POSTER 27

PHARMACOKINETICS OF CLOBAZAM ORAL SOLUBLE FILM

Allen H. Heller[1], Stephen Wargacki[2], Cassie Jung[2], David J. Wyatt[3], A. Mark Schobel[2]

[1]Pharma Study Design, LLC, Woodbridge, CT 06525, USA; [2]Aquestive Therapeutics, Warren, NJ 07059, USA; [3]Syneos Health Clinical Solutions Early Phase, Miami, FL 33131, USA

ABSTRACT

Clobazam oral soluble film (COSF) is a novel dosage form under development for adjunctive treatment of seizures in Lennox-Gastaut syndrome. We assessed the pharmacokinetics (PK) and safety-tolerability of clobazam administered as single doses of COSF 20 and 10 mg (Aquestive Therapeutics) compared with clobazam tablets 20 and 10 mg (Lundbeck, US). Healthy adult volunteers (N=51) were enrolled in a single-dose, open-label, randomized four-sequence, four-period, crossover: (A) COSF 20 mg; (B) clobazam tablet 20 mg; (C) COSF 10 mg; and (D) clobazam tablet 10 mg. PK sampling for clobazam and N-desmethylclobazam was carried out until 21 days post-dose with a 28-day washout. Subjects were monitored for adverse events (AE) throughout the study. COSF at single doses of 10 and 20 mg was bioequivalent to the clobazam tablet at equivalent doses for both clobazam and N-desmethylclobazam, its active metabolite. The PK of both formulations were dose-proportional. AE were dose-related across the treatment groups, with somnolence the most common. Most events were mild and none serious or severe. COSF is a novel clobazam dosage form that is bioequivalent to the clobazam tablet. Because of its ease of administration, COSF may be expected to improve adherence, reduce likelihood of error, and provide accurate dosing compared with formulations of clobazam that are currently available.

BACKGROUND

CLOBAZAM FOR ADJUNCTIVE TREATMENT OF SEIZURES IN LENNOX-GASTAUT SYNDROME (LGS)

- Two dosage forms of clobazam (tablets and oral suspension; ONFI®, Lundbeck, Inc., Deerfield, IL, USA) are FDA-approved for the adjunctive treatment of seizures associated with LGS in patients two years of age or older.
- In short- and long-term clinical trials, adjunctive treatment with clobazam significantly reduces the frequency of both drop seizures and total seizures, and improves patient symptoms as measured by physician and caregiver evaluations.[1,2]
- Administration of current clobazam dosage forms to patients with LGS may be problematic in the setting of dysphagia, frequent seizures, cognitive/behavioral challenges, psychomotor delay, or neuropsychiatric disorders that may occur in these patients,[3-5] and dosage errors by caregivers are common in the administration of liquid formulations of medications.[6]

CLOBAZAM ORAL SOLUBLE FILM (COSF)

- COSF, developed using patented PharmFilm® technology (Aquestive Therapeutics, Warren, NJ, USA) is a rapidly dissolving oral soluble film currently under clinical development for the adjunctive treatment of seizures associated with LGS in patients two years of age or older.
- COSF, approximately the size of a postage stamp, is intended for one-step application to the surface of the tongue where it immediately adheres and rapidly dissolves, releasing clobazam into the saliva to be swallowed naturally, allowing for gastrointestinal absorption.
- This innovative, patient-friendly formulation may eliminate the need for the patient's active cooperation in swallowing.

OBJECTIVES

PRIMARY OBJECTIVES

- To assess the pharmacokinetics (PK) of clobazam administered as single doses of COSF 20 and 10 mg compared with clobazam tablets 20 and 10 mg in healthy adults
- To determine whether COSF is bioequivalent to clobazam tablets at the same nominal dose with respect to clobazam and its active metabolite, N-desmethylclobazam

SECONDARY OBJECTIVES

- To assess the dose-proportionality of single doses of clobazam 20 and 10 mg administered as either COSF or as a tablet
- To assess the safety and tolerability of single doses of COSF 20 and 10 mg

METHODS

SUBJECTS

- Healthy adults, 18-64 years of age
- Nonsmokers for at least six months prior to screening, body mass index (BMI) 18.6-29.9 kg/m^2, able to give written informed consent, and able to communicate effectively with clinic staff; women of child-bearing potential were required to avoid pregnancy by complete abstinence or medically acceptable contraception.
- Exclusion criteria — History of psychiatric symptoms, drug abuse, or alcohol abuse within the previous year, or evidence of any clinically significant organ system disease.
- The study protocol and the consent form were approved by the Chesapeake Institutional Review Board, Columbia, MD, USA.

