COMPARATIVE PHARMACOKINETICS OF DIAZEPAM BUCCAL SOLUBLE FILM AND DIAZEPAM RECTAL GEL

Allen H. Heller[1], Stephen Wargacki[2], Tracy J. Stalvey[2], David J. Wyatt[3], A. Mark Schobel[2]

[1]Pharma Study Design, LLC, Woodbridge, CT 06525, USA; [2]Aquestive Therapeutics, Warren, NJ 07059, USA; [3]Syneos Health Clinical Solutions Early Phase, Miami, FL 33131, USA

ABSTRACT

Objective: To assess the pharmacokinetics (PK) of Diazepam Buccal Soluble Film (DBSF) compared with diazepam rectal gel (DRG).

Background: DBSF is a novel dosage form of diazepam under development for the management of selected, refractory, patients with epilepsy, on stable regimens of anti-epileptic drugs, who require intermittent use of diazepam to control bouts of increased seizure activity. DBSF PK following a single dose is dose-proportional over the range 5 to 15 mg.

Design/Methods: Healthy men and women age 18-65 received single doses of DBSF 15 mg (buccal), or DRG 5 mg, 12.5 mg, or 20 mg (per rectum) under fasting conditions in an open-label, randomized sequence crossover with 28-day washout. Subjects were confined from 14 hours before until 24 hours post-dose. Plasma samples for diazepam were drawn pre-dose and at intervals until 240 hours. Using log-transformed data, PK parameters following DBSF 15 mg were compared with the corresponding parameters following DRG. Dose-proportionality for DRG was assessed by 2 statistical methods. Treatment-emergent adverse events (TEAE) were recorded.

Results: Thirty-six subjects received at least one dose of DBSF. Mean diazepam C_{max} values were highest following 15 mg DBSF, consistent with observation that C_{max} was less than dose-proportional with increasing dose of DRG. Variability (AUC, C_{max}, T_{max}) was consistently greater for DRG than for DBSF. Two subjects exhibited extremely low plasma concentrations following DRG. TEAEs were as expected for diazepam with somnolence being the most frequent. Two subjects were discontinued because of excessive somnolence following DBSF.

Conclusions: With increasing dose of DRG, C_{max} was less than dose-proportional (unlike DBSF). Additionally, PK parameters following DRG exhibited consistently greater variability than DBSF. Diazepam was safe and generally well tolerated in healthy adults following single doses of DBSF 15 mg, DRG 5 mg, 12.5 mg, and 20 mg.

BACKGROUND

- The route of administration diazepam rectal gel (DRG) is not ideal, and may affect adherence[1]
- Diazepam Buccal Soluble Film (DBSF) is a novel dosage form of diazepam under development as a therapeutic alternative to DRG
- DBSF contains diazepam in a dissolvable polymer-based matrix designed to facilitate transport across the buccal mucosa
- We have demonstrated that diazepam pharmacokinetics (PK) after single doses of 5 mg, 10 mg, and 15 mg DBSF are dose-proportional for C_{max} and AUC[2]
- Pilot studies (Aquestive Therapeutics, data on file) suggested that diazepam PK after single doses of 5 mg and 20 mg DRG were dose-proportional for AUC, but less than dose-proportional for C_{max}
- The current investigation compared the PK of diazepam following single doses of DRG and DBSF

OBJECTIVES

- **Primary Objective**: Describe the diazepam PK of DRG over its prescribed dose range (single doses: 5 mg, 12.5 mg, and 20 mg) and compare with the PK of DBSF (single dose: 15 mg) in healthy male and female adult volunteers
- **Secondary Objectives**:
 - Investigate the dose-proportionality of single doses of DRG over the dose range 5-20 mg
 - Evaluate the safety and tolerability of single doses of DBSF 15 mg and DRG 5 mg, 12.5 mg, and 20 mg

METHODS

STUDY DESIGN

- Single-dose, randomized, 4-period, 4-sequence, open-label crossover at a single site in Miami, FL (**Table 1**)

SUBJECTS

- Healthy, non-smoking (≥6 months prior to screening), male and female volunteers, 18-65 years of age with a body mass index (BMI) 18.5-29.9 kg/m² were eligible for participation
- Individuals were excluded if they had clinically significant illnesses or conditions, history of drug abuse or dependence, use of benzodiazepines within 30 days, or tobacco use within 6 months of study drug administration
- All subjects provided informed consent. The study protocol and informed consent were reviewed and approved by an ethics committee (Chesapeake IRB) prior to study initiation

PROCEDURES

- Eligibility was established at a screening visit within 30 days of study drug administration
- Subjects were confined from 14 hours prior to drug administration until 24 hours post-dose, returning to the clinic for subsequent blood draws

TABLE 1: SUBJECTS RANDOMIZED TO ONE OF FOUR TREATMENT SEQUENCES

Sequence	Period 1		Period 2		Period 3		Period 4
1	15 mg DBSF	Washout	20 mg DRG	Washout	12.5 mg DRG	Washout	5 mg DRG
2	10 mg DRG		15 mg DBSF		12.5 mg DRG		20 mg DRG
3	12.5 mg DRG		10 mg DRG		20 mg DRG		15 mg DBSF
4	20 mg DRG		12.5 mg DRG		15 mg DBSF		10 mg DRG

DBSF, diazepam buccal soluble film; DRG, diazepam rectal gel

Washout periods were 28 days

PHARMACOKINETIC SAMPLING AND BIOANALYSIS

- Plasma samples for diazepam were drawn pre-dose and post-dose at 0.25, 0.5, 0.75, 1, 1.5, 2, 3, 4, 6, 8, 12, 24, 48, 72, 96, 120, 144, 192, and 240 hours in each study period
- There was a 28 day washout period between treatments
- Plasma was analyzed for diazepam by a high-performance liquid chromatography method using tandem mass spectroscopy and automated extraction for diazepam (1.00 to 1000 ng/mL) in human EDTA K_2 plasma

