Poster 2.129

PATIENT AND CAREGIVER PREFERENCE FOR ROUTE OF ADMINISTRATION OF A BENZODIAZEPINE FOR CONTROL OF INCREASED SEIZURE ACTIVITY IN STABLE PATIENTS

Ayanna Santos[1], Gary Slatko[2]

[1]TVP Management Company, LLC, Succasunna, NJ; [2]Aquestive Therapeutics, Inc., Warren, NJ

INTRODUCTION

- Despite the use of antiepileptic medications, many patients with epilepsy experience cluster seizures[1]
- Adverse consequences associated with cluster seizures include:
 - Increased emergency room visits and hospital admissions[2]
 - Potential evolution to status epilepticus, a life-threatening medical emergency[3]
 - Possible lasting morphological cortical brain damage[4]
 - An increased risk of death[5]
- Until recently, diazepam rectal gel[6] was the only FDA-approved rescue medication for patients with epilepsy experiencing increased seizure activity; however, the mechanics of administering diazepam rectal gel can be difficult, time-consuming, and embarrassing for patients and caregivers[7-9], which may affect compliance[10,11]
 - Midazolam nasal spray was approved by the FDA in May 2019 for use in this setting,[12] and at the time this study was conducted was not available to patients
- Patient and caregiver preferences are important considerations in rescue medication usage
 - Results from a survey assessing caregivers' preferences for intranasal midazolam versus rectal diazepam have recently been examined[13]; however, little is known regarding caregiver and patient preferences regarding a buccal versus an intranasal route of administration for rescue treatment
- Patients with epilepsy on stable treatment regimens requiring intermittent use of rescue medications may soon have access to novel formulations of diazepam, including a diazepam buccal film

OBJECTIVE

- To determine preferences regarding oral dissolving film strip versus nasal spray for rescue treatment in a real-world population of patients with epilepsy and their caregivers

METHODS

- A convenience sample of patients with seizures who had been prescribed diazepam for intermittent use to control increased seizure activity and their caregivers participated in a 10-minute online survey
- Respondents were initially asked to identify a preference for route of administration (nasal spray or oral dissolving film) without seeing administration instructions, and then were asked about the most important factors influencing their selection of one seizure rescue medication versus another
 - Choices were effectiveness, tolerability, portability, ease of use, immediacy, discretion, and social acceptability, each rated on a scale from 0 (least important) to 6 (most important)
- Respondents were also asked to indicate:
 - The importance of tolerability factors (no/limited administration site irritation; no/limited unpleasant smell/taste; limited somnolence) in an ideal rescue medication
 - The importance of portability factors (ability to carry in a wallet/small purse; ability to store at most temperatures; ability to carry in a discreet container) in an ideal rescue medication
 - The ideal amount of time it should take to access, prepare, and administer rescue medication (immediacy), and past experiences with use of nasal sprays

RESULTS

- A total of 29 respondents (15 patients and 14 caregivers) participated in the survey
- Demographics are summarized in **Table 1**
 - Overall, 17 (59%) respondents reported ≥5 episodes of cluster seizures during the 12 months prior to the survey

TABLE 1. PATIENT AND CAREGIVER DEMOGRAPHICS

	Patients (n=15)	Caregivers (n=14)	All Respondents (N=29)
Sex, n (%)			
Male	5 (33)	7 (50)	12 (41)
Female	10 (67)	7 (50)	17 (59)
Patient age, n (%), years			
18–39	9 (60)	N/A	—
40–65	6 (40)	N/A	—
Caregiver age, n (%), years			
2–20	N/A	7 (50)	—
21–65	N/A	7 (50)	—
Cluster seizures, last 12 mo, n (%)			
<5	8 (53)	4 (29)	12 (41)
5–25	6 (40)	7 (50)	13 (45)
>25	1 (7)	3 (21)	4 (14)
Mean, n	9	28	18
Region, n (%)*			
West/Southwest	—	—	10 (34)
Midwest	—	—	6 (21)
Southeast	—	—	6 (21)
Northeast	—	—	7 (24)

**West/Southwest: AK, CA, CO, HI, ID, MT, NV, OR, UT, WA, WY, AZ, NM, OK, TX; Midwest: IA, IN, IL, KS, MI, MN, NE, OH, ND, SD, WI, MO; Southeast: AL, AR, FL, GA, NC, LA, MS, KY, SC, TN, WV, VA; Northeast: CT, DE, ME, MD, MA, NJ, NY, PA, RI, VT, NH.*
N/A, not available.