STUDY DESIGN

- Single-dose, open-label, randomized four-sequence, four-period, four-treatment crossover under fasting conditions. Eligible subjects were randomly assigned to one of four dosing sequences according to a Williams design. Washout between doses was 28 days.

Figure 1. Study Design

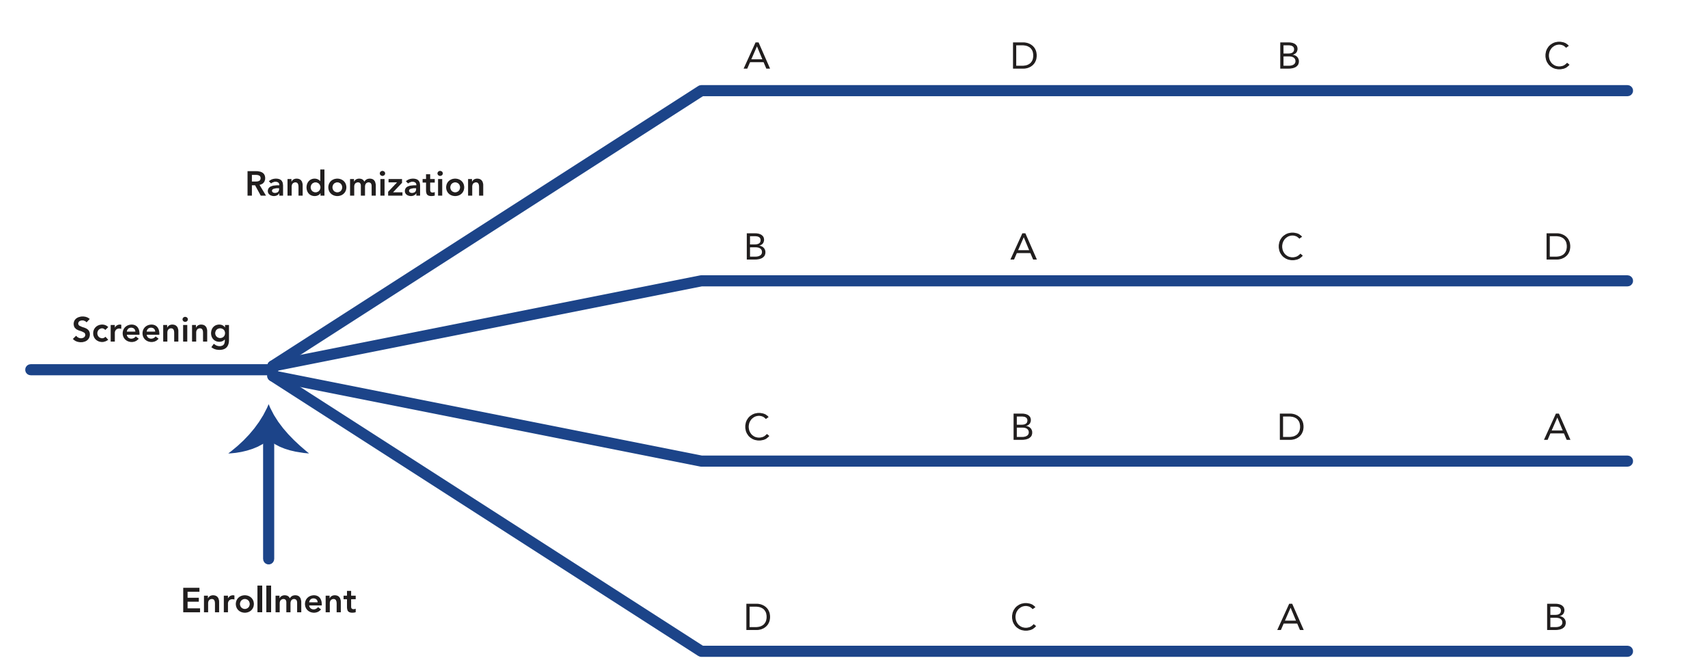
Treatment Sequences
Randomization
Screening
Enrollment
A D B C
B A C D
C B D A
D C A B

Treatment A=COSF 20 mg
Treatment B=clobazam tablet 20 mg
Treatment C=COSF 10 mg
Treatment D=clobazam tablet 10 mg

COSF: clobazam oral soluble film

- For each of the four periods, subjects were confined to the clinical research unit from at least 14 hours prior to drug administration on Day 1 until 36 hours post-dose, and subjects returned to the clinical facility for subsequent blood draws for PK sampling.

