POSTER 28

POPULATION PHARMACOKINETIC MODELING OF DIAZEPAM BUCCAL SOLUBLE FILM

Allen H. Heller[1], Stephen Wargacki[2], Cassie Jung[2], David J. Wyatt[3], Guillaume Bonnefois[4], Pierre-Olivier Tremblay[4], A. Mark Schobel[2]

[1]Pharma Study Design, LLC, Woodbridge, CT 06525, USA; [2]Aquestive Therapeutics, Warren, NJ 07059, USA; [3]Syneos Health Clinical Solutions Early Phase, Miami, FL 33131, USA; [4]Syneos Health Early Phase, Montreal, Quebec H3X 2H9, Canada

ABSTRACT

Diazepam buccal soluble film (DBSF) is a novel dosage form of diazepam under development for the management of selected, refractory, patients with epilepsy, on stable regimens of AEDs, who require intermittent use of diazepam to control bouts of increased seizure activity. We have observed that diazepam pharmacokinetics (PK) following a single dose of DBSF are dose-proportional over the range of 5 to 15 mg for both maximum plasma concentration (C_{max}) and area under the curve (AUC). In contrast, diazepam PK following a single dose of diazepam rectal gel (DRG) are dose-proportional for AUC, but less than dose-proportional for C_{max}. To identify doses of DBSF expected to be therapeutically equivalent to labeled doses of DRG, population PK modeling (NONMEM 7.3) for DBSF and DRG was performed using PK data obtained in Phase I studies in adult men and women. DBSF and DRG were separately modeled using a two-compartment model with zero-order or Michaelis-Menten absorption for DBSF and DRG, respectively, with linear elimination and mixed residual error. Using an algorithm intended to apply equal weight to matching on both C_{max} and $AUC_{0\text{-}12}$, preliminary results indicate that DRG doses of 7.5, 10, 12.5, 15, 17.5, and 20 mg are best matched by DBSF doses of 5, 7.5, 7.5, 10, 12.5, and 12.5 mg, respectively. This modeling approach provides the flexibility to match DBSF with DRG doses on any selected PK parameter(s).

BACKGROUND

- Diazepam rectal gel (DRG; Diastat, Valeant Pharmaceuticals, Bridgewater, NJ, USA) is indicated for the management of selected, refractory, patients with epilepsy, on stable regimens of antiepileptic drugs (AEDs), who require intermittent use of diazepam to control bouts of increased seizure activity.
- Use of DRG may be limited by social concerns (embarrassment and desire for privacy)[1,2] and practical concerns (use prohibited in some schools and potential liability,[1,3,4] and absence of trained personnel to administer in a timely manner[1]).
- Diazepam buccal soluble film (DBSF), developed using patented PharmFilm® technology (Aquestive Therapeutics, Warren, NJ, USA), is a buccal soluble film currently under clinical development for the same indication as DRG.
- Phase I studies (as described below) indicated that diazepam pharmacokinetics (PK) following a single dose of DBSF are dose-proportional over the range of 5 to 15 mg for both maximum plasma concentration (C_{max}) and area under the curve (AUC), whereas diazepam PK following a single dose of DRG are dose-proportional for AUC, but less than dose-proportional for C_{max}.
- Population PK (Pop-PK) modeling was undertaken to identify doses of DBSF that best approximate the diazepam exposure (eg, C_{max} and $AUC_{0\text{-}12}$) expected after administration of single doses of DRG at doses (weight-based) according to the approved labeling for DRG.

OBJECTIVES

- To characterize the Pop-PK of diazepam administered as DBSF and DRG in healthy adult volunteers
- To determine the doses of DBSF that best approximate the diazepam exposure (eg, C_{max} and $AUC_{0\text{-}12}$) expected after administration of single doses of DRG at doses (weight-based) according to the approved labeling for DRG

METHODS

DATA EXTRACTION

- Data for the Pop-PK analysis were obtained from three Phase I studies.
- Study 1: Healthy adult male subjects (N=30) received single doses of 1 × 5 mg DBSF, 1 × 10 mg DBSF, or 1 × 15 mg DBSF under fasting conditions in a three-period, six-sequence randomized crossover with a washout of 21 days between doses.
- Study 2: Healthy adult subjects (N=36) received single doses of DRG 5, 12.5, and 20 mg, and DBSF 1 × 15 mg under fasting conditions in a four-period, four-sequence randomized crossover with minimum washout of 28 days between doses.
- Study 3: Healthy adult subjects (N=18) received single doses of 1 × 15 mg DBSF under fasting conditions and 1 × 15 mg DBSF following a high-fat meal in a two-period, two-sequence crossover with a washout of at least 28 days between doses. Only data from the fasting condition were used for this Pop-PK analysis.
- The derived dataset for the modeling was programmed using R, release 3.3.1. The values of the covariates were the contemporaneous values when available and the most recently available values (including screening) when not. The final analysis included 81 subjects dosed with DBSF and 34 subjects dosed with DRG.
- Four subjects were excluded from the Pop-PK analysis due to extremely low diazepam exposure following DRG.

POP-PK MODELS

Base Structural Model

- Both time courses of DBSF and DRG were modeled separately using NONMEM 7.3.0 with the first-order conditional estimation (FOCE) or with the interactions (FOCE-I) method.
- Distribution and clearance were modeled as multiple compartments controlled by first-order processes. The model was parameterized using clearance and volume terms. The final number of compartments was adjusted based on the model fit to the data. The developed structural models ranged between one and three compartments.

Variability Models

- Between-subject variability (BSV): Between-subject random effects on the model parameters were added as exponential terms.
- Residual variability: The residual error model (ie, intrasubject variability) was included as a proportional plus additive components model. With this structure most of the error would be expected to be proportional with high predicted concentrations, whereas additive error would be expected to be most influential with low predicted concentrations (near the lower limit of quantitation).