SAFETY AND TOLERABILITY

- Treatment-emergent adverse events (TEAE), vital signs, and standard laboratory evaluations were recorded

STATISTICAL ANALYSIS

Summary statistics

- PK parameters were calculated and tabulated by treatment group with summary statistics

Comparison of DRG PK parameters with DBSF

- Diazepam exposure following DRG at each of the 3 studied doses (5 mg, 12.5 mg, and 20 mg) was compared with diazepam exposure following DBSF 15 mg. Pairwise comparisons between each dose of DRG (DRG 5 mg, DRG 12.5 mg, and DRG 20 mg) and DBSF 15 mg were evaluated using analysis of variance (ANOVA), with sequence, treatment, and period as fixed factors and subject nested within sequence as a random factor on the In-transformed AUC_{0-t}, AUC_{0-inf}, and C_{max}
- The ratios of geometric means (15 mg DBSF/5 mg DRG; 15 mg DBSF/12.5 mg DRG; and 15 mg DBSF/20 mg DRG) and corresponding 90% confidence intervals (CI), based on least-squares mean (LSM) from the ANOVA of the In-transformed data, were calculated for AUC_{0-t}, AUC_{0-inf}, and C_{max}

Tests of dose-proportionality for DRG

- Dose proportionally analysis was performed for DRG (5 mg, 12.5 mg and 20 mg)
- Comparisons between treatment groups (DRG 5 mg vs 12.5 mg, and 20 mg vs 12.5 mg) were evaluated using ANOVA, with sequence, treatment, and period as fixed factors and subject nested within sequence as a random factor on the in-transformed AUC_{0-t}, AUC_{0-inf}, and C_{max} dose-normalized to the 12.5 mg dose
- The ratios of geometric means (5 mg/12.5 mg and 20 mg/12.5 mg, dose-normalized) and corresponding 90% CI, based on LSM from the ANOVA of the In-transformed data, were calculated for AUC_{0-t}, AUC_{0-inf}, and C_{max}
- A secondary dose-proportionality analysis evaluated dose-proportionality of DRG from 5 mg to 20 mg on AUC_{0-t}, AUC_{0-inf}, and C_{max} using the power method
- Criteria for dose-proportionality:
 - Using paired comparisons (DRG 5 mg vs 12.5 mg, and 20 mg vs 12.5 mg) dose-normalized to 12.5 mg, the 90% geometric CIs of the ratios of LSMs of the In-transformed AUC_{0-t}, AUC_{0-inf}, and C_{max} were to be between 80% and 125%
 - Using the power method, the 90% CIs for the slopes for AUC_{0-t}, AUC_{0-inf}, and C_{max} were to contain 1

RESULTS

SUBJECTS

- Of the total 36 subjects, the 52.8% were female with a mean age of 40.4 years and a mean BMI of 25.20 kg/m². The majority were white (88.9%) and Hispanic or Latino (88.9%)
- All 36 subjects received at least 1 dose of DBSF or DRG
- Baseline demographic characteristics are summarized in **Table 2**

TABLE 2: SUMMARY OF DEMOGRAPHIC CHARACTERISTICS (SAFETY POPULATION; N=36

Category	Result
Age (years), mean ± SD	40.4 ± 11.8
18-40, n (%)	20 (55.6)
>40, n (%)	16 (44.4)
Female, n (%)	19 (52.8)
Ethnicity	
Not Hispanic or Latino, n (%)	4 (11.1)
Hispanic or Latino, n (%)	32 (88.9)
Race	
White, n (%)	32 (88.9)
Black, n (%)	4 (11.1)
BMI (kg/m²), mean ± SD	25.20 ± 2.69

BMI, body mass index; SD, standard deviation

PHARMACOKINETICS

- Mean plasma concentration-time profiles by treatment are shown in **Figure 1**
 - Diazepam mean concentration-time curve between 0.5 h and 3 h is higher for 15 mg DBSF than for any dose of DRG
 - As expected, diazepam mean concentration-time curves for DRG 5 mg, 12.5 mg, and 20 mg are higher with increasing dose
- Summary statistics of pharmacokinetic parameters by treatment are shown in **Table 3**
 - Mean C_{max} for DBSF 15 mg is higher than mean C_{max} for all doses of DRG
 - AUC_{0-t} and AUC_{0-inf} values for DBSF 15 mg are intermediate between the values for DRG 12.5 mg and 15 mg
 - Variability (AUC, C_{max}) was consistently greater for DRG than for DBSF
 - Two subjects exhibited extremely low plasma concentrations following all doses of DRG

Figure 1. Mean diazepam plasma concentrations over time (0–4 hours)