DOSING PROCEDURES

COSF

- Clinic staff placed the film centered directly on the dorsal surface of the tongue and instructed the subject to close his/her mouth in a natural way. Subjects were permitted to swallow saliva naturally but not to chew, bite, or swallow the film. COSF was administered without water or other liquid.

Clobazam Tablets

- Subjects were administered the assigned tablet with 240 mL water at ambient temperature. Subjects were instructed not to chew, bite, or break the tablet.

PHARMACOKINETICS

- Blood samples for determination of clobazam and N-desmethylclobazam were collected prior to drug administration and at 0.333, 0.667, 1.00, 1.50, 2.00, 2.50, 3.00, 3.50, 4.00, 6.00, 8.00, 10.0, 12.0, 24.0, 28.0, 36.0, 48.0, 72.0, 96.0, 144, 240, 360, and 504 hours post-dose.

Pharmacokinetic Analysis

- *C_{max} and T_{max} taken from the observed plasma concentration-time profile*
- *K_{el} estimated as the negative of the slope from linear regression from the ln-transformed plasma concentration-time profile in the terminal elimination phase*
- *$T_{½\,el}$ calculated as ln(2)/K_{el}*
- *AUC_{0-t} (area under the concentration-time curve from time zero until the last measurable concentration or last sampling time [t], whichever occurred first) calculated using the trapezoidal method*
- *AUC_{0-inf} calculated as AUC_{0-t} + C_{last}/K_{el}, in which C_{last} was the last measurable concentration*
- *CL/F calculated as dose/AUC_{0-inf}*
- *V_d/F estimated from CL/F/K_{el}*
- *Residual area calculated as 100*(1-AUC_{0-t} /AUC_{0-inf})*

SAFETY AND TOLERABILITY

- Before the study and at study completion subjects underwent a complete physical examination, electrocardiogram (ECG), and routine clinical laboratory tests.
- All subjects were monitored for adverse events (AEs) throughout the study, and vital signs were monitored at specified intervals throughout the period of confinement.
- Visual inspections of the administration site were performed prior to all administrations of COSF and at 0.167, 0.5, and 1 hour after administration to check for any sign of mucosal irritation.

STATISTICAL ANALYSIS

Average Bioequivalence

- Comparisons between treatment groups (A versus B and C versus D) were evaluated using analysis of variance (ANOVA) with sequence and treatment as fixed factors and subject (sequence) as a random factor for ln-transformed values of AUC_{0-t}, AUC_{0-inf}, C_{max}, CL/F, and V_d/F, and for the untransformed values of T_{max}, K_{el}, and $T_{1/2\,el}$.
- T_{max} was analyzed using an additional nonparametric test (Wilcoxon signed-rank test).
- Inter- and intrasubject coefficients of variation were estimated.
- The ratios of geometric means (treatments A/B and treatments C/D) and corresponding 90% confidence interval (CI) based on least-squares means (LSMs) from the ANOVA of the ln-transformed data were calculated for AUC_{0-t}, AUC_{0-inf}, and C_{max}.
- Criteria for average bioequivalence (90% CIs between 80% to 125% for AUC_{0-t}, AUC_{0-inf}, and C_{max}) were based on clobazam. Results for N-desmethylclobazam were analyzed for average bioequivalence as supportive data.

Dose-proportionality

- Clobazam and N-desmethylclobazam comparisons between treatment groups (treatments C versus A for COSF and treatments D versus B for clobazam tablets, respectively) were evaluated using ANOVA with sequence, treatment, and period as fixed factors and subject (sequence) as a random factor for the ln-transformed values of AUC_{0-t}, AUC_{0-inf}, and C_{max}, dose normalized to 20 mg.
- The ratios of geometric means (C/A and D/B) and corresponding 90% CIs based on LSMs from the ANOVA of the ln-transformed values for AUC_{0-t}, AUC_{0-inf}, and C_{max}, dose normalized to 20 mg, were calculated.
- Inter- and intrasubject coefficients of variation were estimated.