Stepwise Covariate Model

- The covariate model was established using the stepwise covariate model (SCM) building procedure, run by the Perl-speaks-NONMEM tool SCM. This approach was accomplished through a forward selection step followed by a backward elimination step.
- Covariates investigated for both DBSF and DRG included, but were not limited to, age, albumin, bilirubin, body mass index (BMI), creatinine, ethnicity, fasting status, gender, height, hemoglobin, potassium, protein, race, sodium, urate, and weight.

MODEL EVALUATION

Visual Predictive Check

- Parameter estimates obtained by fitting the final Pop-PK model to the data were used to simulate population profiles. A nonparametric prediction interval around the median, 5th, and 95th percentiles of simulated values was constructed to quantify the variability in the model predictions and to visually compare with the observed values.

Nonparametric Bootstrap

- The mean and the 95% confidence intervals of the parameter estimates from the bootstrap replicates were compared with the estimated parameters from the original dataset to qualify the estimates of the model parameters.

SELECTION OF DBSF DOSES TO MATCH DRG DOSES

- Predicted concentrations for both DBSF and DRG for 500 subjects were simulated for different weight ranges and their corresponding mean, and the median concentration for 500 subjects at each time point was calculated.
- Doses of DBSF were selected that best matched the labeled doses of DRG with respect to diazepam C_{max}, $AUC_{0\text{-}12}$, or a combination of C_{max} and $AUC_{0\text{-}12}$.

RESULTS

POP-PK MODEL OF DBSF

- Base models with different numbers of compartments, different types of absorption or clearance, and different residual error models were tested. A two-compartment model with zero-order absorption and linear elimination was selected. A lag time estimated at 9 minutes was included. Allometric scaling was added for clearance (× [weight/70]$^{0.75}$) and volume of distribution (× weight/70).

Figure 1. DBSF Model Structure

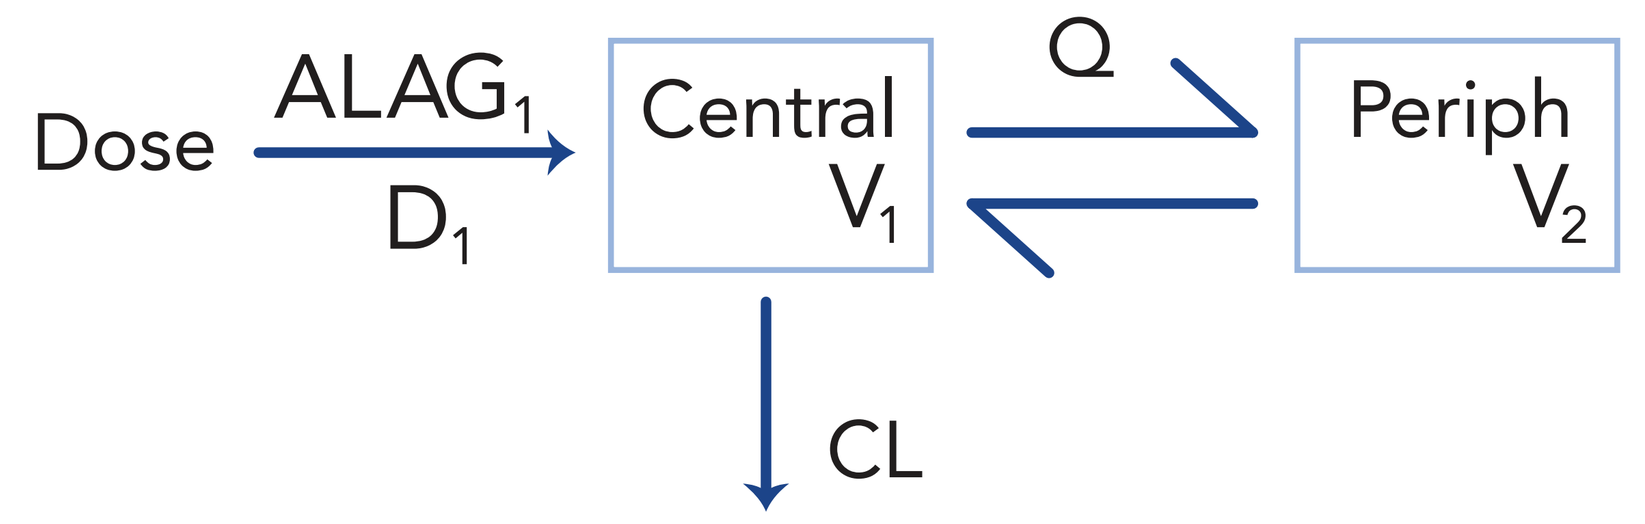
Dose
ALAG1
D1
Central
V1
Q
Periph
V2
CL

ALAG1 (hour): lag time of concentrations to the central compartment; D1 (hour): duration of "infusion-like" administration; V1 (L): central volume of distribution (plasma volume); CL (L/hour): clearance of DBSF diazepam from the central compartment; Q (L/hour): intercompartmental flow; V2 (L): peripheral volume

- Following forward inclusion and backward elimination no covariates were included in the model.
- Visual predictive check validation, based on 500 replications, indicated that the proposed PK model is appropriate to describe the time courses of DBSF plasma concentrations during the absorption or elimination phases for three different dose levels (5 mg/m², 10 mg/m², and 15 mg/m²).
- Nonparametric bootstrap validation (N=207 on 300 replicates) indicated that the precision on all parameters of the model was very good (residual error ≤23%).

POP-PK MODEL OF DRG

- Base models with different numbers of compartments, different types of absorption or clearance, and different residual error models were tested. A two-compartment model with Michaelis-Menten absorption and linear elimination was selected. Allometric scaling was added for clearance (× [weight/70]$^{0.75}$) and volume of distribution (× weight/70).