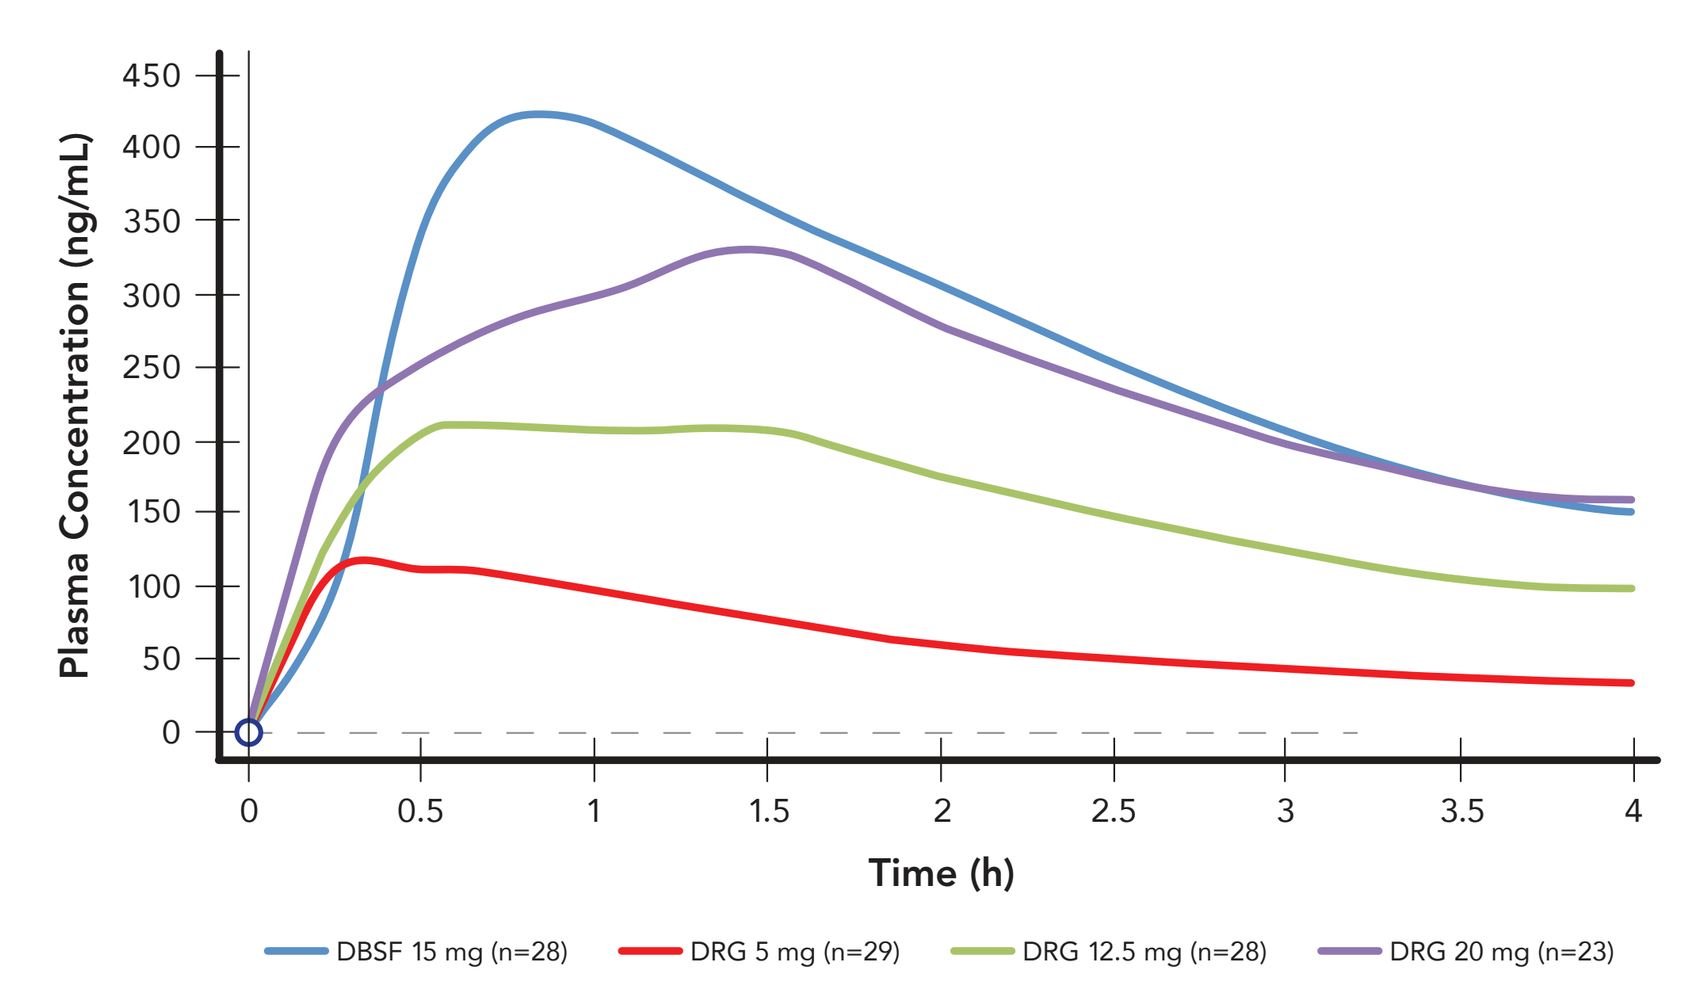
Plasma Concentration (ng/mL)
Time (h)
DBSF 15 mg (n=28)
DRG 5 mg (n=29)
DRG 12.5 mg (n=28)
DRG 20 mg (n=23)

TABLE 3: SUMMARY OF PHARMACOKINETIC PARAMETERS FOR DIAZEPAM

Parameter (units)	DBSF (n=28)		DRG 5 mg (n=29)		DRG 12.5 mg (n=28)		DRG 20 mg (n=23)[a]	
	Mean	CV%	Mean	CV%	Mean	CV%	Mean	CV%
AUC_{0-t} (h*ng/mL)	11289.03	22.72	3365.68	31.59	8377.30	33.58	13716.90[b]	29.78[b]
AUC_{0-inf} (h*ng/mL)	13515.15	28.11	4036.85	40.18	9477.73	33.80	16435.21	35.69
Residual area (%)	14.48	83.97	14.46	74.47	11.10	71.09	14.52	79.53
C_{max} (ng/mL)	469.67	23.02	129.08	37.60	248.01	43.98	364.93[b]	33.23[b]
$T_{1/2el}$ (h)	91.78	57.58	83.76	50.70	76.90	43.89	93.06	56.92
K_{el} (/h)	0.0097	47.3559	0.0100	42.7581	0.0106	41.2718	0.0093	43.7750
Cl/F (L/h)	1.20	29.16	1.73	119.30	2.06	158.28	1.70	124.77
Cl/F/Kg (L/h/kg)	0.0164	24.4	0.0232	97.1	0.0270	134	0.0233	129
V_d/F (L)	145.73	43.31	176.52	75.61	189.73	96.13	198.35	101.97
V_d/F/Kg (L/kg)	1.99	40.32	2.41	61.64	2.52	84.45	2.70	105.56