RESULTS

SUBJECTS

- 51 subjects were randomized to one of the four treatment sequences and comprised the safety population (all subjects who received at least one dose of clobazam); 45 subjects completed all treatment periods.

Figure 2. Subject Disposition

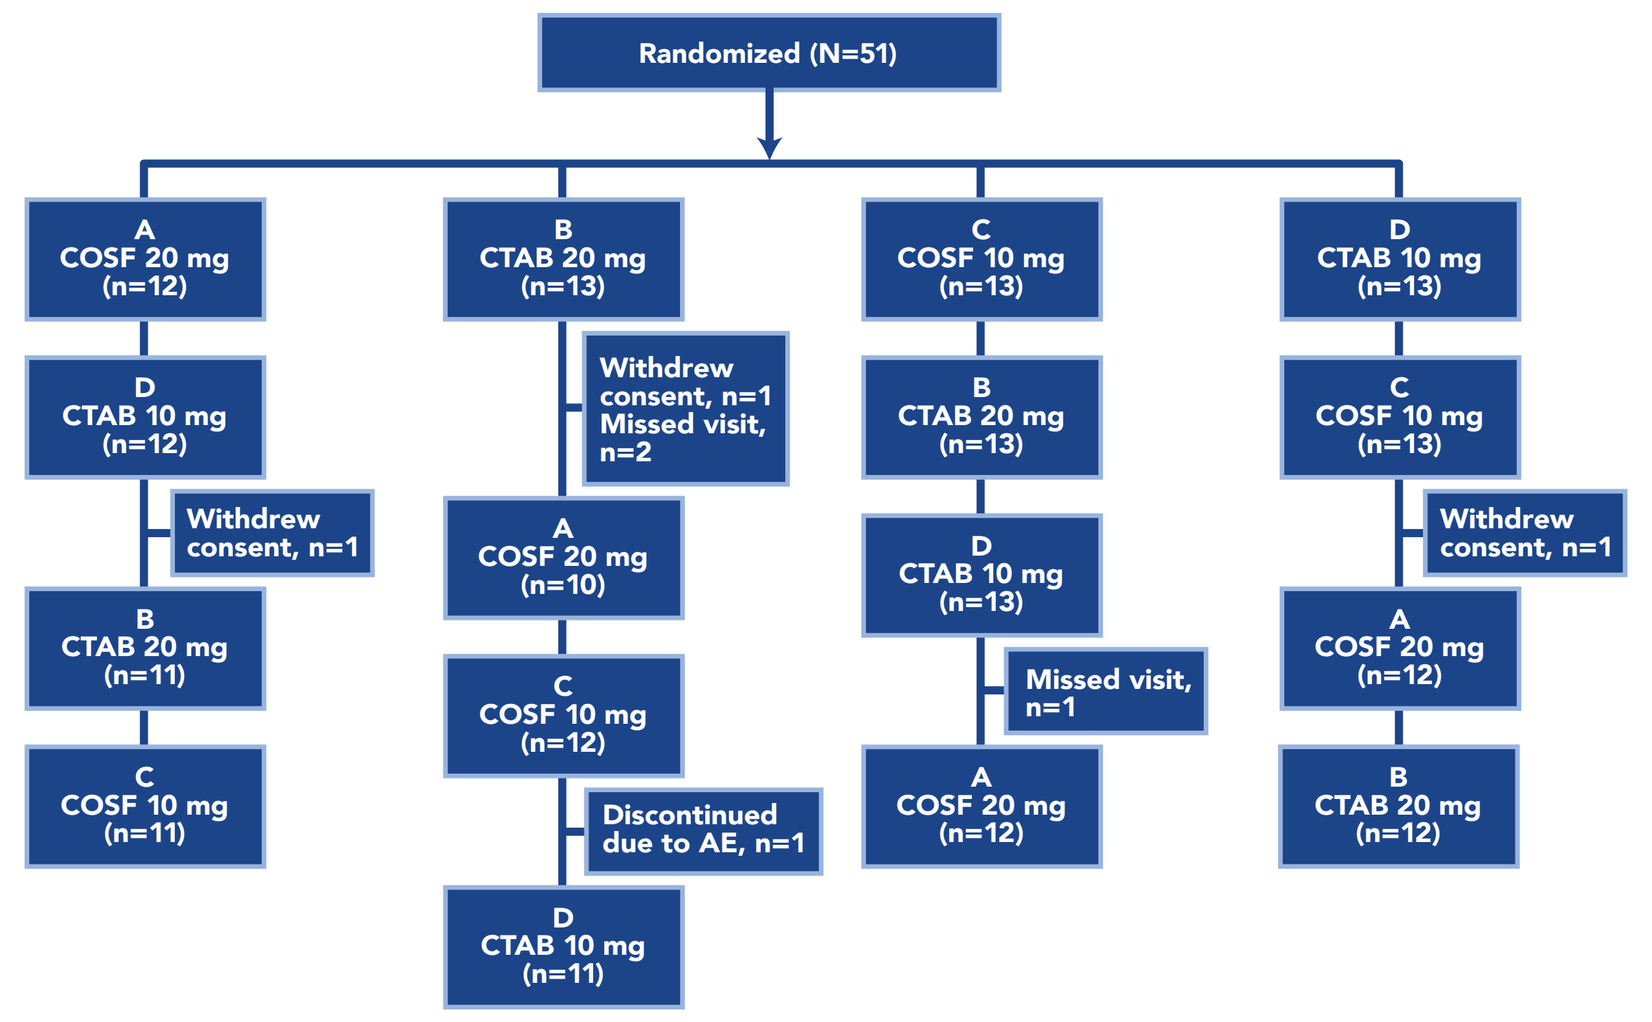
Randomized (N=51)
A COSF 20 mg (n=12)
D CTAB 10 mg (n=12)
Withdrew consent, n=1
B CTAB 20 mg (n=11)
C COSF 10 mg (n=11)
B CTAB 20 mg (n=13)
Withdrew consent, n=1 Missed visit, n=2
A COSF 20 mg (n=10)
C COSF 10 mg (n=12)
Discontinued due to AE, n=1
D CTAB 10 mg (n=11)
C COSF 10 mg (n=13)
B CTAB 20 mg (n=13)
D CTAB 10 mg (n=13)
Missed visit, n=1
A COSF 20 mg (n=12)
D CTAB 10 mg (n=13)
C COSF 10 mg (n=13)
Withdrew consent, n=1
A COSF 20 mg (n=12)
B CTAB 20 mg (n=12)