Figure 2. DRG Model Structure

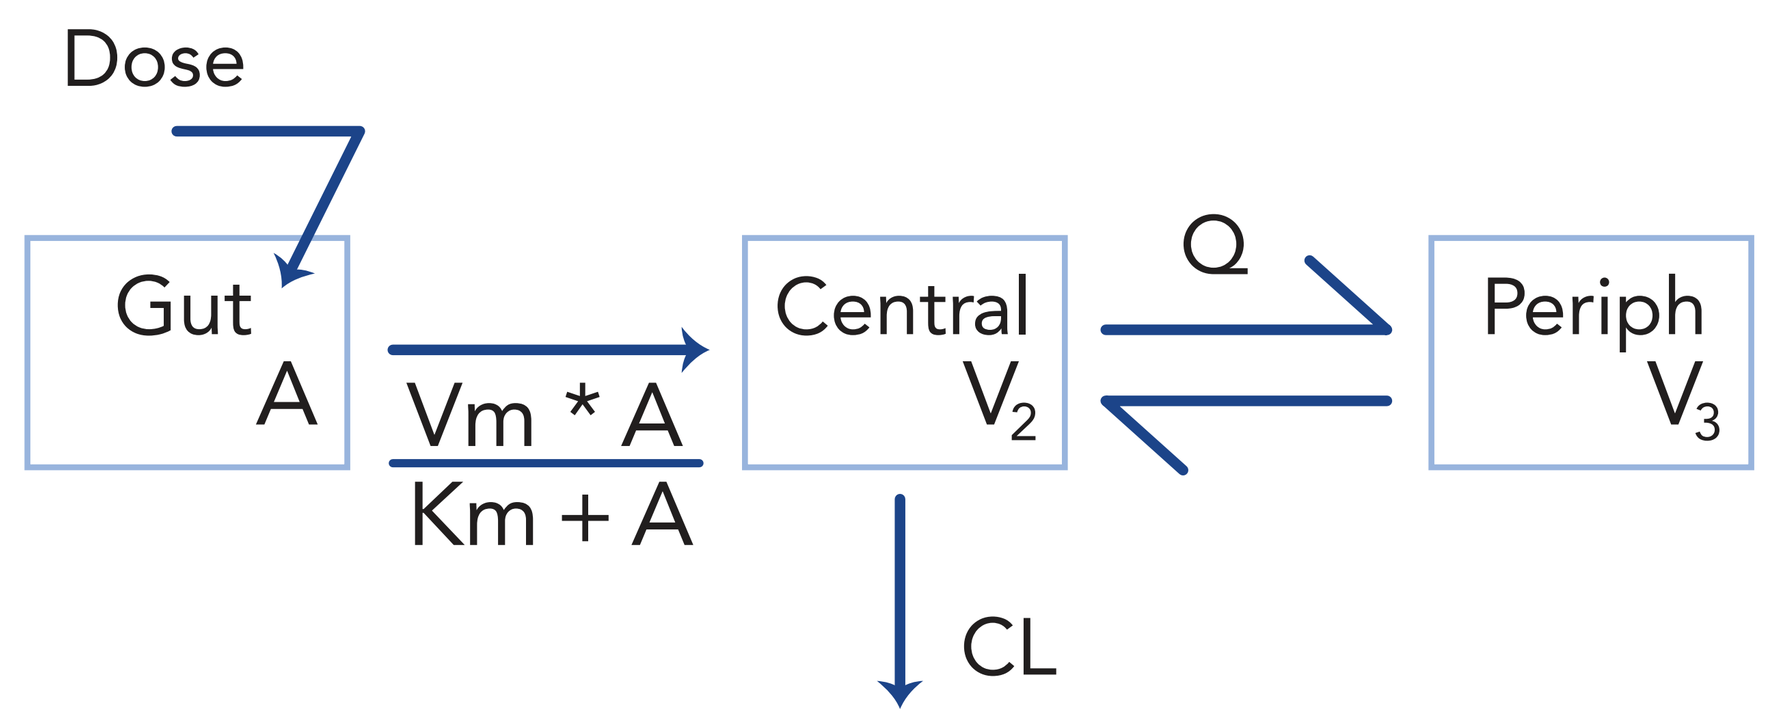
Dose
Gut
A
Vm * A
Km + A
Central
V2
Q
Periph
V3
CL

A: amount of drug; Vm (ng/hour): maximum rate at saturating substrate concentration; Km (ng): substrate concentration at which the reaction rate is half of Vm (Michaelis constant); V2 (L): central volume of distribution (plasma volume); CL (L/hour): clearance of DRG diazepam from the central compartment; Q (L/hour): intercompartmental flow; V3 (L): peripheral volume

- Following forward inclusion and backward elimination no covariates were included in the model.
- Visual predictive check validation, based on 500 replications, indicated that the proposed PK model is appropriate to describe the time courses of DRG plasma concentrations during the absorption or elimination phases for three different dose levels (5 mg/m², 12.5 mg/m², and 20 mg/m²).
- Nonparametric bootstrap validation (N=283 on 300 replicates) indicated that BSV is <50% for all parameters, and precision on these parameters showed residual error <37%.

DBSF DOSES VERSUS DRG DOSES

- The dose of DBSF for each weight category is generally lower than the corresponding dose of DRG whether matching is based on C_{max}, $AUC_{0\text{-}12}$, or a combination of C_{max} and $AUC_{0\text{-}12}$. Matching on C_{max}, the DBSF doses corresponding to DRG doses (depending on weight) range from 2.5 to 10 mg. Matching on $AUC_{0\text{-}12}$, the DBSF doses corresponding to DRG doses range from 5 to 15 mg. Matching on a combination of C_{max} and $AUC_{0\text{-}12}$, the DBSF doses corresponding to DRG doses range from 2.5 to 12.5 mg.

