The Usability of Diazepam Buccal Film as an Oral Rescue Treatment in Adult Patients with Epilepsy

Cassie Jung[1], Jordan Dubow[2], Hui Gong[3], Kore Liow[4], Sami Aboumatar[5], Pavel Klein[6], Michael A. Gelfand[7], Stephen Wargacki[1], Rikin Mehta[1], Michael A. Rogawski[8], Allen H. Heller[9]

[1]Aquestive Therapeutics Inc, Warren, NJ, USA; [2]Clinical and Medical Affairs, Clintrex; [3]Epilepsy Monitoring Unit, Rancho Los Amigos National Rehabilitation Center, Downey, CA, USA; [4]Department of Medicine (Neurology), University of Hawaii John Burns School of Medicine, Honolulu, HI, USA; [5]Austin Epilepsy Care Center, Austin, TX, USA; [6]Mid-Atlantic Epilepsy and Sleep Center, Bethesda, MD, USA; [7]Department of Neurology, University of Pennsylvania, Philadelphia, PA, USA; [8]Departments of Neurology and Pharmacology, School of Medicine, University of California, Davis, Sacramento, CA, USA; [9]Pharma Study Design LLC, Woodbridge, CT, USA

BACKGROUND

- Seizure clusters, also termed acute repetitive seizures, commonly occur in patients with drug refractory epilepsy[1]
- Benzodiazepines such as diazepam are the mainstay of rescue therapy for seizure clusters[1]
- Diazepam buccal film (DBF; formerly DBSF) is a novel dosage form of diazepam under development as an alternative approach for the management of seizure clusters
- The intended target population is selected patients with refractory epilepsy who require intermittent use of diazepam to control episodes of increased seizure activity
- DBF has the potential to be the first orally administered treatment for this indication and would provide an alternative to rectal diazepam gel
- DBF is administered by placing the film against the inner aspect of the cheek, where it adheres, dissolves, and releases drug onto the buccal mucosa
- This phase 2 study assessed the safety, pharmacokinetics (PK) and usability of DBF in adults with epilepsy under interictal and ictal/periictal conditions. This poster focuses on usability. A separate poster reports the PK results (Rogawski MA, Gong H, Liow K, Aboumatar S, Klein P, Gelfand MA, Jung J, Stephen Wargacki S, Mehta R and Heller AH. Pharmacokinetics of diazepam buccal film in adult patients with epilepsy: Comparison of bioavailability with periictal and interictal administration, *AES Abstracts* 2018). The safety results reported here are taken from this report.

OBJECTIVE

- To assess the usability of DBF in adult patients with epilepsy under interictal (Treatment A) and ictal/periictal (Treatment B) conditions

METHODS

STUDY SUBJECTS

- Adult men and women ages 17-65 years with poorly controlled tonic-clonic seizures or focal seizures with impaired awareness (N=35)
- Subjects had on average ≥1 seizure every 3 days or ≥10 seizures/month
- The subjects were receiving at least one antiseizure drug
- Baseline medications were adjusted according to clinical judgement
- Eligibility was established at a screening visit within 28 days of the first study drug administration

STUDY DESIGN

- This was a single-dose crossover study in which subjects were investigated while undergoing a clinical epilepsy monitoring unit (EMU) evaluation (Treatment B) and on another PK-only visit (Treatment A), with the visits separated by in most cases ≥3 weeks (but not <14 days)
- Treatment A and Treatment B could occur in either order
- During Treatment A, DBF 12.5 mg was administered when no seizure activity had been observed in the preceding 3 hours ("interictal")
- During Treatment B, DBF 12.5 mg was administered during a clinically observed seizure or within 5 minutes of cessation of a seizure ("ictal/periictal")

- Usability endpoints for investigator placement of DBF included:
 1. Whether successful placement was achieved
 2. Whether more than one attempt was needed to successfully insert the film
 3. Whether the subject spit or blew out the film
 4. Whether DBF was swallowed

PROCEDURES

- DBF was administered by placing the film against the inner aspect of the cheek, where it adhered, dissolved and released drug onto the buccal mucosa