Parameter (units)	DBSF Median	DBSF Min	DBSF Max	DRG 5 mg Median	DRG 5 mg Min	DRG 5 mg Max	DRG 12.5 mg Median	DRG 12.5 mg Min	DRG 12.5 mg Max	DRG 20 mg Median	DRG 20 mg Min	DRG 20 mg Max
T_{max} (h)	0.998	0.498	3.000	0.500	0.250	8.000	1.000	0.500	8.000	1.500	0.500	3.000

AUC_{0-t}, Area under the concentration-time curve from time zero until the last measurable concentration; AUC_{0-inf}, area under the concentration-time curve from time zero to infinity (extrapolated); C_{max}, maximum plasma concentration; $T_{1/2el}$, terminal elimination half-life, calculated as ln(2)/K_{el}; K_{el}, terminal elimination rate constant; Cl/F, clearance, calculated as Dose/AUC_{0-inf}; V_d/F, volume of distribution, calculated as Cl/F/K_{el}; volume of distribution/kg estimated from (Cl/F/weight)/K_{el}; T_{max}, time of C_{max}

[a]Unless otherwise specified

[b]n=24

TESTS FOR DOSE-PROPORTIONALITY OF DRG

- In the test for dose-proportionality based on paired comparisons (DRG 5 mg vs 12.5 mg, and 20 mg vs 12.5 mg) dose-normalized to 12.5 mg, the 90% geometric CIs of the ratios of LSMs of the In-transformed C_{max} are not between 80% and 125%, indicating that DRG is less than dose-proportional for C_{max} (**Table 4**)
- In the test for dose-proportionality by the power method, the upper limit of the 90% CI for the slope for C_{max} is less than 1 (data not shown), indicating that C_{max} is less than dose-proportional over the studied dosing range
- By both methods, AUC_{0-t} and AUC_{0-inf} are dose-proportional

TABLE 4: SUMMARY OF DOSE-PROPORTIONALITY RESULTS USING ANOVA (PAIRED COMPARISONS: DRG 5 MG VS 12.5 MG, AND 20 MG VS 12.5 MG, DOSE-NORMALIZED TO 12.5 MG)

Treatment comparison	N	Parameter	Geometric LSM (5 mg)	Geometric LSM (12.5 mg)	Ratio[a]	90% Geometric CI[b] Lower	90% Geometric CI[b] Upper	Intra-subject CV	Inter-subject CV
DRG 5 mg vs DRG 12.5 mg	24	AUC_{0-t}	7599.58	8485.95	89.55%	82.73%	96.94%	14.44%	62.83%
	24	AUC_{0-inf}	8698.67	9680.62	89.86%	82.13%	98.31%	16.39%	61.04%
	24	C_{max}	284.79	246.42	115.57%	97.33%	137.23%	31.90%	71.47%
DRG 20 mg vs DRG 12.5 mg	21	AUC_{0-t}	7970.57	8202.70	97.17%	85.74%	110.13%	20.38%	46.93%
	21	AUC_{0-inf}	9357.44	9651.23	96.96%	86.08%	109.21%	19.36%	48.29%
	21	C_{max}	208.42	227.84	91.48%	81.66%	102.48%	18.46%	65.84%

CV, coefficient of variation; AUC_{0-t}, area under the concentration-time curve from time zero until the last measurable concentration; AUC_{0-inf}, area under the concentration-time curve from time zero to infinity (extrapolated); C_{max}, maximum plasma concentration; LSM, least-squares mean; CI, confidence interval

[a]Calculated using least-squares means according to the formula: e(difference) x 100

- Plots comparing LS mean C_{max}, AUC_{0-t} and AUC_{0-inf} values for DBSF and DRG are shown in **Figure 2**
- The relationship between C_{max} and dose for DBSF and DRG is illustrated in **Figure 3A**
- The relationship between AUC_{0-t} and dose for DBSF and DRG is illustrated in **Figure 3B**

Figure 2. LS mean values for (A) C_{max}, (B) AUC_{0-t}, and (C) AUC_{0-inf} after dosing with DBSF and DRG

