Pharmacokinetics of Diazepam Buccal Film in Adult Patients with Epilepsy: Comparison of Bioavailability with Periictal and Interictal Administration

Michael A. Rogawski[1], Hui Gong[2], Kore Liow[3], Sami Aboumatar[4], Pavel Klein[5], Michael A. Gelfand[6], Cassie Jung[7], Stephen Wargacki[7], Rikin Mehta[7] and Allen H. Heller[8]

[1]Departments of Neurology and Pharmacology, School of Medicine, University of California, Davis, Sacramento, CA, USA; [2]Epilepsy Monitoring Unit, Rancho Los Amigos National Rehabilitation Center, Downey, CA, USA; [3]Department of Medicine (Neurology), University of Hawaii John Burns School of Medicine, Honolulu, HI, USA; [4]Austin Epilepsy Care Center, Austin, TX, USA; [5]Mid-Atlantic Epilepsy and Sleep Center, Bethesda, MD, USA; [6]Department of Neurology, University of Pennsylvania, Philadelphia, PA, USA; [7]Aquestive Therapeutics Inc, Warren, NJ, USA; [8]Pharma Study Design LLC, Woodbridge, CT, USA

BACKGROUND

- Seizure clusters, also termed acute repetitive seizures, commonly occur in patients with drug refractory epilepsy[1]
- Seizure clustering is associated with reduced quality of life, may be a risk factor for less satisfactory long-term outcomes for people with epilepsy, and may devolve to status epilepticus, a life threatening medical emergency[1]
- Benzodiazepines such as diazepam are the mainstay of rescue therapy for seizure clusters[1]
- Diazepam buccal film (DBF; formerly DBSF) is a novel dosage form of diazepam under development as an alternative approach to the management of seizure clusters
- DBF is expected to have advantages in relation to other diazepam dosage forms for acute treatment in terms of patient and caregiver acceptance, portability, and reliability
- DBF is a thin soluble mucoadhesive polymer matrix that incorporates diazepam, distributed in a uniform fashion, as well as permeation enhancers and other excipients
- The film adheres to the inner cheek and delivers diazepam across the buccal mucosa and also by the enteric route
- The intended target population is selected patients with refractory epilepsy who require intermittent use of diazepam to control episodes of increased seizure activity
- During a seizure, changes might occur in the oral environment that could affect diazepam absorption. This study sought to assess whether such changes, if any, are associated with alterations in the absorption of DBF-delivered diazepam

OBJECTIVES

PRIMARY OBJECTIVE

- Compare the bioavailability of diazepam in adult patients with epilepsy following single doses of 12.5 mg DBF administered under interictal and ictal/periictal conditions by comparison of pharmacokinetic (PK) parameters derived from blood level measurements obtained at various times after dosing

SECONDARY OBJECTIVES

- Evaluate the safety and tolerability of single doses of DBF 12.5 mg under interictal and ictal/periictal conditions
- Evaluate the usability of single doses of DBF 12.5 mg under interictal and ictal/periictal conditions

METHODS

STUDY SUBJECTS

- Adult men and women ages 17-65 years with poorly controlled tonic-clonic seizures or focal seizures with impaired awareness (N=35)
- Subjects had on average ≥1 seizure every 3 days or ≥10 seizures / month
- The subjects were receiving at least one antiseizure drug
- Baseline medications were adjusted according to clinical judgement
- Eligibility was established at a screening visit within 28 days of the first study drug administration

STUDY DESIGN

- This was a single-dose crossover study in which subjects were investigated while undergoing a clinical epilepsy monitoring unit (EMU) evaluation (Treatment B) and on another PK-only visit (Treatment A), with the visits separated by in most cases ≥3 weeks (but not <14 days)
- Treatment A and Treatment B could occur in either order
- During Treatment A, DBF 12.5 mg was administered when no seizure activity had been observed in the preceding 3 hours ("interictal")
- During Treatment B, DBF 12.5 mg was administered during a clinically observed seizure or within 5 minutes of cessation of a seizure ("ictal/periictal")
- In Treatment A, subjects were confined on the day of treatment for at least 8 hours after DBF dosing. Subjects returned to the clinic for later blood draws
- In Treatment B, subjects remained in the EMU for at least 4 hours after treatment