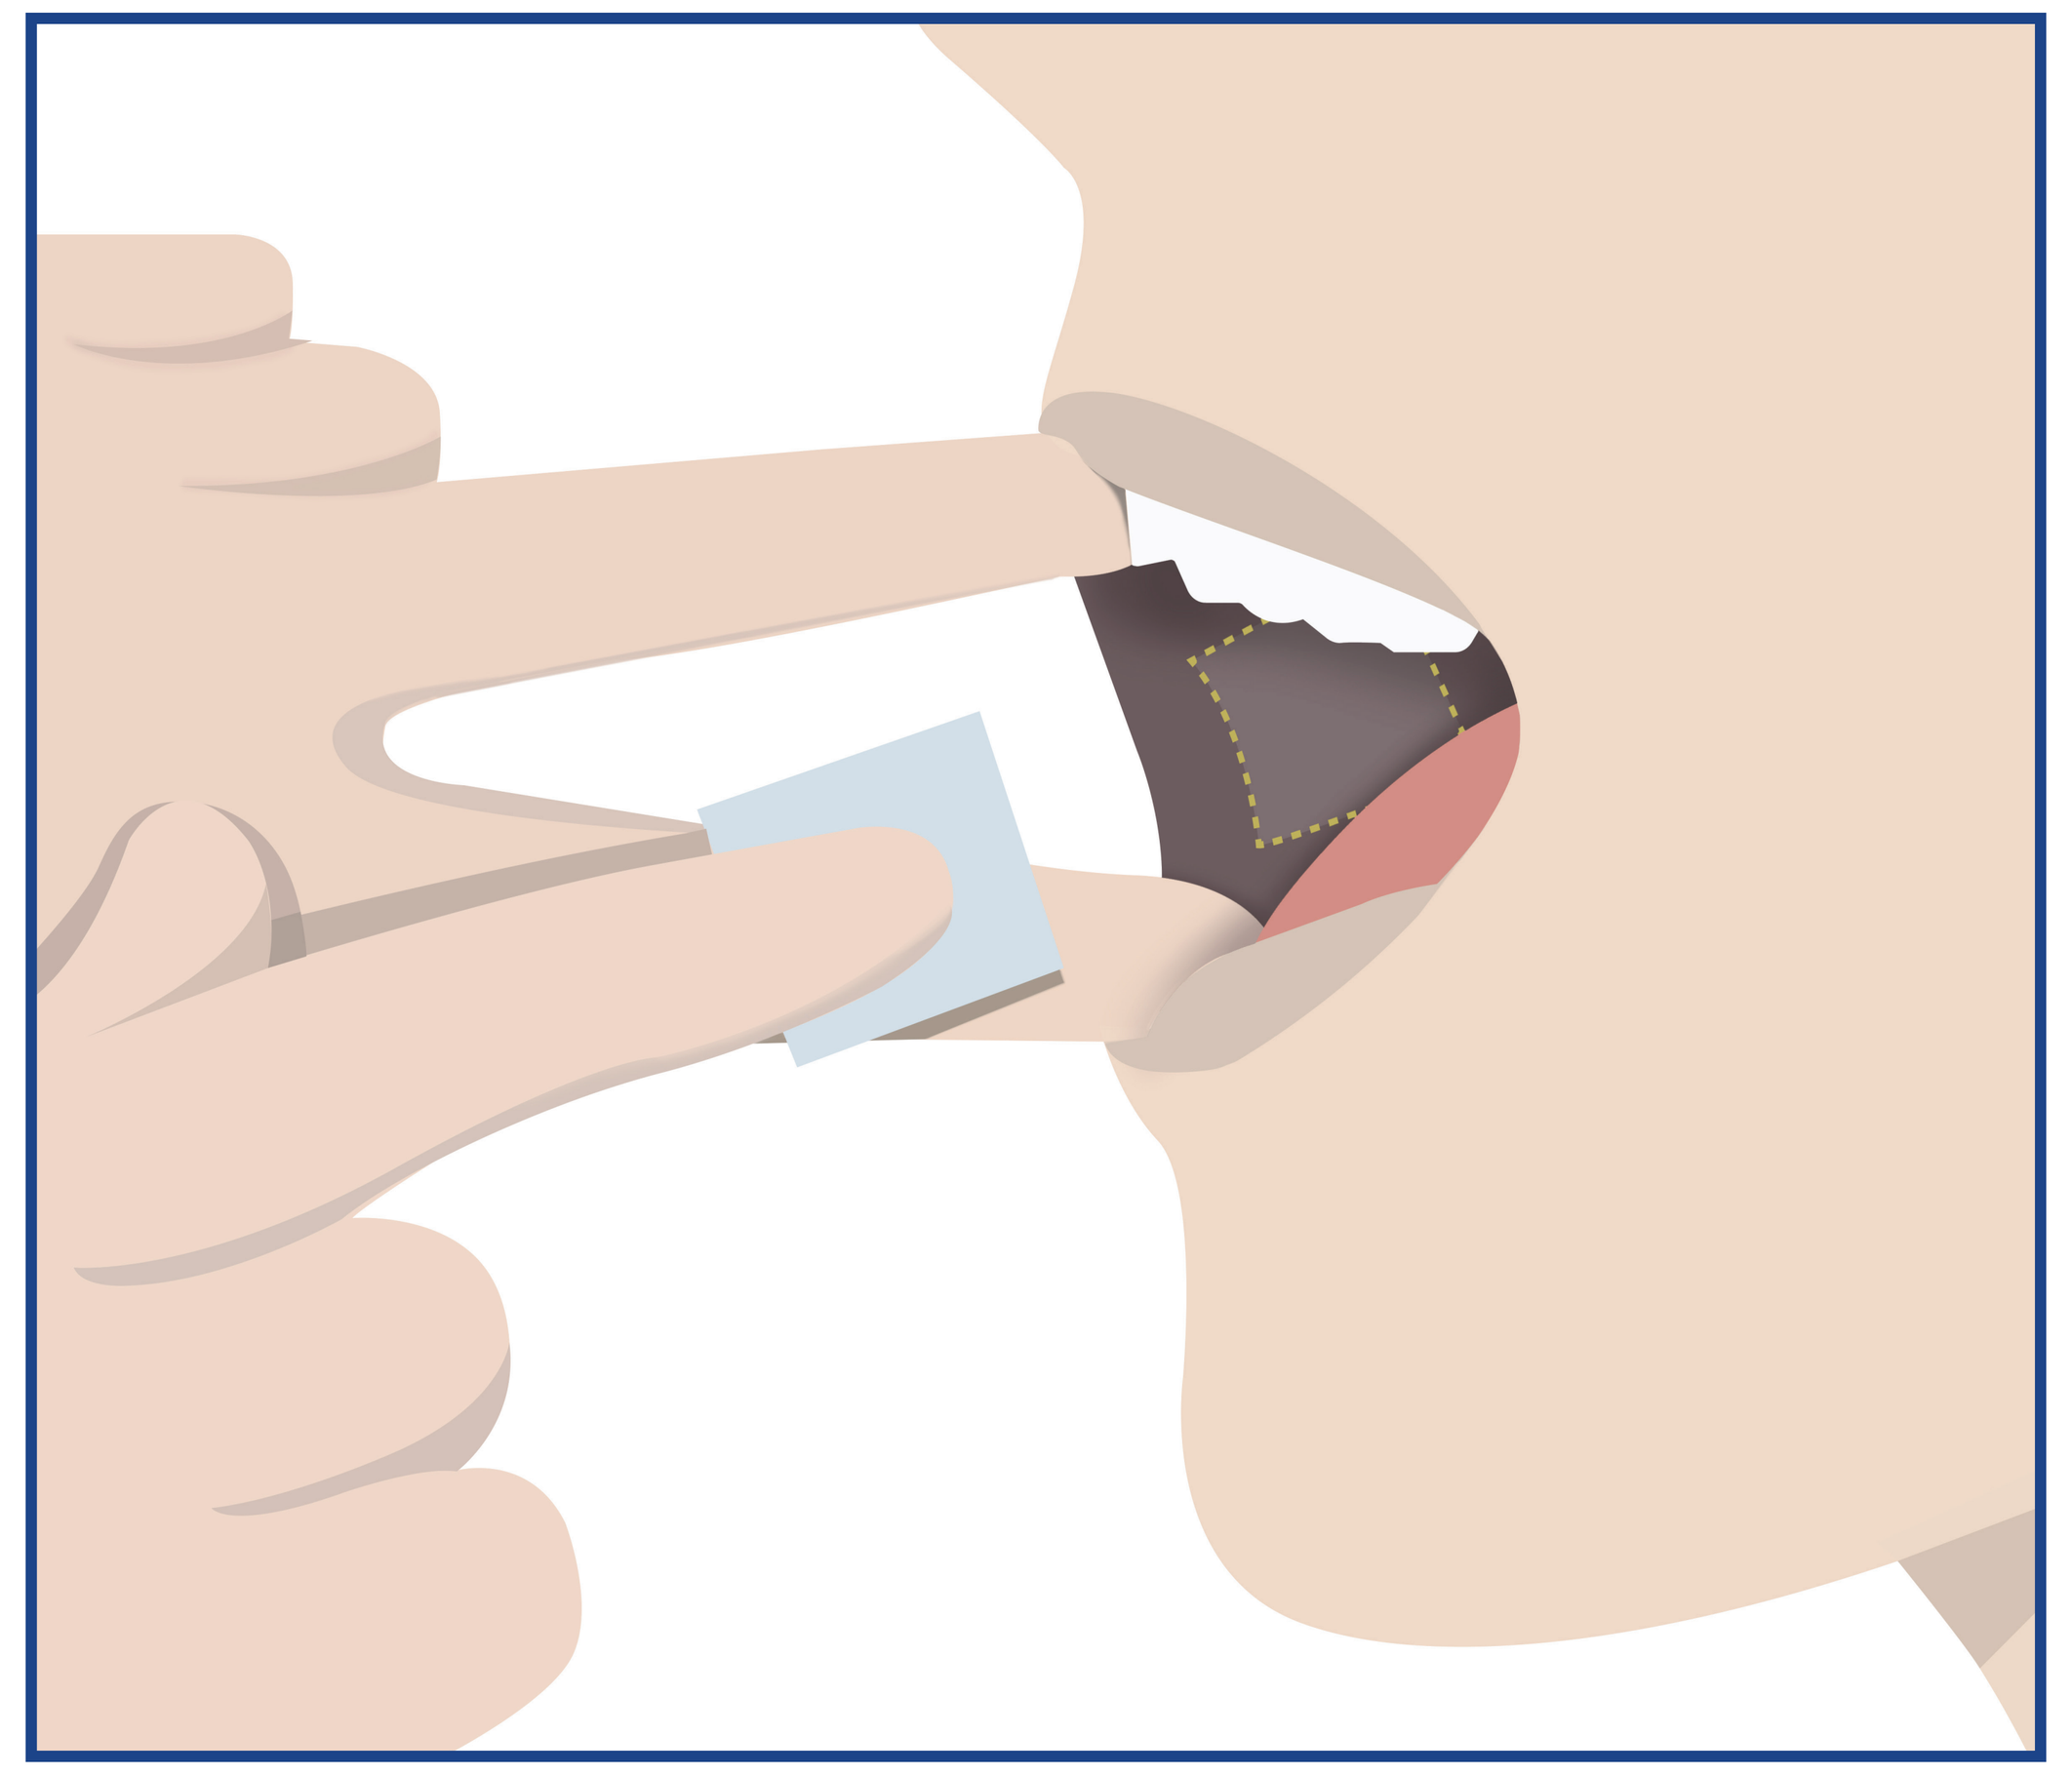

SAFETY AND TOLERABILITY

- Subjects were monitored for adverse events (AE) throughout the study

RESULTS

SUBJECTS

- A total of 35 subjects received at least one dose of study drug and comprised the safety population. The demographic characteristics of these subjects is summarized in **Table 1**