- Only 7% of patients and 36% of caregivers reported current use of any rescue medication for cluster seizures
- Approximately one-half of all respondents had used diazepam to control episodes of increased seizure activity (**Figure 1**)

Figure 1. Experience With Previous Diazepam Use to Control Increased Seizure Activity Among Patients and Caregivers

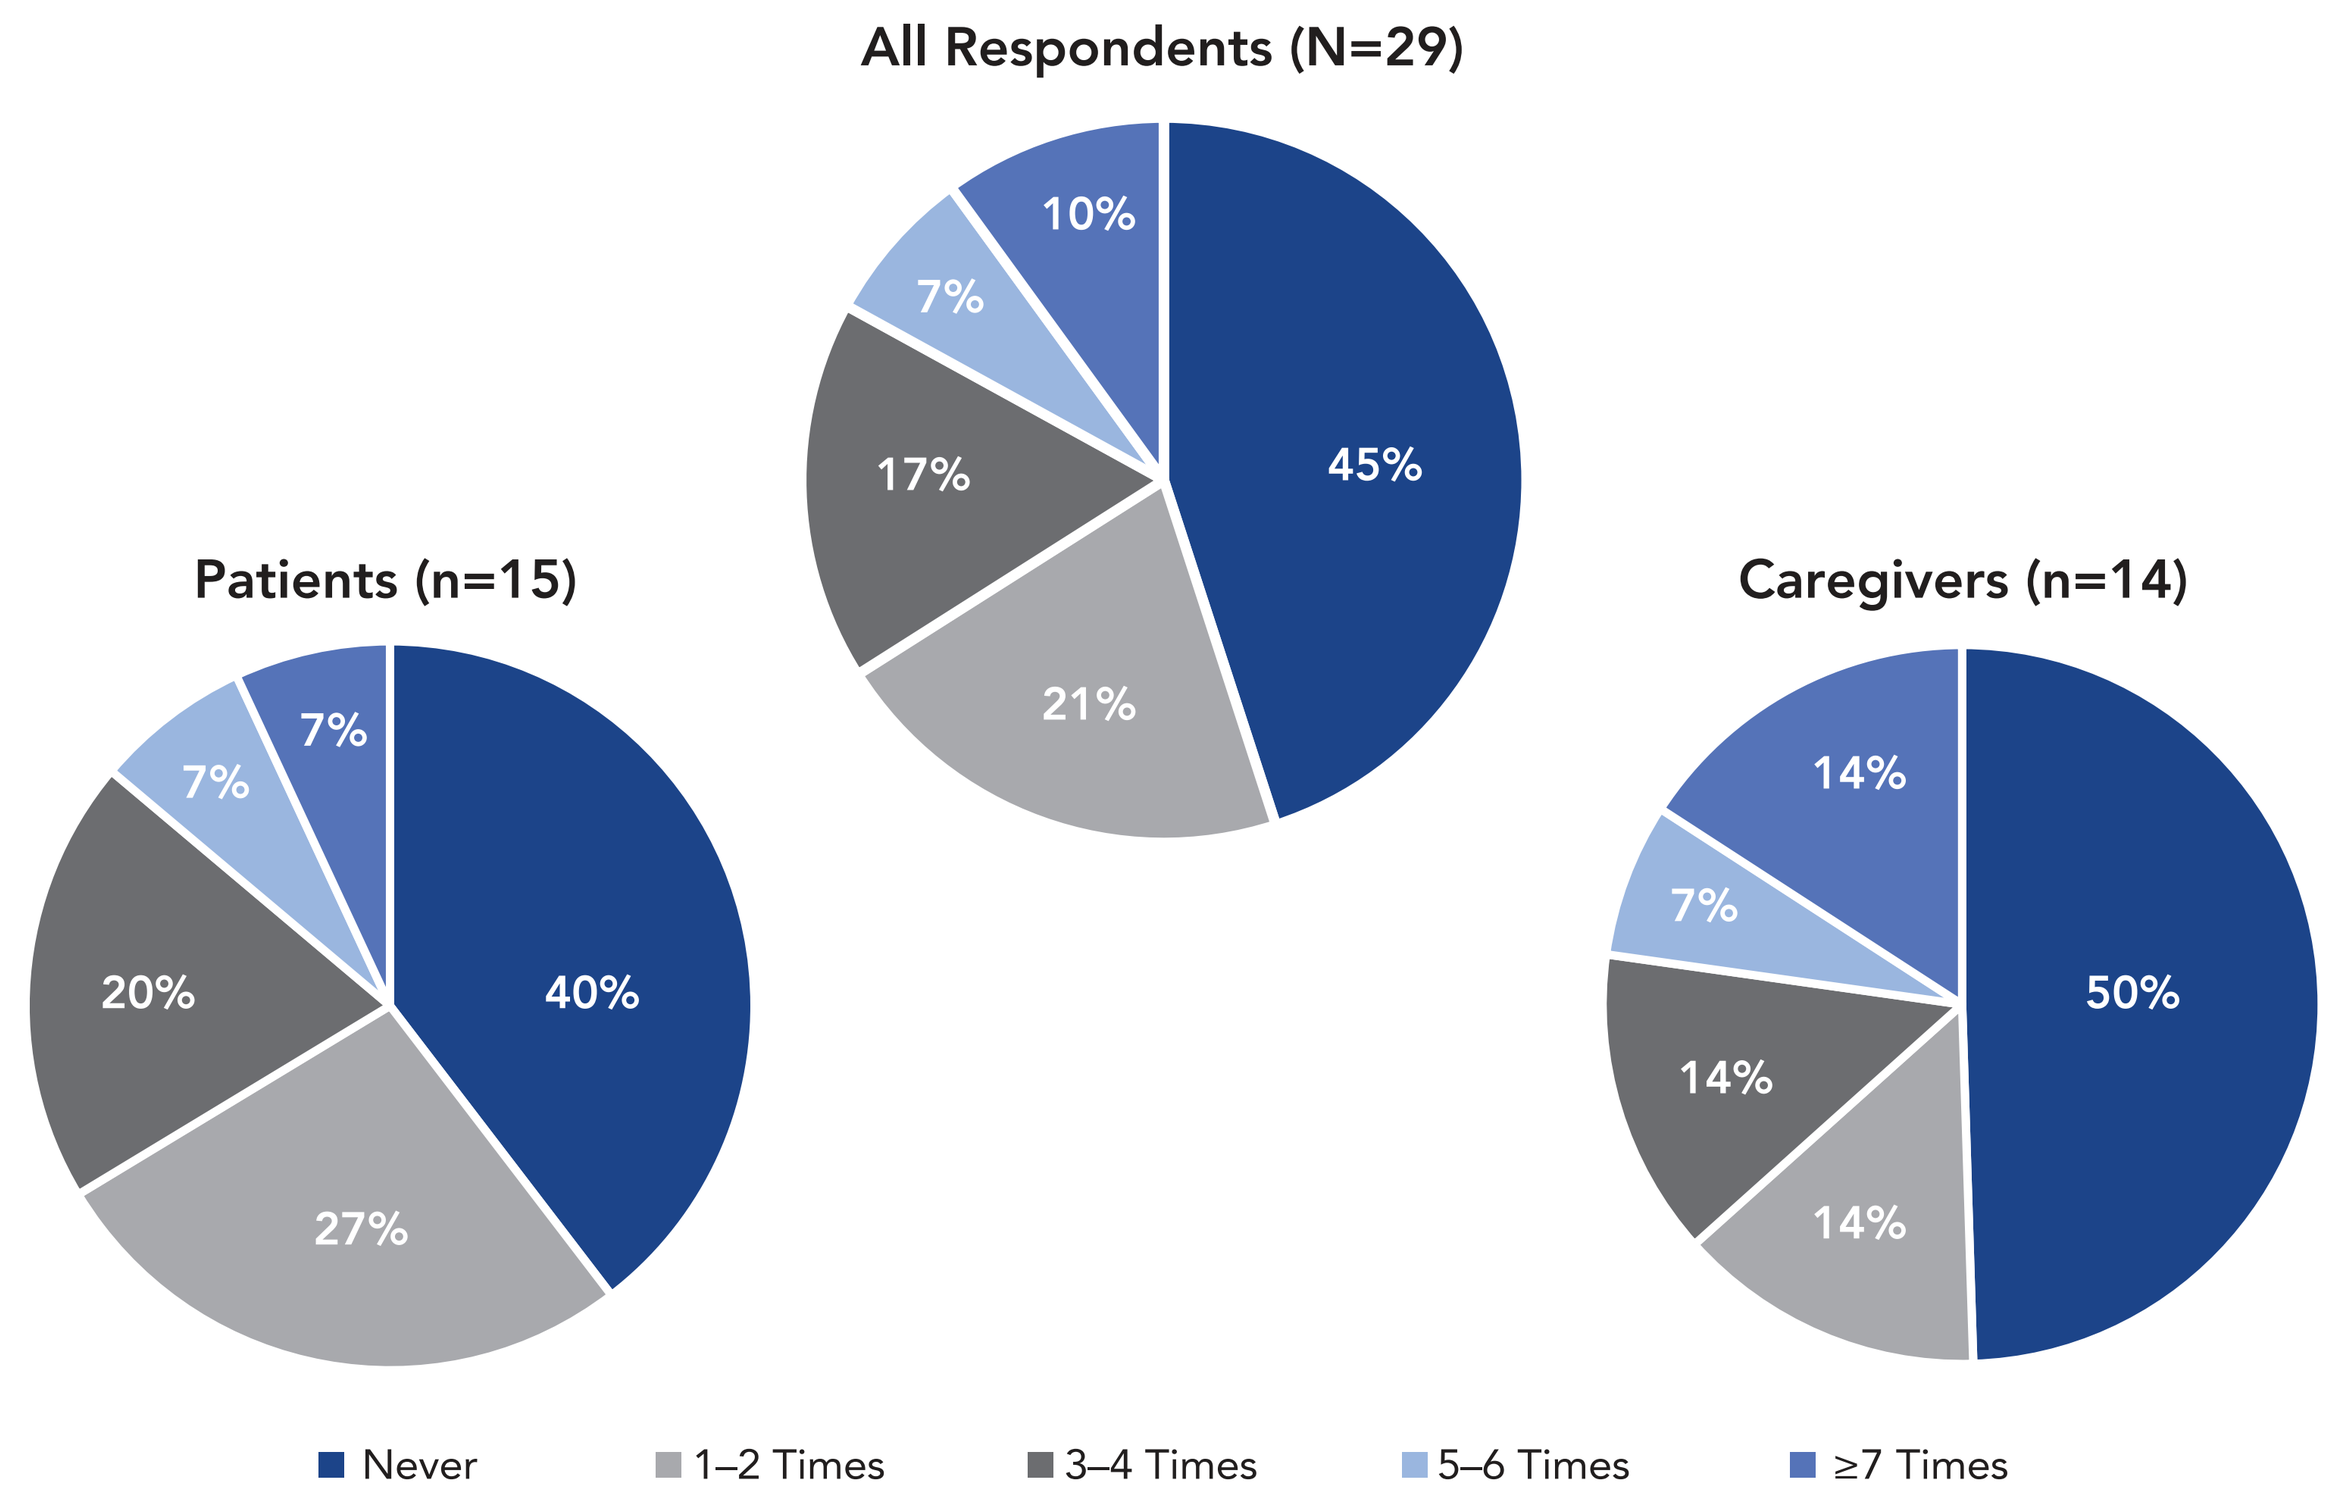
All Respondents (N=29)
45%
21%
17%
7%
10%
Patients (n=15)
40%
27%
20%
7%
7%
Caregivers (n=14)
50%
14%
14%
7%
14%
Never
1–2 Times
3–4 Times
5–6 Times
≥7 Times