PHARMACOKINETIC SAMPLING AND ANALYSIS

- Plasma samples for diazepam were drawn pre-dose and post-dose at 0.25, 0.5, 1, 1.5, 2, and 4 hours following both treatments. Additional samples were drawn at 8, 24, 48, 96, 144, 192, and 240 hours following Treatment A
- Plasma was analyzed for diazepam by an HPLC method using tandem mass spectroscopy and automated extraction for diazepam (1.00 to 1000 ng/mL) in human EDTA K2 plasma
- Maximal plasma concentration (C_{max}), time to maximal concentration (T_{max}), and partial area under the curve (partial AUC) at 2 or 4 hours were calculated

SAFETY AND TOLERABILITY

- Patients were monitored for adverse events (AE) throughout the study

STATISTICAL ANALYSIS

Summary Statistics

- PK parameters were calculated and tabulated by treatment group with summary statistics

Comparison of Treatment A and Treatment B

- Diazepam exposure following Treatment B was compared with exposure following Treatment A. Pairwise comparisons within subject were evaluated by analysis of variance (ANOVA) with treatment as a fixed factor and subject as a random factor on the In-transformed $AUC_{(0\text{-}4h)}$, $AUC_{(0\text{-}2h)}$, and C_{max}
- T_{max} was compared using the non-parametric Wilcoxon signed-rank test

RESULTS

SUBJECTS

- A total of 35 subjects received at least one dose of study drug and comprised the safety population. The demographic characteristics of these subjects is summarized in **Table 1**

TABLE 1: SUMMARY OF DEMOGRAPHIC CHARACTERISTICS OF SUBJECTS INCLUDED IN THE SAFETY POPULATION

Demographic Characteristic	Overall (N=35)
Age (years), mean ± SD	32.8 ± 11.1
Age group (years), n (%)	
< 18 years	0
18-40 years	29 (82.9)
41-65 years	6 (17.1)
> 65 years	0
Sex, n (%)	
Female	18 (51.4)
Male	17 (48.6)
Ethnicity, n (%)	
Not Hispanic or Latino	26 (74.3)
Hispanic or Latino	9 (25.7)
Race, n (%)	
White	19 (54.3)
Black	5 (14.3)
Asian	1 (2.9)
Am Indian	2 (5.7)
Hawaiian	6 (17.1)
Other	2 (5.7)
Height (cm), mean ± SD	165.64 ± 10.81
Weight (kg), mean ± SD	76.24 ± 19.58
BMI (kg/m²), mean ± SD	27.84 ± 7.01

Am Indian: American Indian or Alaskan Native; Black: Black or African American; BMI: body mass index; Hawaiian: Native Hawaiian or Pacific Islander; SD: standard deviation

COMPARISON OF PHARMACOKINETIC PARAMETERS FOLLOWING ADMINISTRATION IN THE INTERICTAL AND ICTAL/PERIICTAL STATES

- PK parameters obtained from blood draws following Treatment A and Treatment B are summarized in **Tables 2** and **3**
- Patients were excluded from analysis if both treatments were not completed (n=4), critical PK time points were missed as adjudicated by an expert blinded to the concentration data (n=3), pre-dose diazepam concentrations were >5% of the subsequent C_{max} (n=2), or DBF was administered in a manner contrary to instructions (n=5)
- PK profiles valid for analysis until 2 hours after dosing in both treatment conditions were available for 21 patients. The results are shown in **Table 2**. There were no statistical differences between the geometric mean C_{max}, AUC, or T_{max} values in the two treatment conditions

TABLE 2: PHARMACOKINETIC PARAMETERS 2 HOURS FOLLOWING DBF 12.5 MG IN THE INTERICTAL AND ICTAL/PERIICTAL STATE

2-hour Pharmacokinetic Profiles (n=21)				
	A. Interictal	B. Ictal/periictal	Ratio B/A (%)[1]	90% CI (%)[2]
C_{max} (ng/mL), geo. mean	198.76	184.40	92.77	74.5 – 115.53
$AUC_{(0\text{-}2h)}$ (ng.h/mL), geo. mean	264.57	248.06	93.76	73.86 – 119.02
T_{max} (h), median	0.680	0.983	*	

[1]Ratio of geometric means
[2]90% geometric confidence interval using ln-transformed data
**Difference in T_{max} not significant, p = 0.8620 (Wilcoxon signed-rank test)*
AUC, area under the plasma concentration-time curve; C_{max}, maximum plasma drug concentration; CI, confidence interval; T_{max}, time to reach maximum plasma concentration