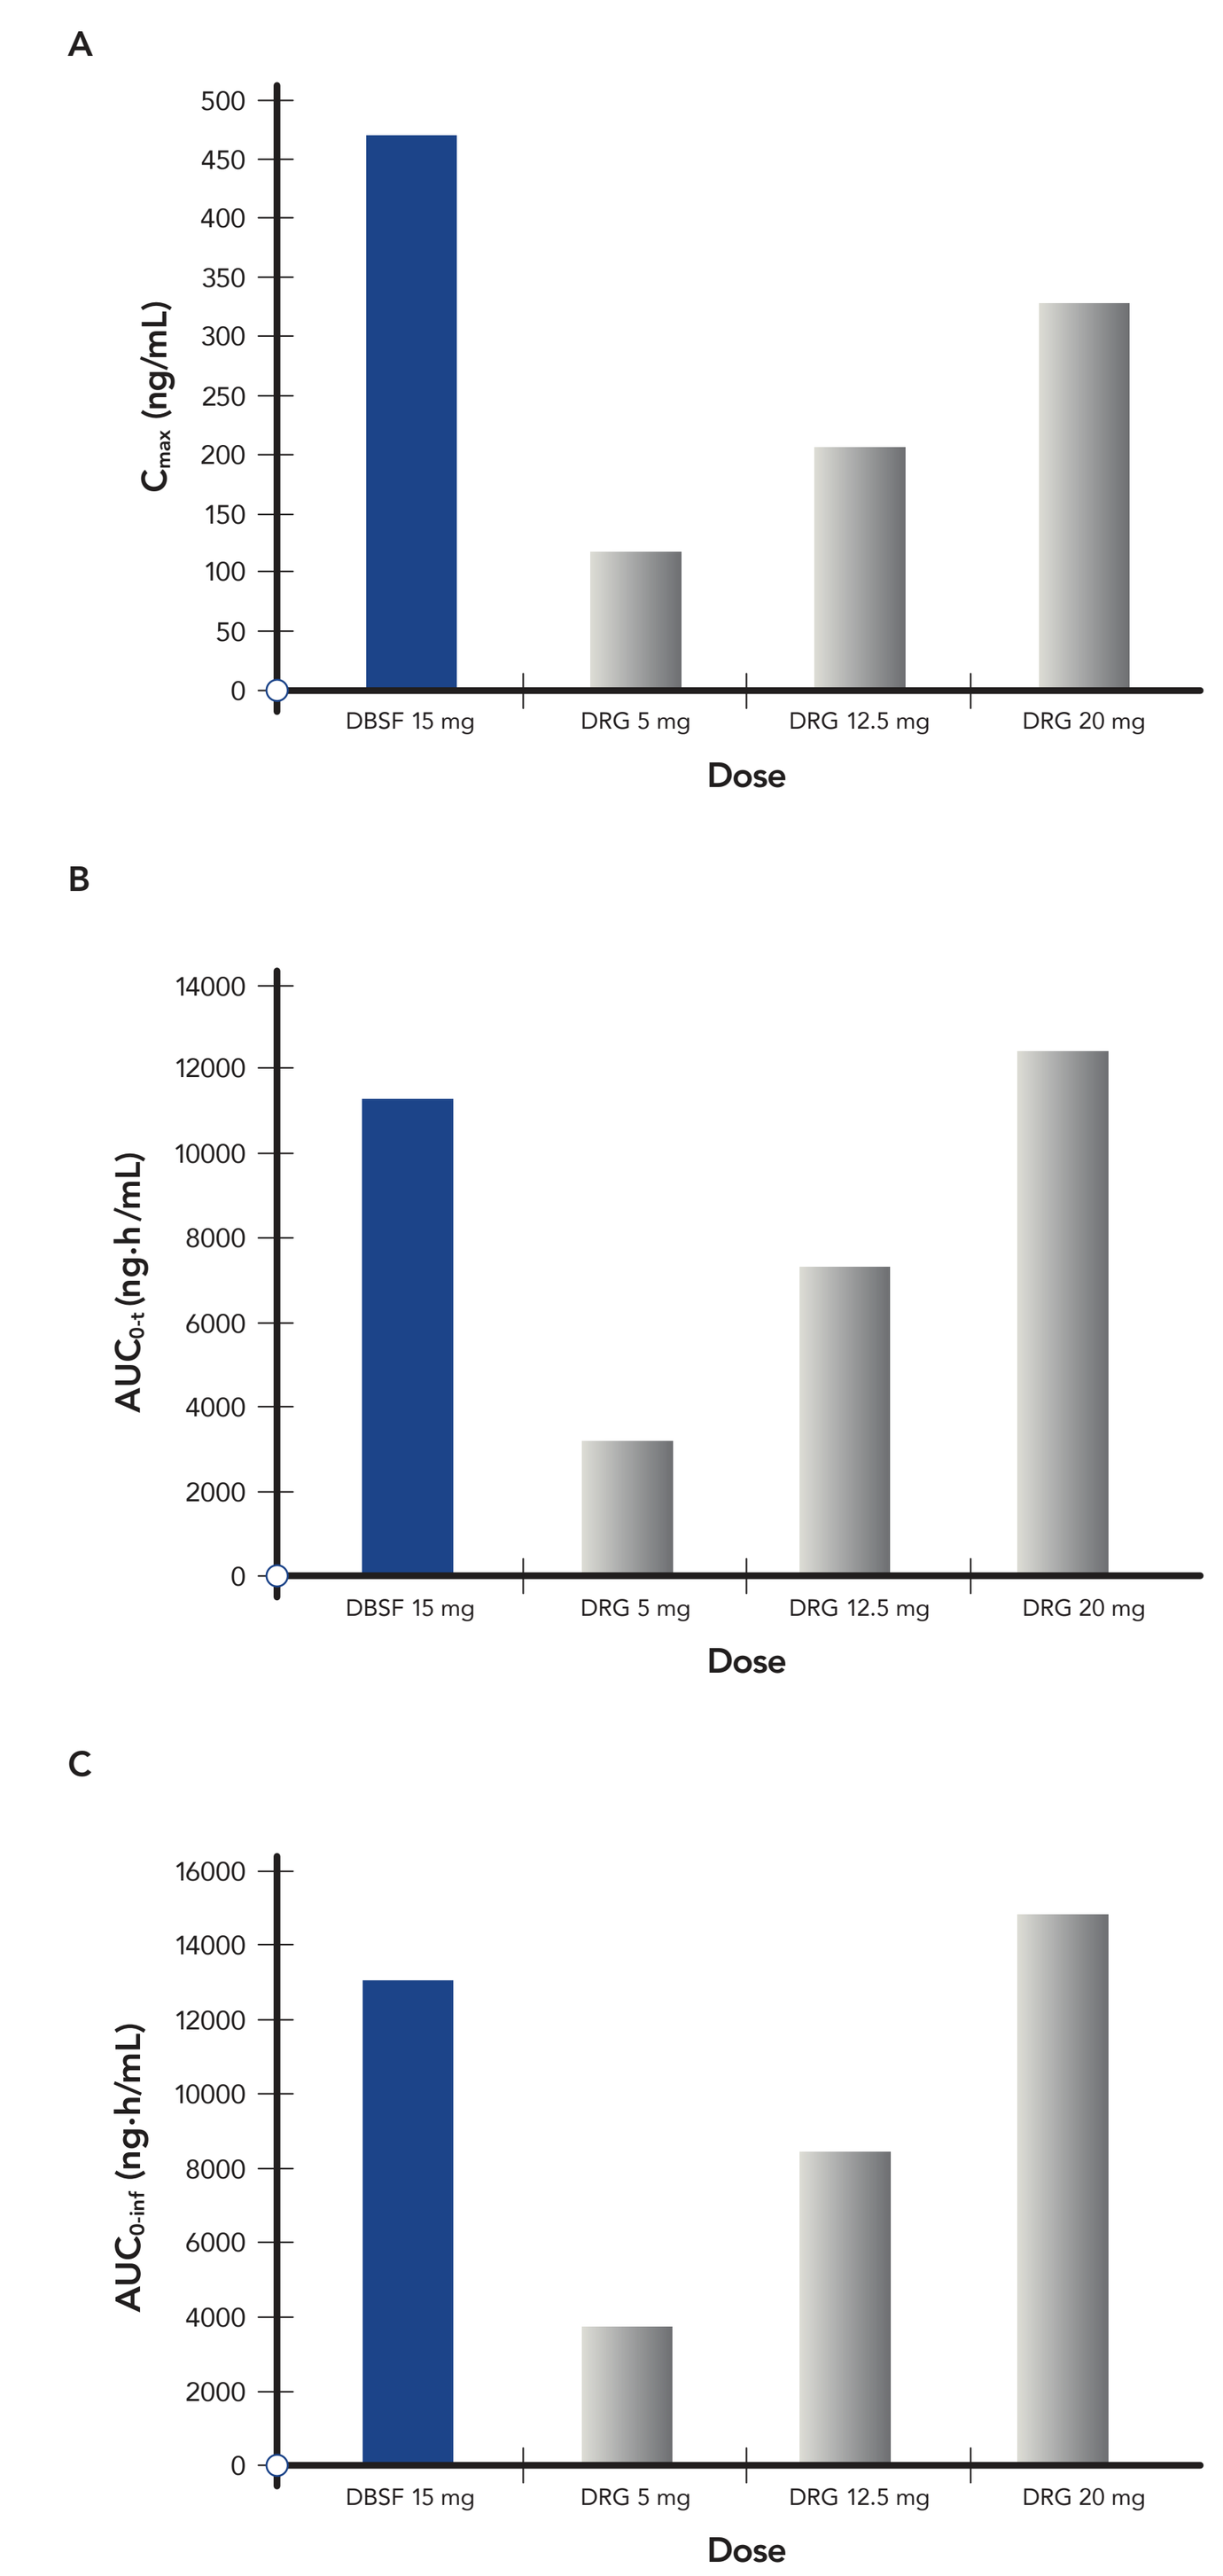
A
C_{max} (ng/mL)
B
AUC_{0-t} (ng·h/mL)
C
AUC_{0-inf} (ng·h/mL)
DBSF 15 mg
DRG 5 mg
DRG 12.5 mg
DRG 20 mg
Dose