- The majority of patients (80%) and caregivers (71%) initially preferred an oral film strip to a nasal spray formulation. Following a review of instructions of each medication, 87% of patients and 86% of caregivers indicated a preference for oral film strip over nasal spray (**Figure 2**).

Figure 2. Patient and Caregiver Preference Before and After Reviewing Rescue Seizure Medication Administration Instructions

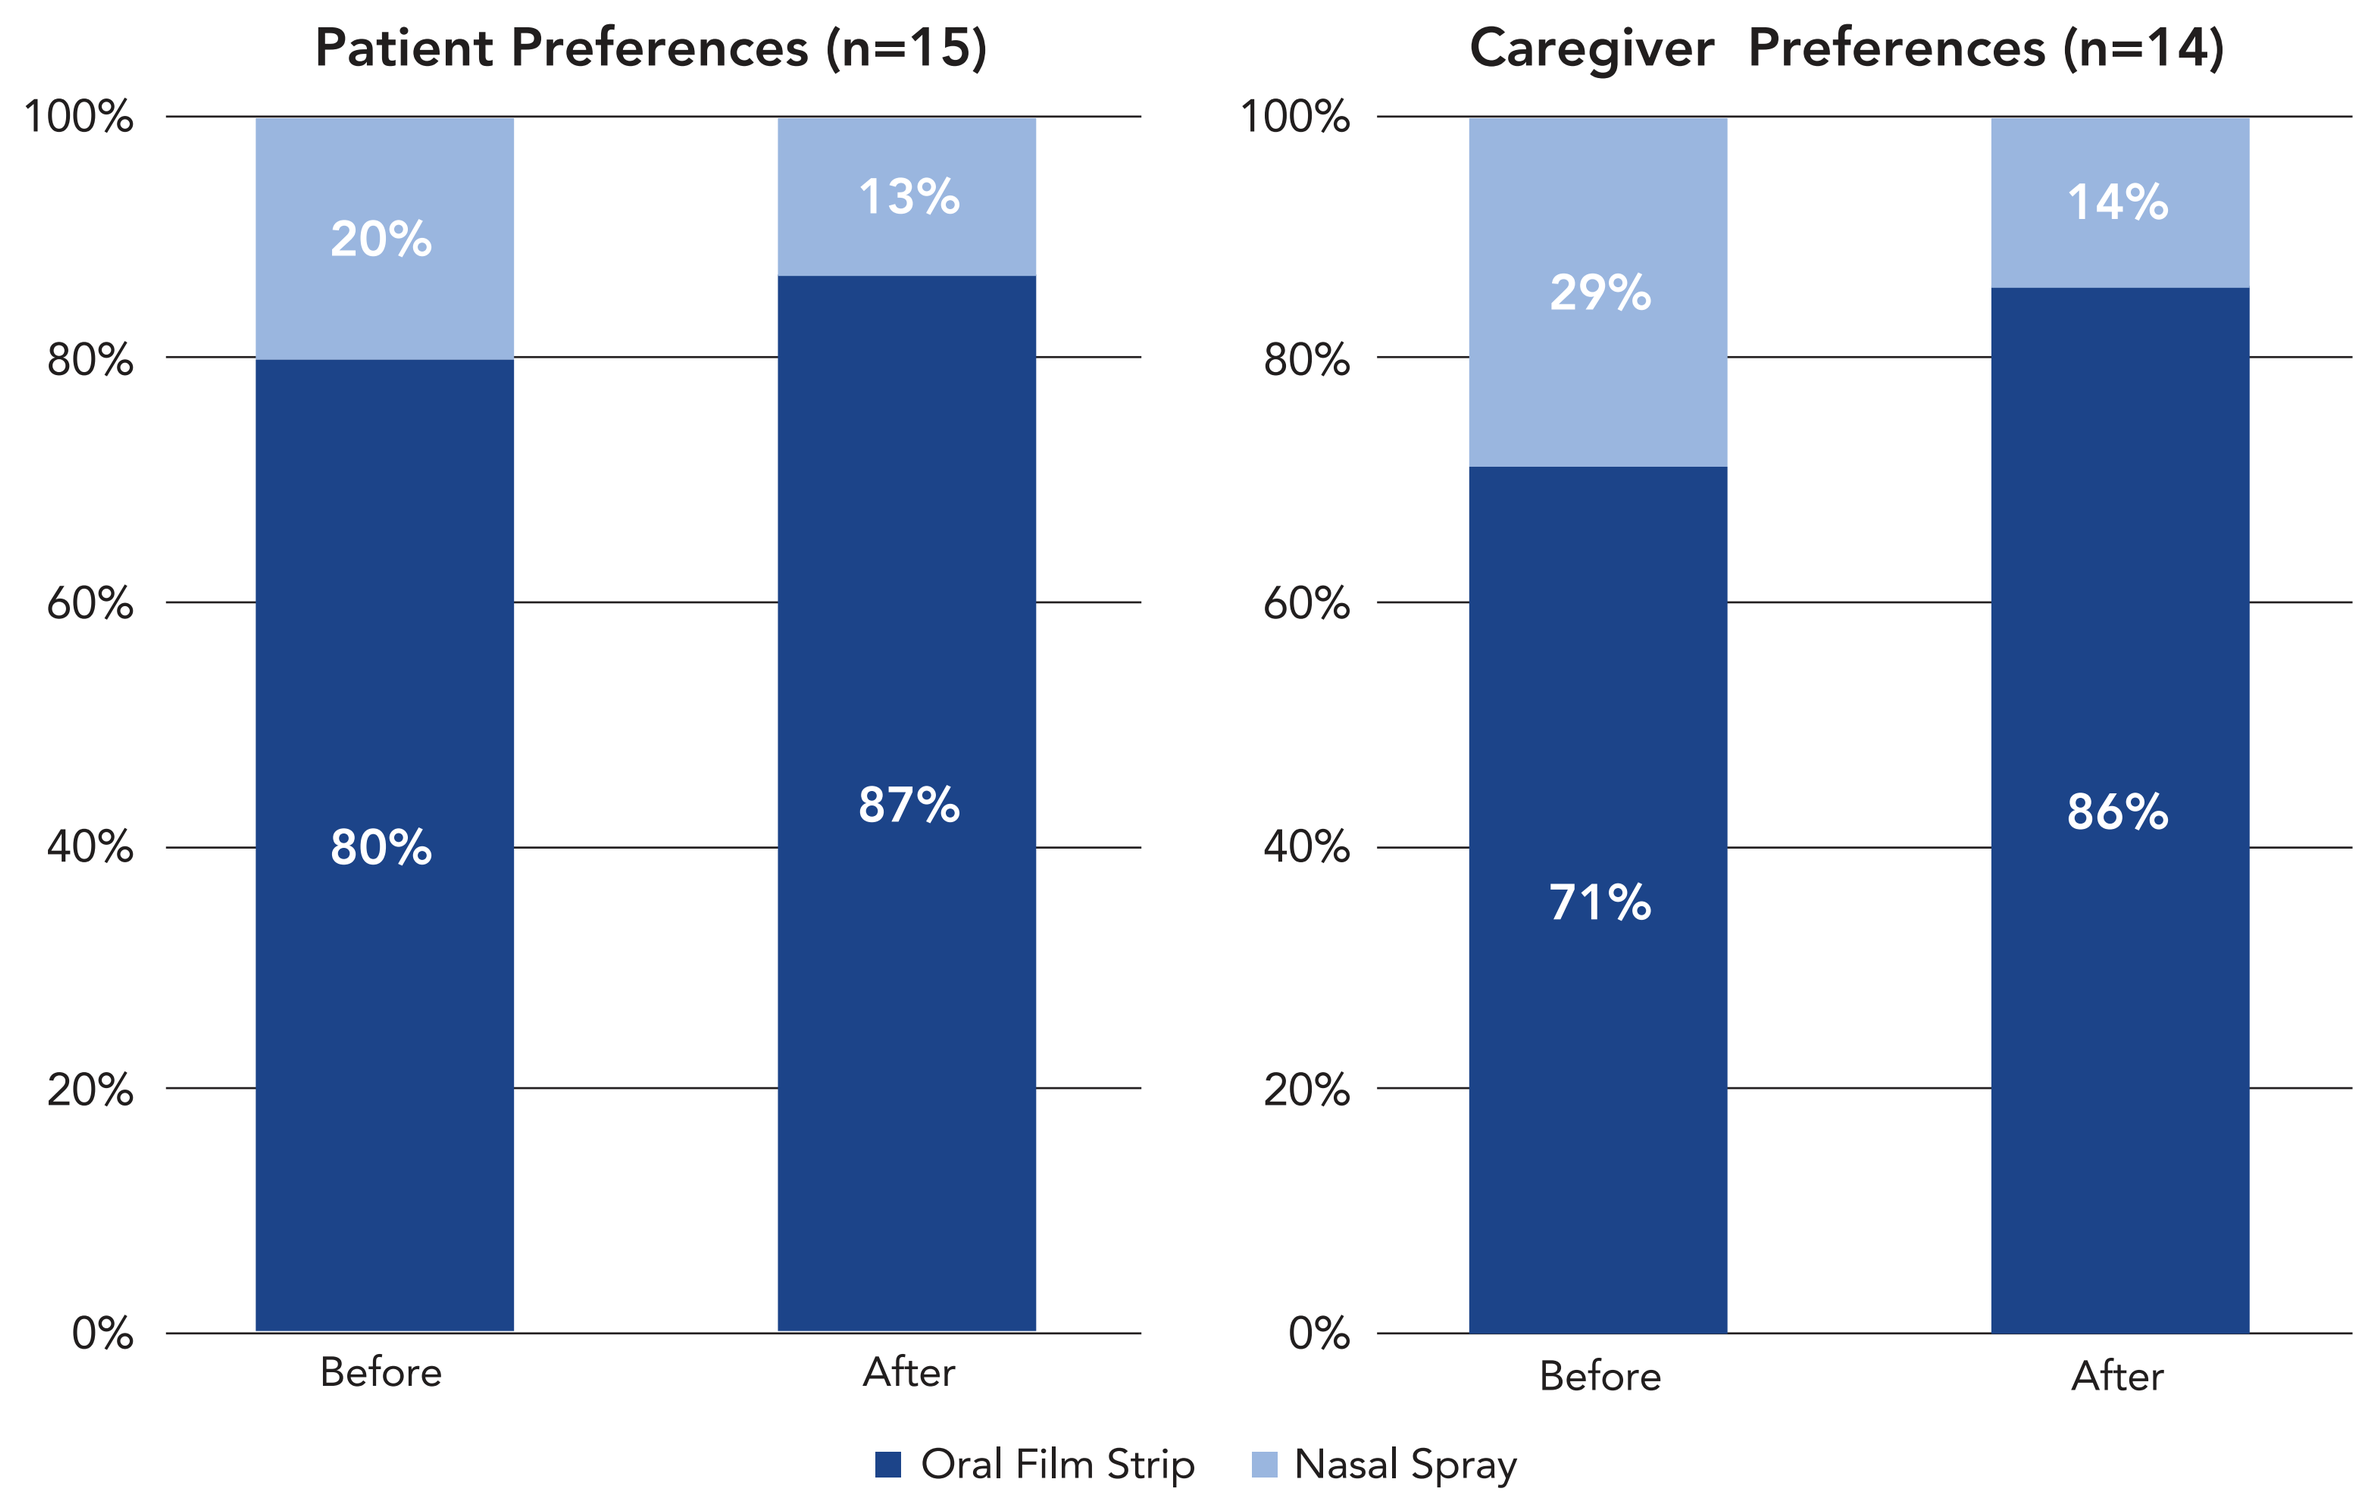
Patient Preferences (n=15)
Caregiver Preferences (n=14)
100%
80%
60%
40%
20%
0%
20%
80%
13%
87%
29%
71%
14%
86%
Before
After
Oral Film Strip
Nasal Spray