- PK profiles valid for analysis until 4 hours after dosing for both treatment conditions were available for 18 of the 21 subjects included in the analysis of Table 2. The PK parameters from these 18 subjects are provided in **Table 3**
- As was the case for the analysis presented in Table 2, there were no statistical differences between the geometric mean C_{max}, AUC, or T_{max} values in the two treatment conditions

TABLE 3: PHARMACOKINETIC PARAMETERS 4 HOURS FOLLOWING DBF 12.5 MG IN THE INTERICTAL AND ICTAL/PERIICTAL STATE

4-hour Pharmacokinetic Profiles (n=18)				
	A. Interictal	B. Ictal/periictal	Ratio B/A (%)[1]	90% CI (%)[2]
C_{max} (ng/mL), geo. mean	190.34	179.96	95.54	73.34 -121.89
$AUC_{(0\text{-}4h)}$ (ng.h/mL), geo. mean	483.79	433.32	89.57	69.21 -115.91
T_{max} (h), median	0.767	0.533	*	

[1]Ratio of geometric means
[2]90% geometric confidence interval using ln-transformed data
**Difference in T_{max} not significant, p=0.5708 (Wilcoxon signed-rank test)*
AUC, area under the plasma concentration-time curve; C_{max}, maximum plasma drug concentration; CI, confidence interval; T_{max}, time to reach maximum plasma concentration

- Mean plasma concentration-time profiles by treatment are shown in **Figure 1**

FIGURE 1: MEAN DIAZEPAM PLASMA CONCENTRATION FOLLOWING ADMINISTRATION OF DBF 12.5 MG IN THE INTERICTAL OR ICTAL/PERIICTAL STATE (N=16-21)*