TABLE 1: DBSF DOSES CORRESPONDING TO LABELED DOSES OF DRG: MATCHING ON C_{max}, MATCHING ON $AUC_{0\text{-}12}$, AND MATCHING ON A COMBINATION OF C_{max} AND $AUC_{0\text{-}12}$

Matching on C_{max}			Matching on $AUC_{0\text{-}12}$			Matching on C_{max} + $AUC_{0\text{-}12}$		
Weight (kg)	Dose (mg)		Weight (kg)	Dose (mg)		Weight (kg)	Dose (mg)	
	DRG[a]	DBSF		DRG[a]	DBSF		DRG[a]	DBSF
14–25	5	2.5	14–25	5	5	14–25	5	2.5
26–37	7.5	5	26–37	7.5	5	26–37	7.5	5
38–50	10	7.5	38–50	10	7.5	38–50	10	7.5
51–62	12.5	7.5	51–62	12.5	10	51–62	12.5	7.5
63–75	15	7.5	63–75	15	10	63–75	15	10
76–87	17.5	10	76–87	17.5	12.5	76–87	17.5	10
88–111	20	10	88–111	20	15[b]	88–111	20	12.5

[a]DRG doses are the doses recommended in the Diastat label (weight-based) for adults (individuals age 12 and older)

[b]Based on the average weight instead of limits of weight range

AUC: area under the concentration-time curve; C_{max}: maximum plasma drug concentration; DBSF: diazepam buccal soluble film; DRG: diazepam rectal gel

- Figures 3-5 depict C_{max}, $AUC_{0\text{-}4}$, and $AUC_{0\text{-}12}$ ranges, based on dose matching by C_{max}, $AUC_{0\text{-}12}$, and a combination of C_{max} and $AUC_{0\text{-}12}$, respectively.

Figure 3. C_{max}, $AUC_{0\text{-}4}$, and $AUC_{0\text{-}12}$ Ranges for DBSF and DRG According to Weight for Doses Matched by C_{max}

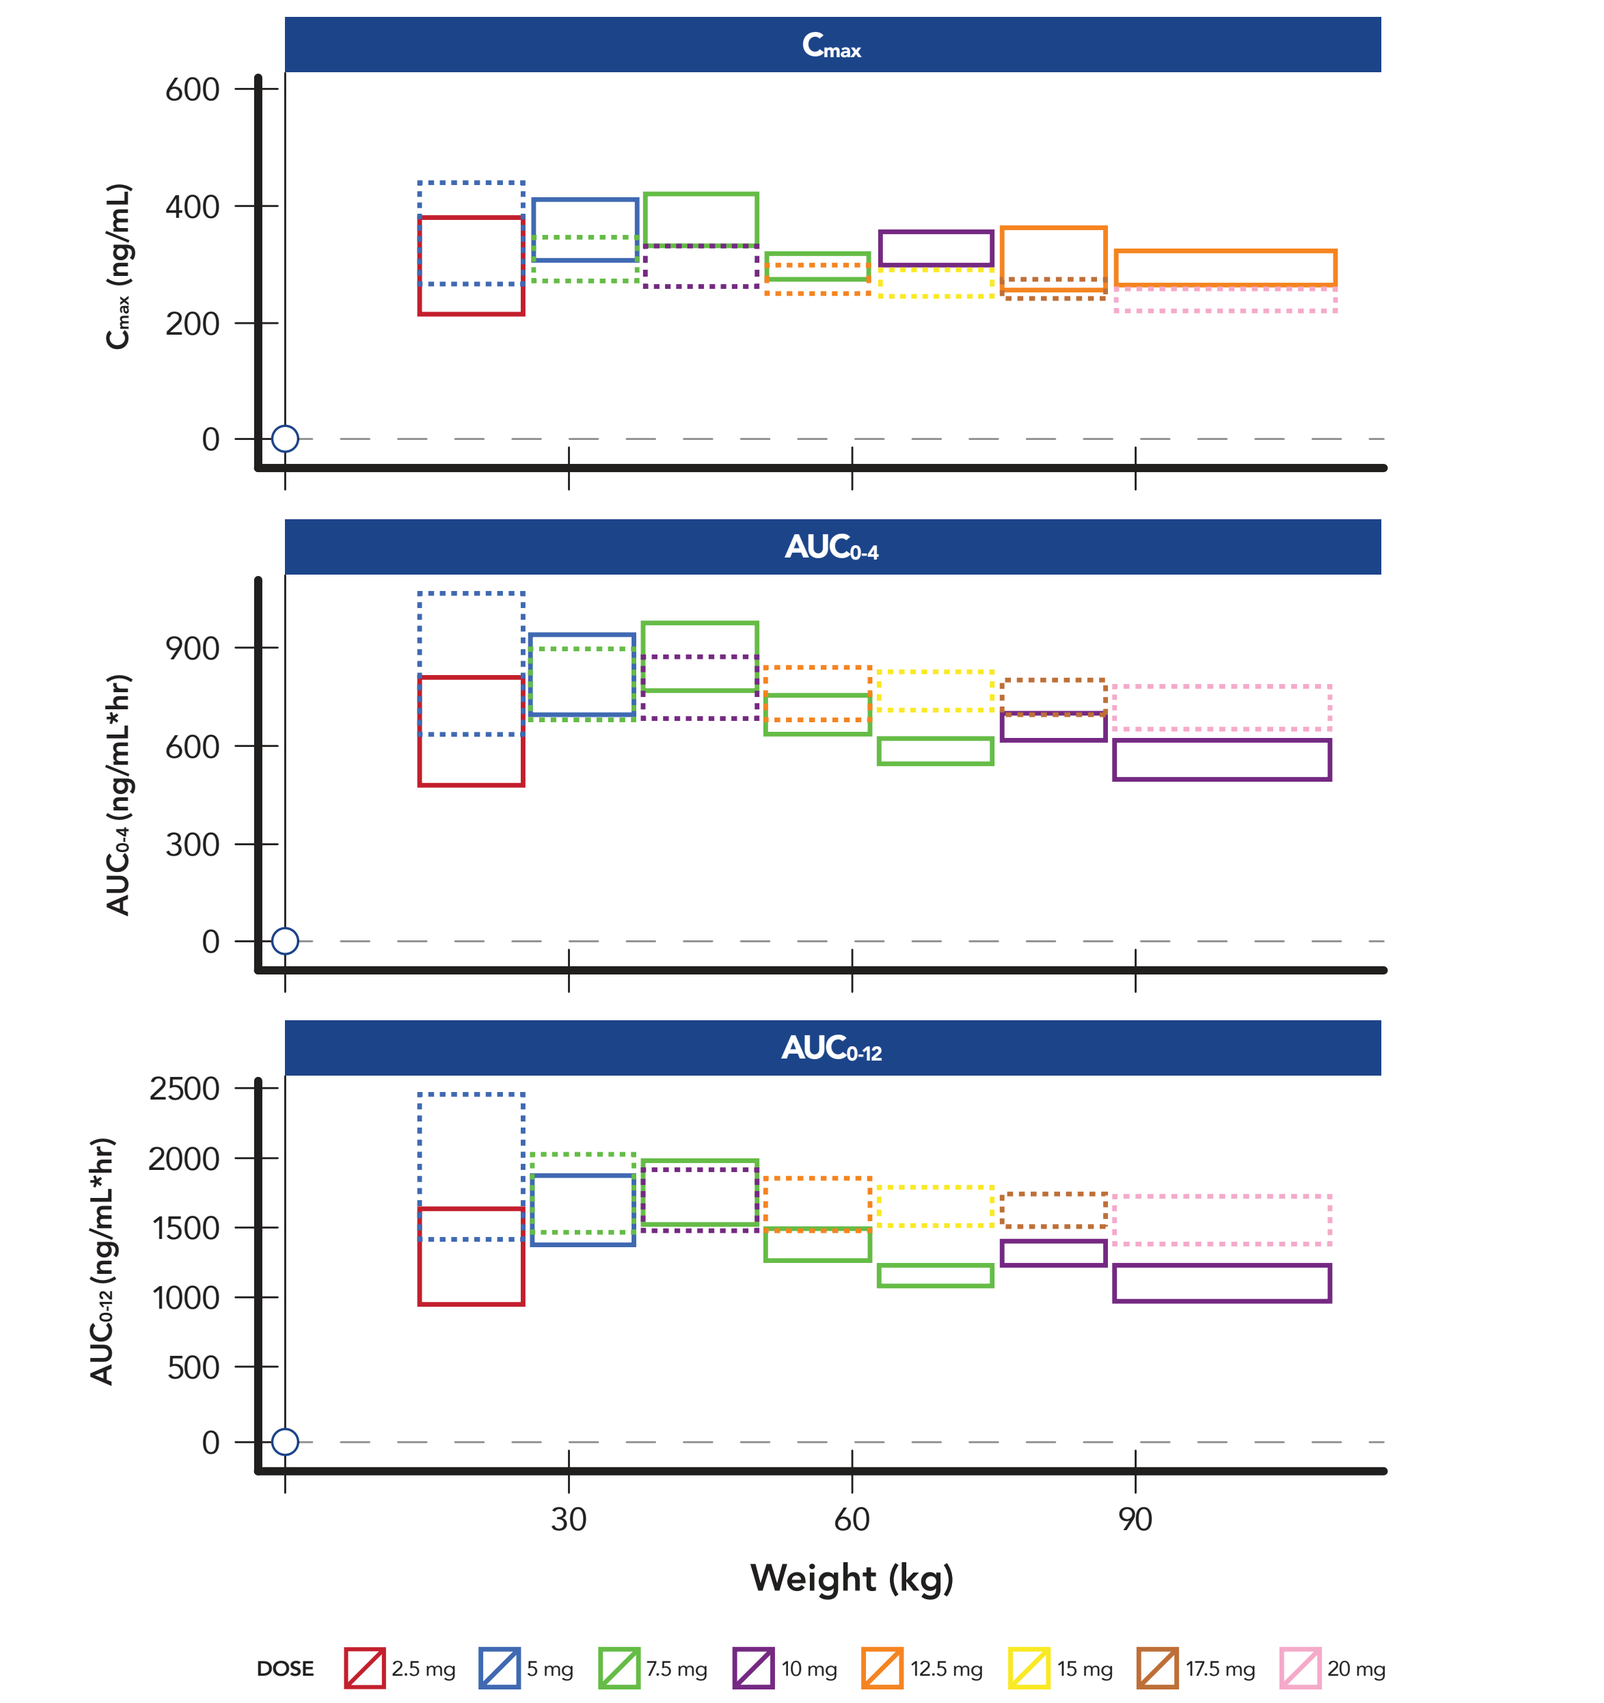
Cmax
Cmax (ng/mL)
AUC0-4
AUC0-4 (ng/mL*hr)
AUC0-12
AUC0-12 (ng/mL*hr)
Weight (kg)
DOSE 2.5 mg 5 mg 7.5 mg 10 mg 12.5 mg 15 mg 17.5 mg 20 mg