- Overall, 8 (28%) respondents (5 patients, 3 caregivers) reported previous use of a nasal spray as rescue medication for any indication (eg, epilepsy, migraine)
- Respondents ranked effectiveness, immediacy, ease of use and tolerability as the most important factors in selecting a seizure rescue medication (**Figure 3**)

Figure 3. Most Important Reasons for Selecting a Seizure Rescue Medication as Reported by All Respondents (N=29)

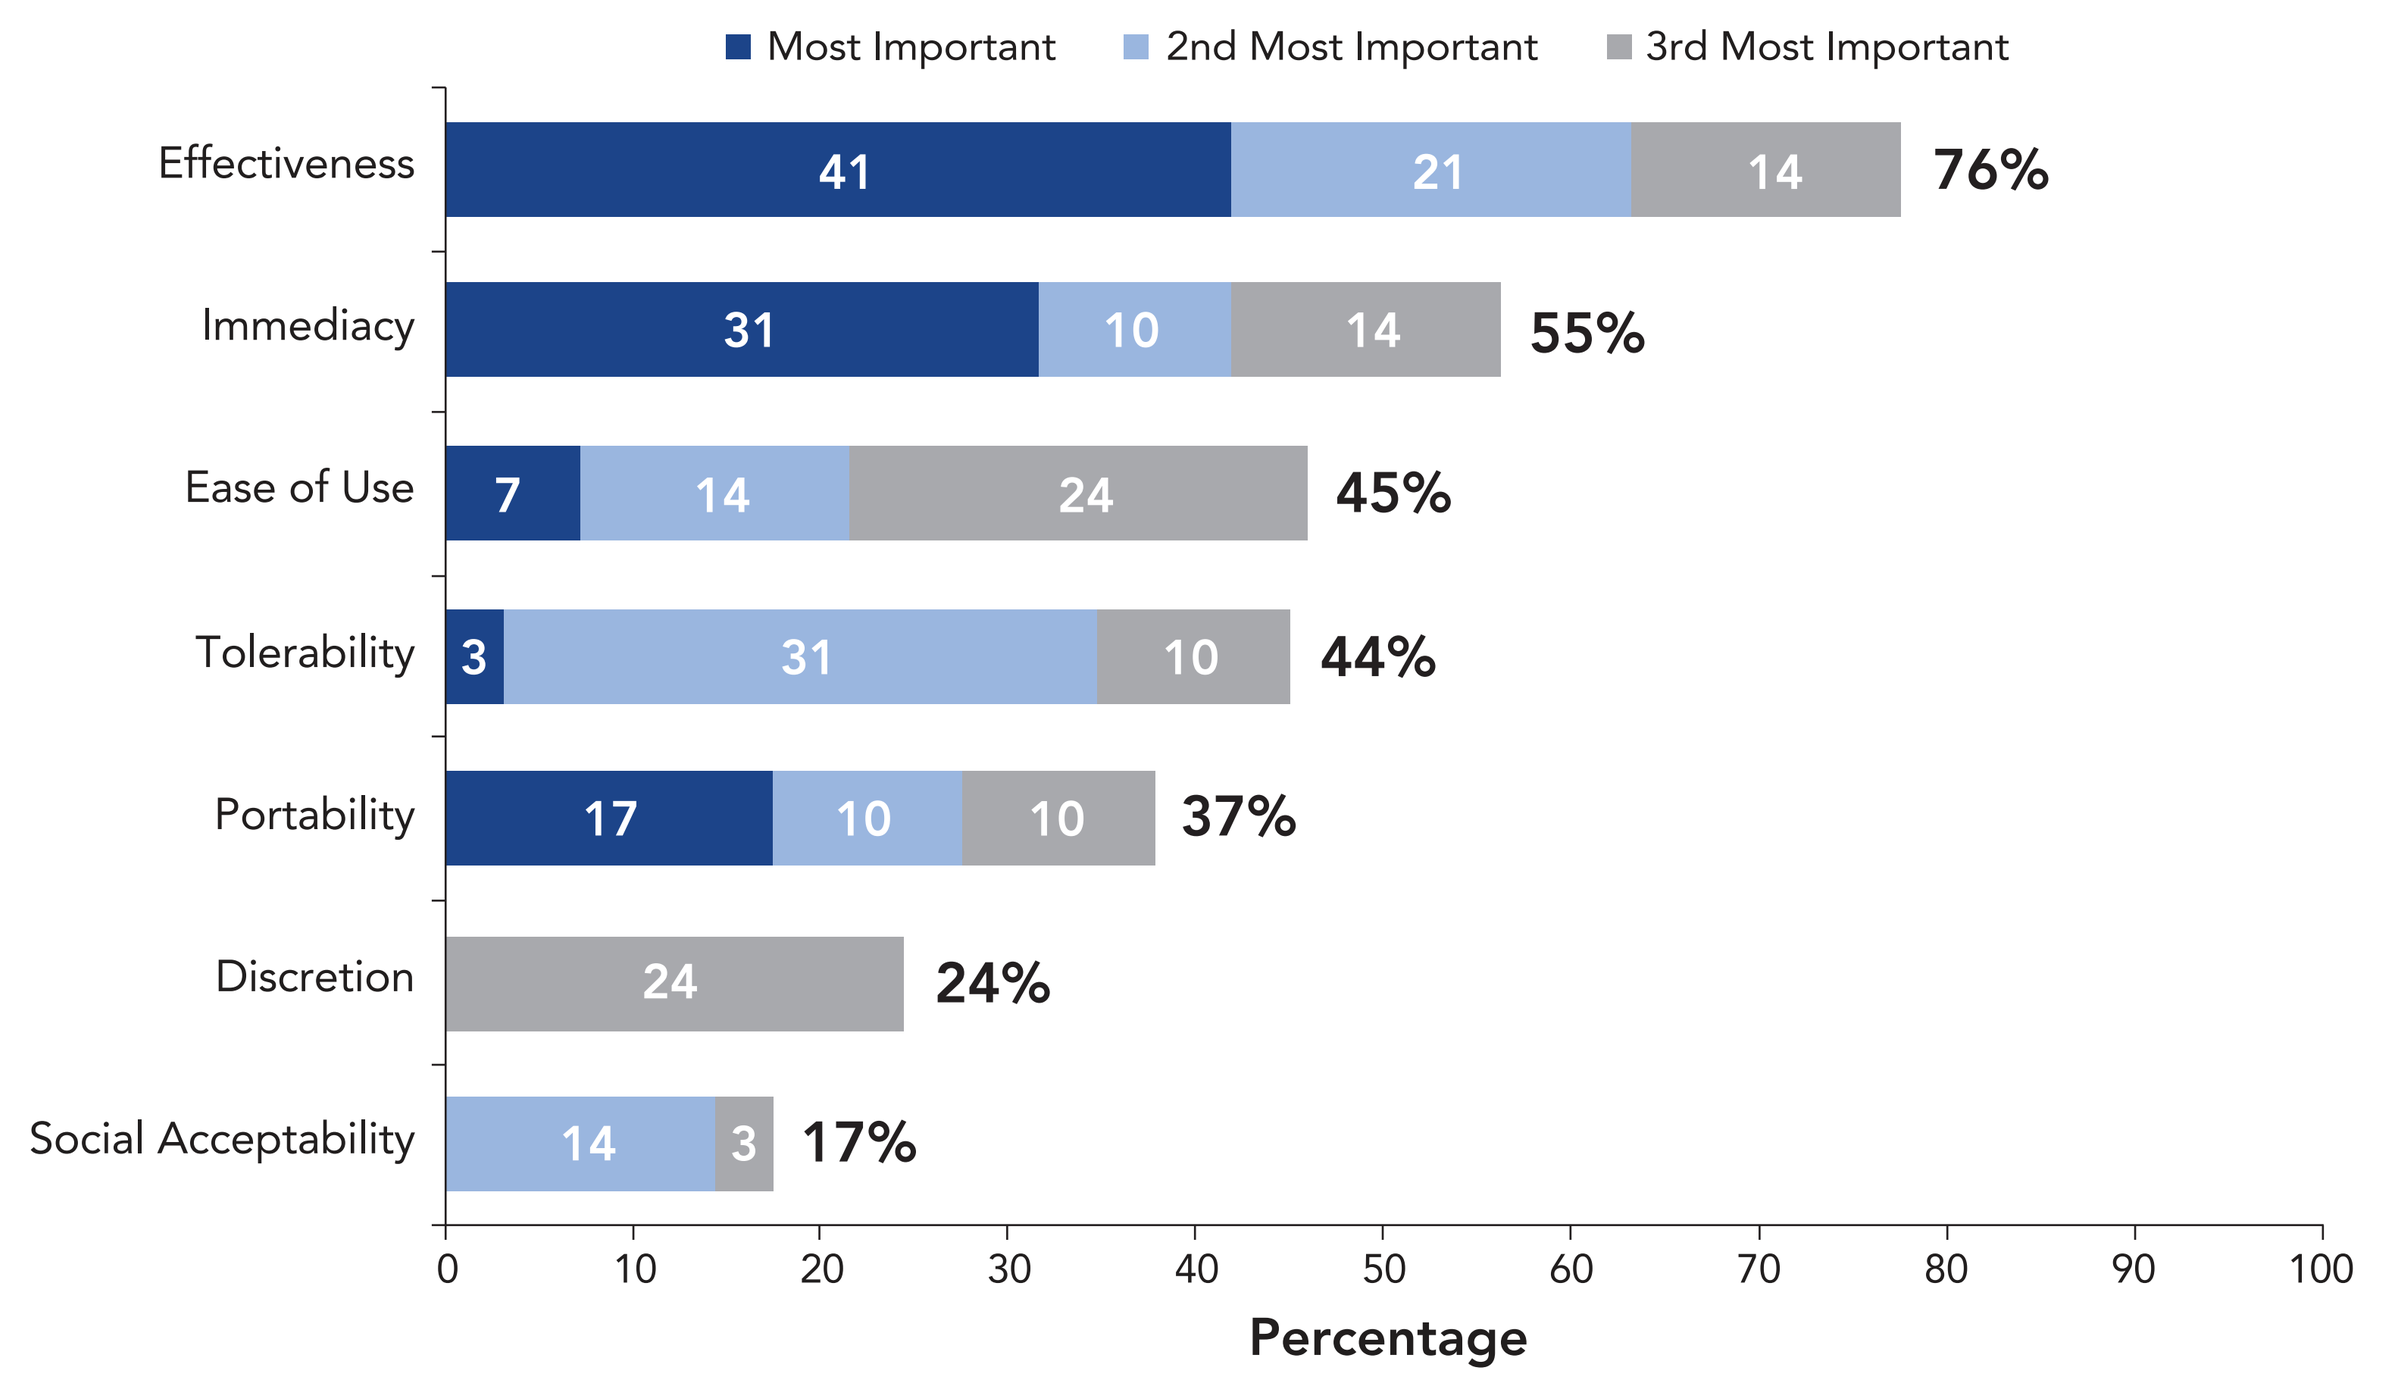
Most Important
2nd Most Important
3rd Most Important
Effectiveness
41
21
14
76%
Immediacy
31
10
14
55%
Ease of Use
7
14
24
45%
Tolerability
3
31
10
44%
Portability
17
10
10
37%
Discretion
24
24%
Social Acceptability
14
3
17%
0
10
20
30
40
50
60
70
80
90
100
Percentage