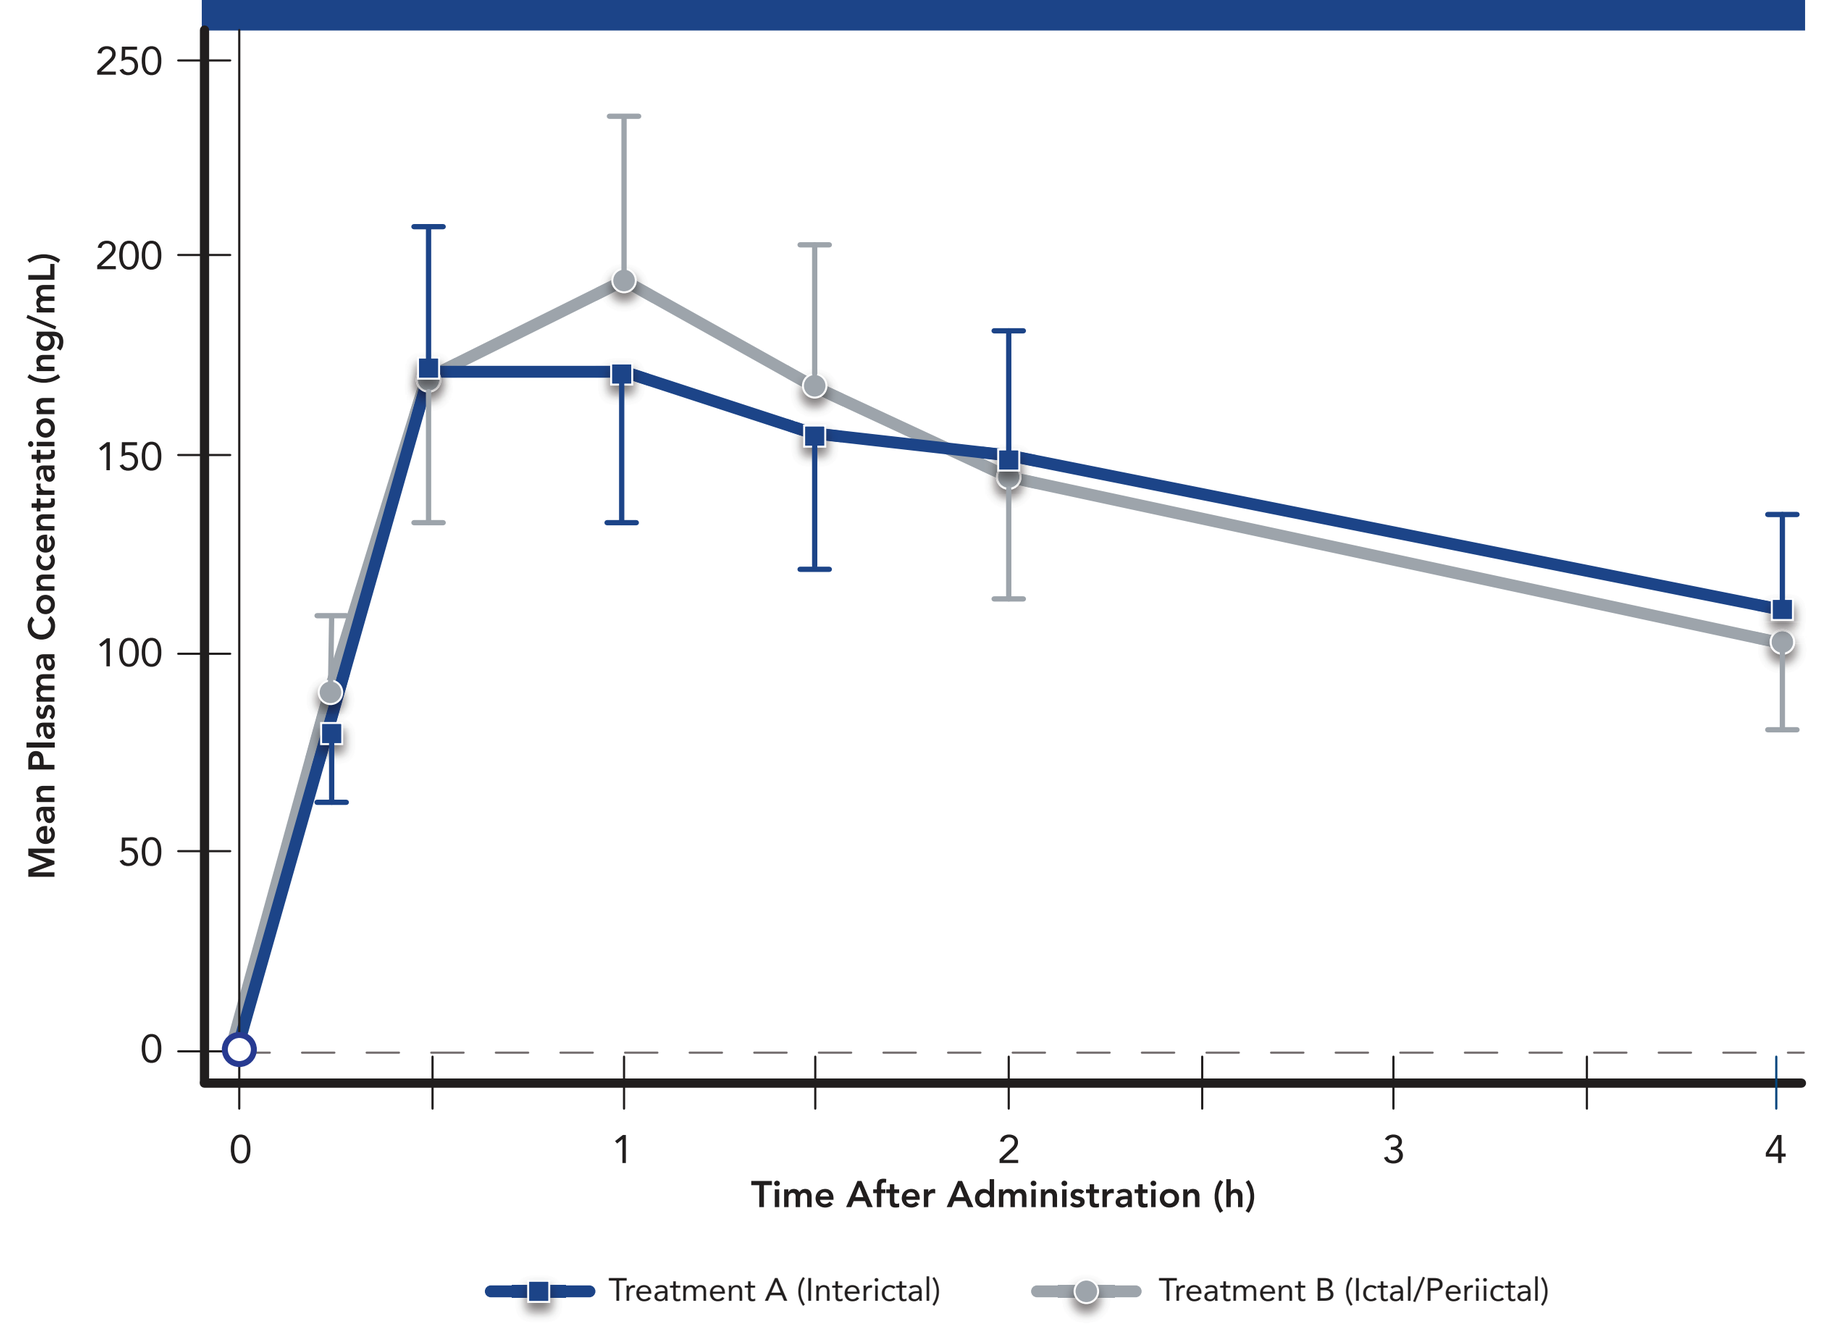
Mean Plasma Concentration (ng/mL)
250
200
150
100
50
0
0
1
2
3
4
Time After Administration (h)
Treatment A (Interictal)
Treatment B (Ictal/Periictal)