Treatment Sequences
1. A→D→B→C
2. B→A→C→D
3. C→B→D→A
4. D→C→A→B

AE: adverse event; COSF: clobazam oral soluble film; CTAB: clobazam tablet

TABLE 1: SUMMARY OF DEMOGRAPHIC CHARACTERISTICS OF THE STUDY POPULATION

Demographic Characteristic	Subjects (N=51)
Age (years), mean ± SD	43.4 ± 11.5
Age group (years), n (%)	
18-40	23 (45.1)
≥40	28 (54.9)
Sex, n (%)	
Female	25 (49)
Male	26 (51)
Ethnicity, n (%)	
Not Hispanic or Latino	3 (5.9)
Hispanic or Latino	48 (94.1)
Race, n (%)	
White	43 (84.3)
Black	8 (15.7)
Height (cm), mean ± SD	167.84 ± 8.41
Weight (kg), mean ± SD	73.82 ± 12.08
Body mass index (kg/m^2), mean ± SD	26.07 ± 2.65

N: number of subjects treated; n (%): number and percentage of subjects; SD: standard deviation.

- Of four subjects who did not complete the study, two withdrew consent for personal reasons, one withdrew because of difficulty with venipuncture, and one was withdrawn due to increased blood pressure and heart rate before receiving the final dose; two subjects who completed the study failed to appear for one of the study treatments.

PHARMACOKINETICS

- All subjects who completed at least two study periods, including two treatments for which a statistical comparison was possible, and for whom the clobazam/N-desmethylclobazam PK profile could be adequately characterized, were included in the PK analyses and comprised the PK population.
- The PK population for clobazam was N=45 for comparisons A versus B and C versus A, and N=47 for comparisons C versus D and D versus B.