Values for parameters are LS means (In-transformed data) from ANOVA of pair-wise comparisons: DBSF 15 mg vs DRG 5 mg (n=24); DBSF 15 mg vs DRG 10 mg (n=23); DBSF 15 mg vs DRG 20 mg (n=21)

Figure 3. (A) Mean C_{max} by dose of diazepam; (B) mean AUC_{0-t} by dose of diazepam. (Linear scale; DRG values fit by linear regression. C_{max} proportional to dose to the ⅔ power; AUC directly proportional to dose.)

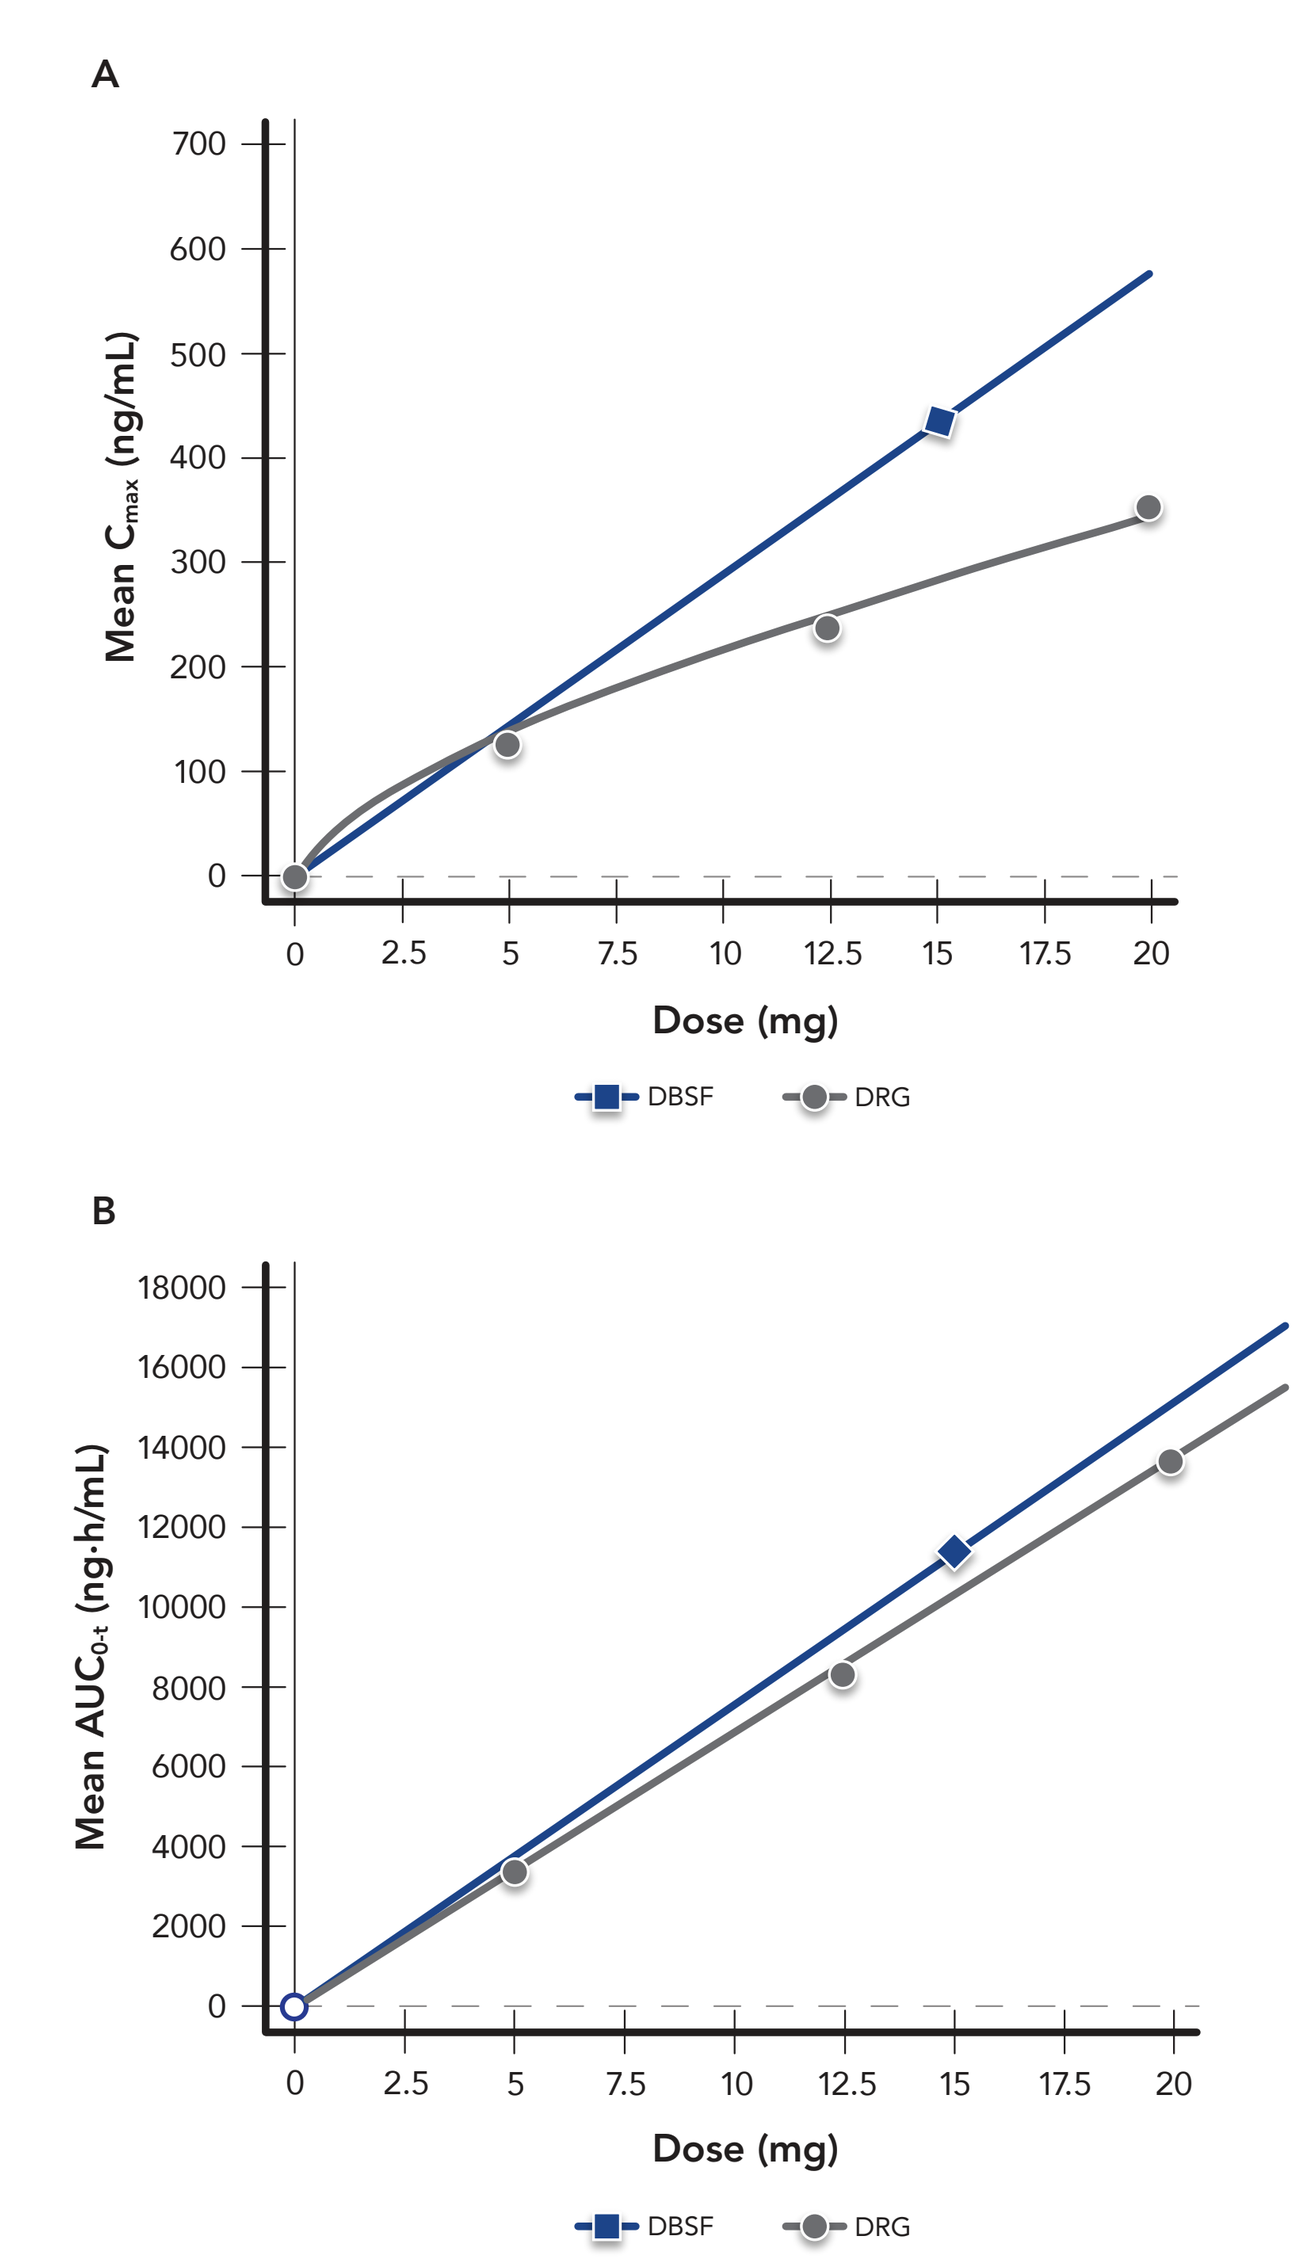
A
Mean C_{max} (ng/mL)
B
Mean AUC_{0-t} (ng·h/mL)
Dose (mg)
DBSF
DRG