**Of 21 subjects with valid profiles for both Treatment A and Treatment B, all available data are included. Each timepoint is the mean of concentration data from 16-21 subjects. Error bars are standard error of the mean.*

SAFETY AND TOLERABILITY

- Majority of the treatment-emergent adverse events (TEAE) in this study were classified as unrelated to study drug
- The most common adverse events that were classified as probably related to study drug were somnolence and dizziness and were relatively infrequent (overall frequency of 5.7% of subjects and 2.9% of subjects, respectively)
- Frequency of patients experiencing TEAEs considered possibly or probably related to study drug and number of events are summarized in **Table 4**

TABLE 4: FREQUENCY OF PATIENTS EXPERIENCING TREATMENT-EMERGENT ADVERSE EVENTS CONSIDERED POSSIBLY OR PROBABLY RELATED TO STUDY DRUG AND NUMBER OF EVENTS - SAFETY POPULATION

Adverse Events–MedDRA® Preferred Term n (%) E	Period A (N=33)	Period B (N=33)	Overall (N=35)
Somnolence	2 (6.1) 3	0	2 (5.7) 3
Dizziness	1 (3.0) 1	0	1 (2.9) 1
Hypoaesthesia	1 (3.0) 1	0	1 (2.9) 1
Nausea	0	1 (3.0) 1	1 (2.9) 1
Paraesthesia oral	1 (3.0) 1	1 (3.0) 1	1 (2.9) 2

E: Number of treatment-emergent adverse events; N: Number of patients dosed; n (%): Number and percent of patients with treatment-emergent adverse event
MedDRA®: Medical Dictionary for Regulatory Activities, Version 20.0
Each patient could only contribute once to each of the incidence rates, regardless of the number of occurrences
Overall: Included results from all treatment periods

- Somnolence was rated as severe in one subject following Treatment A
- There were no serious AEs related to study drug and no patient withdrew because of an AE
- There was 1 event of angina pectoris considered unrelated to study drug and 1 event of tachycardia considered unlikely to be related to study drug

CONCLUSIONS

- A single dose of DBF 12.5 mg administered to adults with epilepsy provides exposure to diazepam under ictal/periictal conditions that is comparable to that obtained under interictal conditions
- DBF 12.5 mg is safe and well tolerated

ACKNOWLEDGEMENTS

Assistance in preparing this poster was provided by Nate Connors, PhD, and Farida Khan, PhD, of ACCESS Medical, LLC and funded by Aquestive Therapeutics.

FUNDING AND DISCLOSURES

Financial support for this study was provided by Aquestive Therapeutics. All authors contributed to the development of the poster and maintained control over the content. Michael A. Rogawski, Allen H. Heller and David J. Wyatt are consultants to Aquestive Therapeutics. Cassie Jung, Stephen Wargacki, and Rikin Mehta are employees of Aquestive Therapeutics.

REFERENCE

1. Haut SR. Seizure clusters: characteristics and treatment. *Curr Opin Neurol*. 2015;28(2):143-150.

Aquestive