TABLE 2: SUMMARY OF PHARMACOKINETIC PARAMETERS FOR CLOBAZAM – PK POPULATION

Parameter	A: COSF 20 mg (N=45)			B: CTAB 20 mg (N=47)			C: COSF 10 mg (N=47)			D: CTAB 10 mg (N=47)		
	Mean	SD	CV%	Mean	SD	CV%	Mean	SD	CV%	Mean	SD	CV%
AUC_{0-t} (ng*h/mL)	10871.43	2835.05	26.08	10304.23	2870.75	27.86	4737.49	1333.00	28.14	4735.07	1250.26	26.40
AUC_{0-inf} (ng*h/mL)	11054.21	2908.92	26.32	10493.78	2899.09	27.63	4895.17	1339.79	27.37	4918.00	1291.70	26.26
Residual area (%)	1.59	0.94	58.81	1.85	1.01	54.56	3.40	1.79	52.61	3.72	1.76	47.19
C_{max} (ng/mL)	389.30	102.20	26.25	387.04	102.19	26.40	185.36	49.86	26.90	191.88	37.39	19.49
$T_{1/2el}$ (h)	45.51	12.76	28.03	45.50	13.09	28.78	42.82	12.02	28.08	43.93	13.10	29.82
K_{el} (L/h)	0.0166	0.0055	33.0093	0.0167	0.0057	34.3840	0.0177	0.0059	33.2927	0.0173	0.0059	33.8111
CL/F (L/h)	1.93	0.49	25.22	2.05	0.56	27.25	2.19	0.58	26.68	2.16	0.52	24.23
V_d/F (L)	120.19	24.19	20.12	126.71	25.63	20.23	128.15	27.42	21.40	130.57	32.47	24.87
	Median	Min	Max	Median	Min	Max	Median	Min	Max	Median	Min	Max
T_{max} (h)	1.516	0.333	4.000	2.000	0.658	6.000	1.500	0.663	3.517	1.500	0.664	4.000

AUC: area under the plasma concentration-time curve; CL/F: apparent total clearance of the drug from plasma; C_{max}: maximum plasma drug concentration; COSF: clobazam oral soluble film; CTAB: clobazam tablet; CV%: coefficient of variation; K_{el}: terminal elimination rate constant; Max: maximum; Min: minimum; $T_{1/2el}$: terminal elimination half-life; T_{max}: time to reach maximum plasma concentration; V_d/F: volume of distribution

AVERAGE BIOEQUIVALENCE

- The ratios (A/B and C/D) with 90% geometric CIs for clobazam AUC_{0-t}, AUC_{0-inf}, and C_{max} indicate COSF 20 mg is bioequivalent to clobazam tablet 20 mg, and COSF 10 mg is bioequivalent to clobazam tablet 10 mg.
- Median T_{max} values following the 20 mg dose were 1.5 hours for COSF and 2.0 hours for the clobazam tablet (*P*=0.07). The median T_{max} value following the 10 mg dose was 1.5 hours for both formulations (*P*=0.80).

TABLE 3: RATIOS (A/B AND C/D), 90% GEOMETRIC CONFIDENCE INTERVALS, INTRA- AND INTERSUBJECT CV (%) FOR CLOBAZAM – AUC_{0-t}, AUC_{0-inf}, AND C_{max}

Treatment Comparison	Parameter	Geometric LSM (A)	Geometric LSM (B)	Ratio (%)[1]	90% Geometric CI[2] Lower (%)	90% Geometric CI[2] Upper (%)	Intrasubject CV (%)	Intersubject CV (%)
COSF 20 mg (A) – CTAB 20 mg (B)	AUC_{0-t} (ng*h/mL)	10531.45	10152.24	103.74	101.32	106.21	6.48	24.63
	AUC_{0-inf} (ng*h/mL)	10712.10	10344.68	103.55	101.16	106.00	6.41	24.74
	C_{max} (ng/mL)	386.59	376.84	102.59	95.43	110.28	20.05	14.37
		Geometric LSM (C)	Geometric LSM (D)					
COSF 10 mg (C) – CTAB 10 mg (D)	AUC_{0-t} (ng*h/mL)	4554.83	4583.30	99.38	96.81	102.02	7.49	25.76
	AUC_{0-inf} (ng*h/mL)	4714.55	4759.87	99.05	96.72	101.43	6.79	25.44
	C_{max} (ng/mL)	179.96	188.53	95.45	90.19	101.03	16.29	17.12

[1]*Calculated using LSM according to the formula: $e^{(Difference)}$ * 100*
[2]*90% geometric confidence interval using ln-transformed data*
AUC: area under the plasma concentration-time curve; CI: confidence interval; C_{max}: maximum plasma drug concentration; COSF: clobazam oral soluble film; CTAB: clobazam tablet; CV%: coefficient of variation; LSM: least-squares mean

DOSE-PROPORTIONALITY

- The ratios (C/A and D/B) with 90% geometric CIs for clobazam AUC_{0-t}, AUC_{0-inf}, and C_{max}, dose normalized to 20 mg, indicate that both COSF and clobazam tablets are dose proportional over the 10 to 20 mg dose range.