Dashed rectangles depict ranges for DRG. Solid rectangles depict ranges for DBSF. Height of rectangles corresponds to the range of the PK parameter for subjects within the weight range. For each boundary of the weight range, the median PK profile was calculated based on 500 replications, and the PK parameter was computed based on these median profiles.

AUC: area under the concentration-time curve; C_{max}: maximum plasma drug concentration; DBSF: diazepam buccal soluble film; DRG: diazepam rectal gel

Figure 4. C_{max}, $AUC_{0\text{-}4}$, and $AUC_{0\text{-}12}$ Ranges for DBSF and DRG According to Weight for Doses Matched by $AUC_{0\text{-}12}$

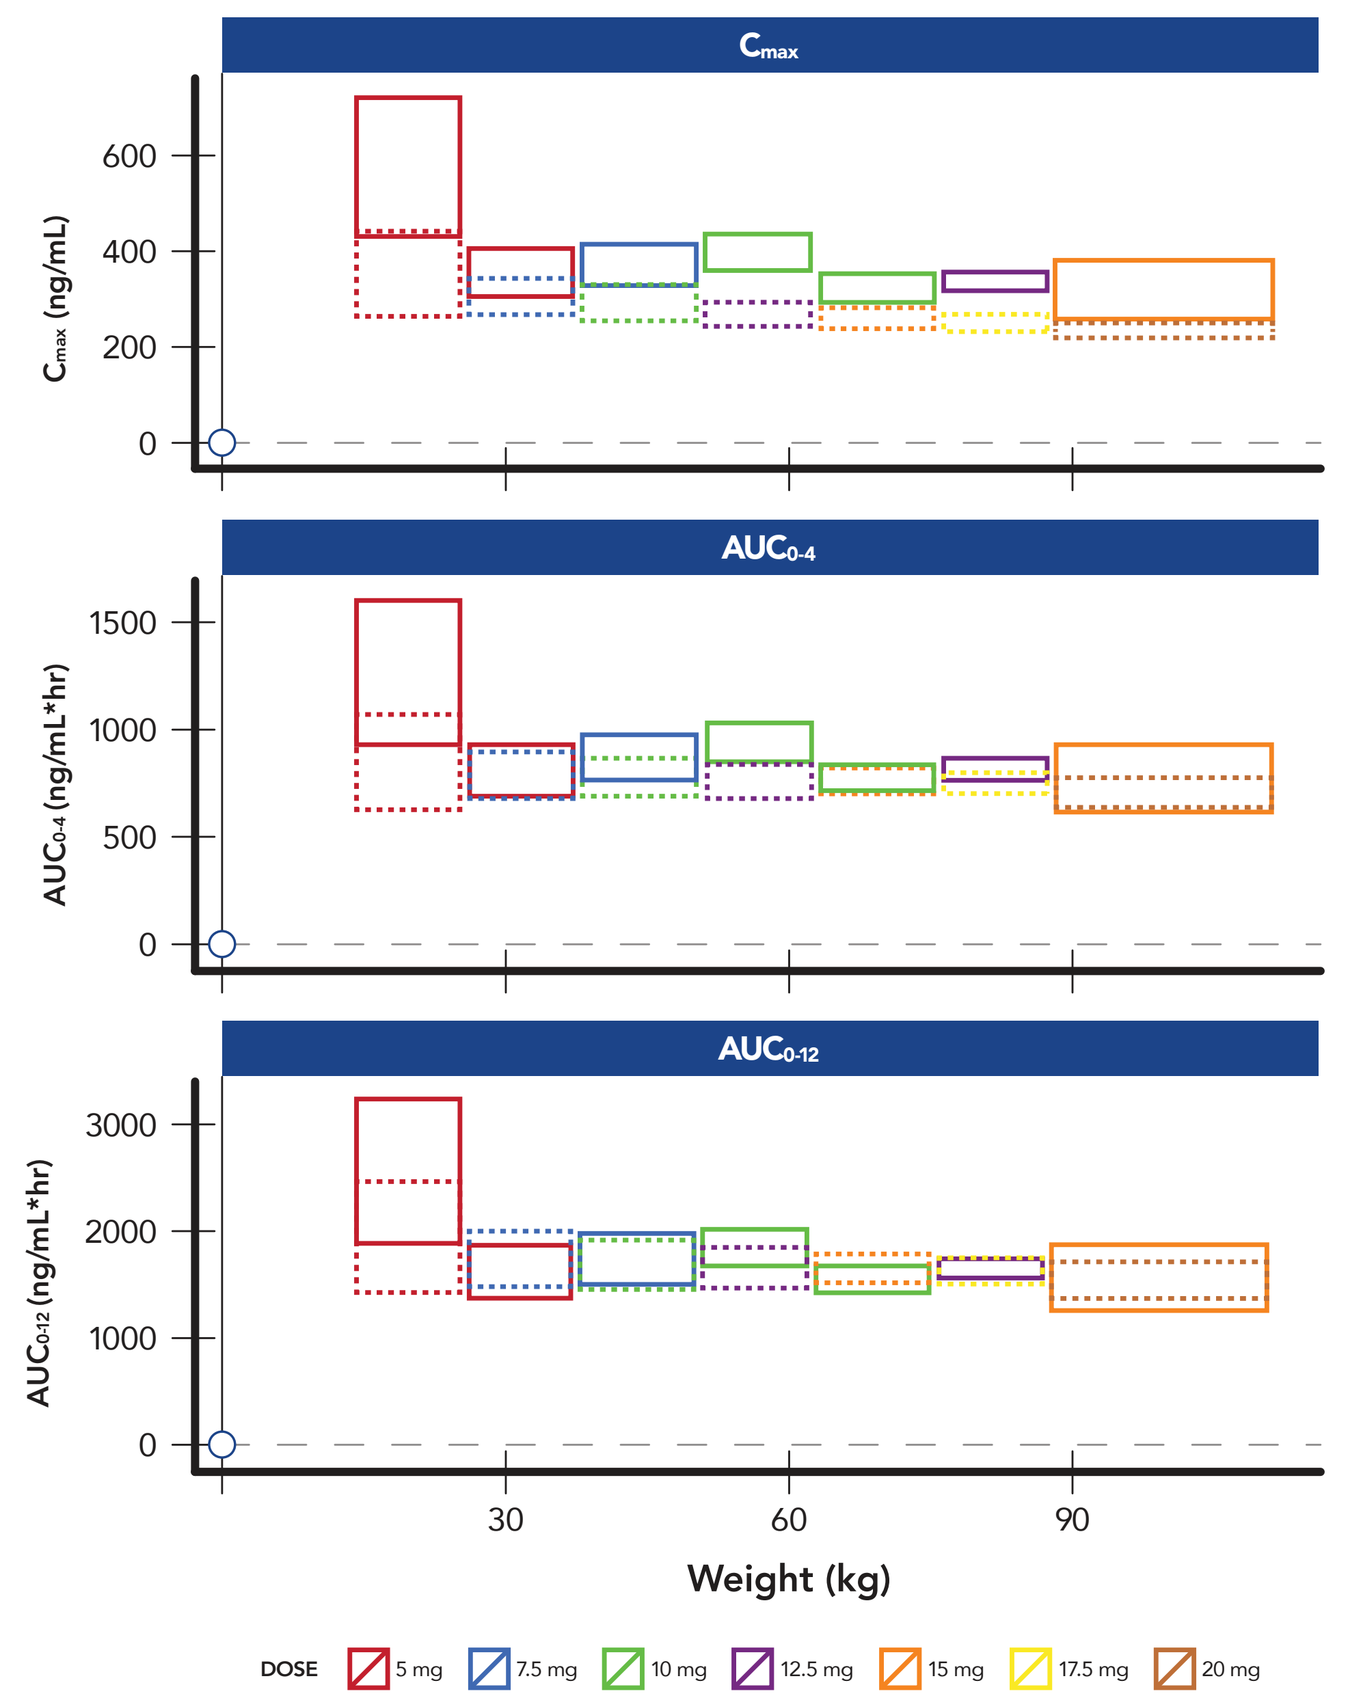
Cmax
Cmax (ng/mL)
AUC0-4
AUC0-4 (ng/mL*hr)
AUC0-12
AUC0-12 (ng/mL*hr)
Weight (kg)
DOSE 5 mg 7.5 mg 10 mg 12.5 mg 15 mg 17.5 mg 20 mg