TABLE 1: SUMMARY OF DEMOGRAPHIC CHARACTERISTICS OF SUBJECTS INCLUDED IN THE SAFETY POPULATION

Demographic Characteristic	Overall (N=35)
Age (years), mean ± SD	32.8 ± 11.1
Age group (years), n (%)	
< 18 years	0
18-40 years	29 (82.9)
41-65 years	6 (17.1)
> 65 years	0
Sex, n (%)	
Female	18 (51.4)
Male	17 (48.6)
Ethnicity, n (%)	
Not Hispanic or Latino	26 (74.3)
Hispanic or Latino	9 (25.7)
Race, n (%)	
White	19 (54.3)
Black	5 (14.3)
Asian	1 (2.9)
Am Indian	2 (5.7)
Hawaiian	6 (17.1)
Other	2 (5.7)
Height (cm), mean ± SD	165.64 ± 10.81
Weight (kg), mean ± SD	76.24 ± 19.58
BMI (kg/m²), mean ± SD	27.84 ± 7.01

Am Indian: American Indian or Alaskan Native; Black: Black or African American; BMI: body mass index; Hawaiian: Native Hawaiian or Pacific Islander; SD: standard deviation

USABILITY OUTCOMES

- Results from usability endpoints for investigator placement of DBF are summarized in **Table 2**

TABLE 2: USABILITY ENDPOINTS FOR DBF DURING INTERICTAL (TREATMENT A) AND ICTAL/PERIICTAL STATES (TREATMENT B)

Outcome	Treatment A (n=33)	Treatment B (n=33)
Successful placement of film, n (%)	33 (100%)	33 (100%)
Number of subjects requiring >1 film placement attempt, n (%)	0	2 (6.1%)
Spit or blew out film, n (%)	0	3 (9.1%)
Swallowed film, n (%)	2 (6.1%)	1 (3.0%)

SAFETY AND TOLERABILITY

- Majority of the treatment-emergent adverse events (TEAE) in this study were classified as unrelated to study drug
- The most common adverse events that were classified as probably related to study drug were somnolence and dizziness and were relatively infrequent (overall frequency of 5.7% of subjects and 2.9% of subjects, respectively)
- Frequency of patients experiencing TEAEs considered possibly or probably related to study drug and number of events are summarized in **Table 3**

TABLE 3: FREQUENCY OF PATIENTS EXPERIENCING TREATMENT-EMERGENT ADVERSE EVENTS CONSIDERED POSSIBLY OR PROBABLY RELATED TO STUDY DRUG AND NUMBER OF EVENTS - SAFETY POPULATION

Adverse Events– MedDRA® Preferred Term n (%) E	Period A (N=33)	Period B (N=33)	Overall (N=35)
Somnolence	2 (6.1) 3	0	2 (5.7) 3
Dizziness	1 (3.0) 1	0	1 (2.9) 1
Hypoaesthesia	1 (3.0) 1	0	1 (2.9) 1
Nausea	0	1 (3.0) 1	1 (2.9) 1
Paraesthesia oral	1 (3.0) 1	1 (3.0) 1	1 (2.9) 2

E: Number of treatment-emergent adverse events; N: Number of patients dosed; n (%): Number and percent of patients with treatment-emergent adverse event
MedDRA®: Medical Dictionary for Regulatory Activities, Version 20.0
Each patient could only contribute once to each of the incidence rates, regardless of the number of occurrences
Overall: Included results from all treatment periods

- Somnolence was rated as severe in one subject following Treatment A
- There were no serious AEs related to study drug and no patient withdrew because of an AE
- There was 1 event of angina pectoris considered unrelated to study drug and 1 event of tachycardia considered unlikely to be related to study drug

CONCLUSIONS

- In the EMU setting, DBF was successfully placed and generally used without difficulty in both interictal and ictal/periictal states
- DBF is safe and well tolerated
- DBF is a practical oral treatment for adults with refractory epilepsy who require intermittent use of diazepam to control episodes of increased seizure activity

ACKNOWLEDGEMENTS

Support in preparing this poster was provided by Nate Connors, PhD, and Farida Khan, PhD, of ACCESS Medical, LLC, and funded by Aquestive Therapeutics.

FUNDING AND DISCLOSURES

Financial support for this study was provided by Aquestive Therapeutics. All authors contributed to the development of the poster and maintained control over the final content. Allen H. Heller and Michael A. Rogawski are consultants to Aquestive Therapeutics. Cassie Jung, Stephen Wargacki, and Rikin Mehta are employees of Aquestive Therapeutics.

REFERENCE

1. Haut SR. Seizure clusters: characteristics and treatment. *Curr Opin Neurol.* 2015;28(2):143-150.

Aquestive