TABLE 4: RATIOS (C/A AND D/B), 90% GEOMETRIC CONFIDENCE INTERVALS, INTRA- AND INTERSUBJECT CV (%) FOR CLOBAZAM – AUC_{0-t}, AUC_{0-inf}, AND C_{max} DOSE NORMALIZED TO 20 mg

Treatment Comparison	Parameter	Geometric LSM (C)	Geometric LSM (A)	Ratio (%)[1]	90% Geometric CI[2] Lower (%)	90% Geometric CI[2] Upper (%)	Intrasubject CV (%)	Intersubject CV (%)
COSF 10 mg (C) – COSF 20 mg (A)	AUC_{0-t} (ng*h/mL)	9307.37	10500.45	88.64	86.22	91.12	7.80	25.22
	AUC_{0-inf} (ng*h/mL)	9637.19	10670.76	90.31	87.95	92.75	7.48	24.96
	C_{max} (ng/mL)	364.68	378.73	96.29	91.49	101.34	14.45	21.08
		Geometric LSM (D)	Geometric LSM (B)					
CTAB 10 mg (D) – CTAB 20 mg (B)	AUC_{0-t} (ng*h/mL)	9178.21	9920.30	92.52	90.23	94.87	7.22	25.93
	AUC_{0-inf} (ng*h/mL)	9531.31	10108.59	94.29	92.05	96.59	6.94	25.86
	C_{max} (ng/mL)	376.03	374.25	100.47	95.04	106.22	16.12	15.72

[1]*Calculated using LSM according to the formula: $e^{(Difference)}$ * 100*
[2]*90% geometric confidence interval using ln-transformed data*
AUC: area under the plasma concentration-time curve; CI: confidence interval; C_{max}: maximum plasma drug concentration; COSF: clobazam oral soluble film; CTAB: clobazam tablet; CV%: coefficient of variation; LSM: least-squares mean

N-DESMETHYLCLOBAZAM

- The PK population for N-desmethylclobazam was N=44 for comparisons A versus B and C versus A, and N=46 for comparisons C versus D and D versus B.
- These analyses indicated that COSF 20 mg is bioequivalent to clobazam tablet 20 mg, and COSF 10 mg is bioequivalent to clobazam tablet 10 mg (data not shown).
- The data further indicated that both COSF and clobazam tablets are dose-proportional over the 10 to 20 mg dose range for N-desmethylclobazam (data not shown).

SAFETY AND TOLERABILITY

Treatment-emergent Adverse Events

- The number of AEs and the number of subjects experiencing AEs were dose-related across the treatment groups, with somnolence the most common event. None of these TEAEs were severe or serious, and most were considered mild.

TABLE 5: MOST FREQUENTLY REPORTED TREATMENT-EMERGENT ADVERSE EVENTS

MedDRA® System Organ Class / MedDRA® Preferred Term	COSF 20 mg (N=46) Number of Events	COSF 20 mg (N=46) Subjects, n (%)	CTAB 20 mg (N=49) Number of Events	CTAB 20 mg (N=49) Subjects, n (%)	COSF 10 mg (N=49) Number of Events	COSF 10 mg (N=49) Subjects, n (%)	CTAB 10 mg (N=49) Number of Events	CTAB 10 mg (N=49) Subjects, n (%)	Overall (N=51) Number of Events	Overall (N=51) Subjects, n (%)
All TEAEs	34	21 (45.7)	28	20 (40.8)	24	15 (30.6)	20	13 (26.5)	106	34 (66.7)
Nervous system disorders	26	20 (43.5)	19	16 (32.7)	16	12 (24.5)	13	11 (22.4)	74	27 (52.9)
Somnolence	16	15 (32.6)	16	14 (28.6)	9	9 (18.4)	5	5 (10.2)	46	22 (43.1)
Headache	8	8 (17.4)	0	0	5	4 (8.2)	6	6 (12.2)	19	14 (27.5)
Dizziness	2	2 (4.3)	3	3 (6.1)	2	1 (2.0)	1	1 (2.0)	8	6 (11.8)
Gastrointestinal disorders	1	1 (2.2)	2	2 (4.1)	4	3 (6.1)	5	4 (8.2)	12	9 (17.6)
Vomiting	0	0	0	0	1	1 (2.0)	3	3 (6.1)	4	4 (7.8)
Diarrhea	0	0	2	2 (4.1)	1	1 (2.0)	0	0	3	3 (5.9)
Conditions related to venipuncture	5	2 (4.3)	1	1 (2.0)	0	0	0	0	6	3 (5.9)
Influenza-like illness	0	0	0	0	0	0	2	2 (4.1)	2	2 (3.9)