SAFETY AND TOLERABILITY

- A total of 131 TEAEs were reported by 34 (94.4%) of the 36 subjects who received at least 1 dose of study medication (**Table 6**)
- As expected, the frequency of TEAEs was related to diazepam exposure: DBSF 15 mg > DRG 20 mg > DRG 12.5mg > DRG 5 mg
 - The most frequent TEAE was somnolence
 - Note that 15 mg DBSF is above the anticipated dose range for DBSF (5 mg to 12.5 mg)
- TEAEs are summarized by relationship to study treatment and severity in **Table 7**
 - Most TEAEs were judged as possibly or probably related to study drug: 40 events were considered probably related to DBSF 15 mg and ≤20 events probably related to any dose of DRG
 - The most frequently reported TEAEs considered probably related to study medication were somnolence (63 TEAEs overall), followed by "feeling of relaxation" (7 TEAEs overall)
 - Most TEAEs were of mild severity
- One subject experienced a serious AE (SAE) of "depressed level of consciousness" associated with bradycardia classified as probably related to study medication, emerging approximately 2 hours after DBSF 15 mg. The subject was kept overnight in hospital for observation
 - The subject's PK exposure (C_{max} and T_{max}) were similar to other subjects following DBSF 15 mg. CYP2C19 genotype was normal, and a toxic screen of plasma at the time of the event revealed no evidence of other drugs. This event was attributed to an unusual sensitivity to diazepam unrelated to the DBSF formulation
- 2 subjects discontinued due to an AE (SAE of depressed level of consciousness of severe intensity, and TEAE of somnolence of severe intensity) following DBSF 15 mg
- The majority of TEAEs resolved spontaneously by the end of the study
- With the exception of one subject who exhibited an unusual sensitivity to diazepam no safety issues were observed for vital signs or clinical laboratory or results

TABLE 5: SUMMARY OF ADVERSE EVENTS

	DBSF 15 mg	DRG 5 mg	DRG 12.5 mg	DRG 20 mg	Overall
Subjects dosed, N	33	33	32	31	36
Subjects with ≥1 TEAE, n (%)	30 (90.9)	16 (48.5)	17 (53.1)	24 (77.4)	34 (94.4)
TEAEs, n	52	25	22	32	131
Severe TEAEs, n	2	0	0	0	2
Serious TEAEs, n	1	0	0	0	1
Discontinuations due to TEAEs, n	2	0	0	0	2
Deaths, n	0	0	0	0	0
Most frequent adverse events, n of patients (%)					
Somnolence	28 (84.8)	11 (33.3)	17 (53.1)	21 (67.7)	34 (94.4)
Dizziness	2 (6.1)	0	0	0	2 (5.6)
Feeling of relaxation	7 (21.2)	0	1 (3.1)	0	8 (22.2)
Hiccups	3 (9.1)	0	0	0	3 (8.3)

DBSF, diazepam buccal soluble film; DRG, diazepam rectal gel

TABLE 6: FREQUENCY OF TEAES BY RELATIONSHIP AND SEVERITY (N TEAES)

	Causality: Unrelated	Unlikely	Possible	Probable	Severity: Mild	Moderate	Severe
DBSF 15 mg	1	8	3	40	44	6	2
DRG 5 mg	2	10	2	11	25	0	0
DRG 12.5 mg	2	2	3	15	21	1	0
DRG 20 mg	1	8	3	20	31	1	0
Overall	6	28	11	86	121	8	2

TEAE, treatment-emergent adverse event; DBSF, diazepam buccal soluble film; DRG, diazepam rectal gel

CONCLUSIONS

- Pharmacokinetics of DBSF and DRG are distinct
 - For DBSF, C_{max} is dose-proportional
 - For DRG, C_{max} is less than dose-proportional
 - Both DBSF and DRG are dose-proportional for AUC
- The difference in PK implies that the safe and effective dose range for of DBSF will be lower than the dose range recommended for DRG
- PK parameters following DRG exhibit consistently greater variability than DBSF
- Diazepam was safe and generally well tolerated in healthy adults following single doses of DBSF 15 mg and DRG 5 mg, 12.5 mg, and 20 mg

REFERENCES

1. Tatum WO. *Epilepsy Behav.* 2002;3:535-538.
2. Heller AH, Wargacki S, Jung C, Wyatt J, Schobel M. Safety and Pharmacokinetics of Diazepam Buccal Soluble Film. Poster presented at: 2018 AAN Annual Meeting; April 21-27, 2018; Los Angeles, CA.

ACKNOWLEDGEMENTS

Medical writing support was provided by Nate Connors, PhD, of ACCESS Medical, LLC; medical writing support was funded by Aquestive Therapeutics.

FUNDING AND DISCLOSURES

Financial support for this study was provided by Aquestive Therapeutics. Aquestive Therapeutics participated in the interpretation of data, review, and approval of the poster. All authors contributed to the development of the poster and maintained control over the final content. Allen H. Heller and David J. Wyatt are consultants to Aquestive Therapeutics. Stephen Wargacki, Tracy J. Stalvey, and A. Mark Schobel are employees of Aquestive Therapeutics.