Dashed rectangles depict ranges for DRG. Solid rectangles depict ranges for DBSF. Height of rectangles corresponds to the range of the PK parameter for subjects within the weight range. For each boundary of the weight range, the median PK profile was calculated based on 500 replications, and the PK parameter was computed based on these median profiles.

AUC: area under the concentration-time curve; C_{max}: maximum plasma drug concentration; DBSF: diazepam buccal soluble film; DRG: diazepam rectal gel

Figure 5. C_{max}, $AUC_{0\text{-}4}$, and $AUC_{0\text{-}12}$ Ranges for DBSF and DRG According to Weight for Doses Matched by the Combination of C_{max} and $AUC_{0\text{-}12}$

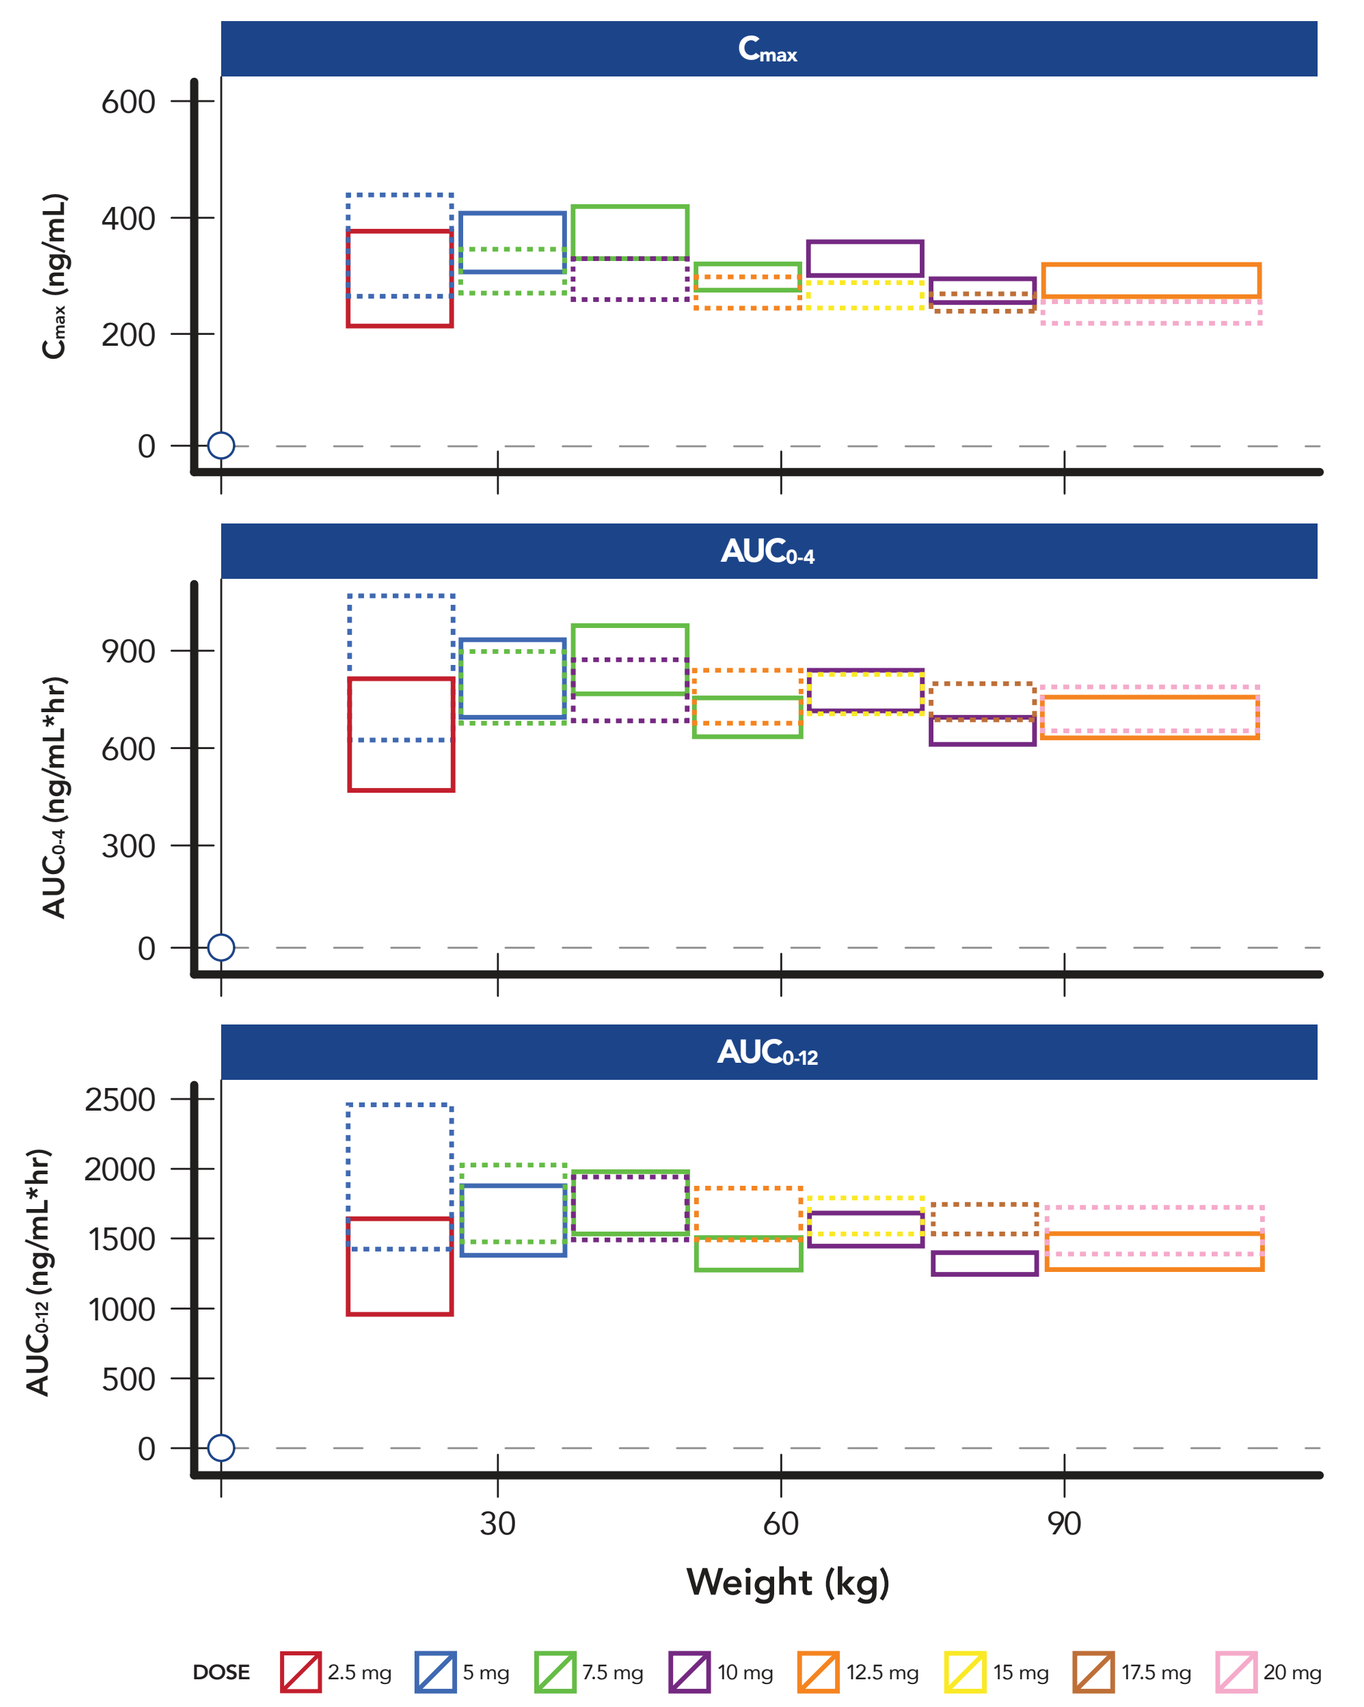
Cmax
Cmax (ng/mL)
AUC0-4
AUC0-4 (ng/mL*hr)
AUC0-12
AUC0-12 (ng/mL*hr)
Weight (kg)
DOSE 2.5 mg 5 mg 7.5 mg 10 mg 12.5 mg 15 mg 17.5 mg 20 mg