Each subject could contribute only once to each of the incidence rates, regardless of the number of occurrences
Overall: includes results from all treatment groups
COSF: clobazam oral soluble film; CTAB: clobazam tablet; MedDRA®: Medical Dictionary for Regulatory Activities Version 20.0; TEAE: treatment-emergent adverse event

- One subject discontinued the study due to increased blood pressure (153/92 mm Hg) and heart rate (110 bpm) without symptoms prior to the scheduled administration of clobazam tablet 10 mg in Period 4. The principal investigator judged that a causal relationship between these events and study drug was unlikely.

Safety Evaluations

- Visual inspections of the administration site following COSF revealed no evidence of mucosal irritation.
- No relevant differences in mean values, or changes from baseline, for vital signs were observed over time, and there were no relevant differences across treatments.
- Physical examination, ECG, and routine clinical laboratory tests revealed no clinically significant abnormalities attributable to study drug.

CONCLUSIONS

- COSF at single doses of 10 and 20 mg is bioequivalent to the clobazam tablet at equivalent doses with respect to both clobazam and its active metabolite N-desmethylclobazam.
- The pharmacokinetics of COSF after single dose administrations are dose-proportional at doses of 10 and 20 mg with respect to both clobazam and its active metabolite N-desmethylclobazam.
- The number of AEs and the number of subjects experiencing AEs were dose-related across the treatment groups; none of the AEs were severe or serious, and most were considered mild.
- COSF provides a simple, reliable therapeutic option with precise dosing.

REFERENCES

1. Ng YT, Conry JA, Drummond R, Stolle J, Weinberg MA. Randomized, phase III study results of clobazam in Lennox-Gastaut syndrome. *Neurology.* Oct 11 2011; 77(15):1473-1481.
2. Ng YT, Conry J, Paolicchi J, et al. Long-term safety and efficacy of clobazam for Lennox-Gastaut syndrome: interim results of an open-label extension study. *Epilepsy Behav.* Dec 2012;25(4):687-694.
3. Arzimanoglou A, French J, Blume WT, et al. Lennox-Gastaut syndrome: a consensus approach on diagnosis, assessment, management, and trial methodology. *Lancet Neurol.* Jan 2009;8(1):82-93.
4. Kerr M, Kluger G, Philip S. Evolution and management of Lennox-Gastaut syndrome through adolescence and into adulthood: are seizures always the primary issue? *Epileptic Disord.* May 2011;13 Suppl 1:S15-26.
5. Ogawa K, Kanemoto K, Ishii Y, et al. Long-term follow-up study of Lennox-Gastaut syndrome in patients with severe motor and intellectual disabilities: with special reference to the problem of dysphagia. *Seizure.* Apr 2001;10(3):197-202.
6. Yin HS, Parker RM, Sanders LM, et al. Liquid medication errors and dosing tools: A randomized controlled experiment. *Pediatrics.* Oct 2016;138(4).

ACKNOWLEDGEMENTS

Medical writing support was provided by Jonathan Sokolowski, PhD, of ACCESS Medical, LLC; medical writing support was funded by Aquestive Therapeutics.

FUNDING AND DISCLOSURES

Financial support for this study was provided by Aquestive Therapeutics. Aquestive Therapeutics participated in the interpretation of data, review, and approval of the poster. All authors contributed to the development of the poster and maintained control over the final content. Allen H. Heller is a consultant to Aquestive Therapeutics. Stephen Wargacki, Cassie Jung, and A. Mark Schobel are employees of Aquestive Therapeutics.

Aquestive