Dashed rectangles depict ranges for DRG. Solid rectangles depict ranges for DBSF. Height of rectangles corresponds to the range of the PK parameter for subjects within the weight range. For each boundary of the weight range, the median PK profile was calculated based on 500 replications, and the PK parameter was computed based on these median profiles.

AUC: area under the concentration-time curve; C_{max}: maximum plasma drug concentration; DBSF: diazepam buccal soluble film; DRG: diazepam rectal gel

CONCLUSIONS

- DBSF and DRG have different structural Pop-PK models. DBSF includes a zero-order absorption with a lag time estimated at 9 minutes, whereas the DRG model includes a Michaelis-Menten absorption. Both models have a linear elimination and both were successfully internally validated using visual predictive check and nonparametric bootstrap methods.
- If a mono-objective optimization based only on C_{max} or $AUC_{0\text{-}12}$ is performed, the DBSF doses that match DRG doses in adults (5 mg to 20 mg) are between 2.5 and 10 mg, and between 5 and 15 mg, respectively. With multi-objective optimization (ie, combined C_{max} and $AUC_{0\text{-}12}$), the DBSF doses that match DRG doses range from 2.5 to 12.5 mg.
- This modeling approach provides the flexibility to match DBSF doses with DRG doses on any selected PK parameter(s).

REFERENCES

1. Hartman AL, Devore CD, and the Section on Neurology, Council on School Health. Rescue medicine for epilepsy in education settings. *Pediatrics*. Jan 2016;137(1).
2. Tatum IW. Adult patient perceptions of emergency rectal medications for refractory seizures. *Epilepsy Behav*. Dec 2002;3(6):535-538.
3. O'Dell C, O'Hara K. School nurses' experience with administration of rectal diazepam gel for seizures. *J Sch Nurs*. Jun 2007;23(3):166-169.
4. Terry D, Paolicchi J, Karn M. Acceptance of the use of diazepam rectal gel in school and day care settings. *J Child Neurol*. Sep 2007;22(9):1135-1138.

ACKNOWLEDGEMENTS

Medical writing support was provided by Jonathan Sokolowski, PhD, of ACCESS Medical, LLC; medical writing support was funded by Aquestive Therapeutics.

FUNDING AND DISCLOSURES

Financial support for this study was provided by Aquestive Therapeutics. Aquestive Therapeutics participated in the interpretation of data, review, and approval of the poster. All authors contributed to the development of the poster and maintained control over the final content. Allen H. Heller is a consultant to Aquestive Therapeutics. Stephen Wargacki, Cassie Jung, and A. Mark Schobel are employees of Aquestive Therapeutics.

Aquestive