- Important tolerability and portability considerations are summarized in **Table 2**
 - Minimal administration site irritation and somnolence were reported as the most important tolerability considerations overall, and ability to store product at most temperatures was reported as the most important portability consideration overall
 - For patients, ability to carry their rescue medication in a wallet or small purse was the most important portability consideration

TABLE 2. MOST IMPORTANT TOLERABILITY AND PORTABILITY CONSIDERATIONS FOR AN IDEAL RESCUE MEDICATION

	Patients (n=15)	Caregivers (n=14)	All Respondents (N=29)
Tolerability characteristic, n (%)			
No/limited administration site irritation	9 (60)	9 (64)	18 (62)
Limited somnolence (sleepiness)	12 (80)	6 (43)	18 (62)
No/limited unpleasant taste or smell	9 (60)	5 (36)	14 (48)
Portability characteristic, n (%)			
Ability to store at most temperatures	10 (67)	12 (86)	22 (76)
Ability to carry in a wallet or small purse	12 (80)	6 (43)	18 (62)
Ability to carry in a discreet container	11 (73)	4 (29)	15 (29)

- When asked about the ideal amount of time it should take to access, prepare, and administer seizure rescue medication, the vast majority of respondents reported the ideal amount of time to be ≤5 minutes, with the majority of those preferring <1 minute (**Figure 4**)

Figure 4. Reported Ideal Amount of Time it Should Take to Access, Prepare, and Administer Seizure Rescue Medication

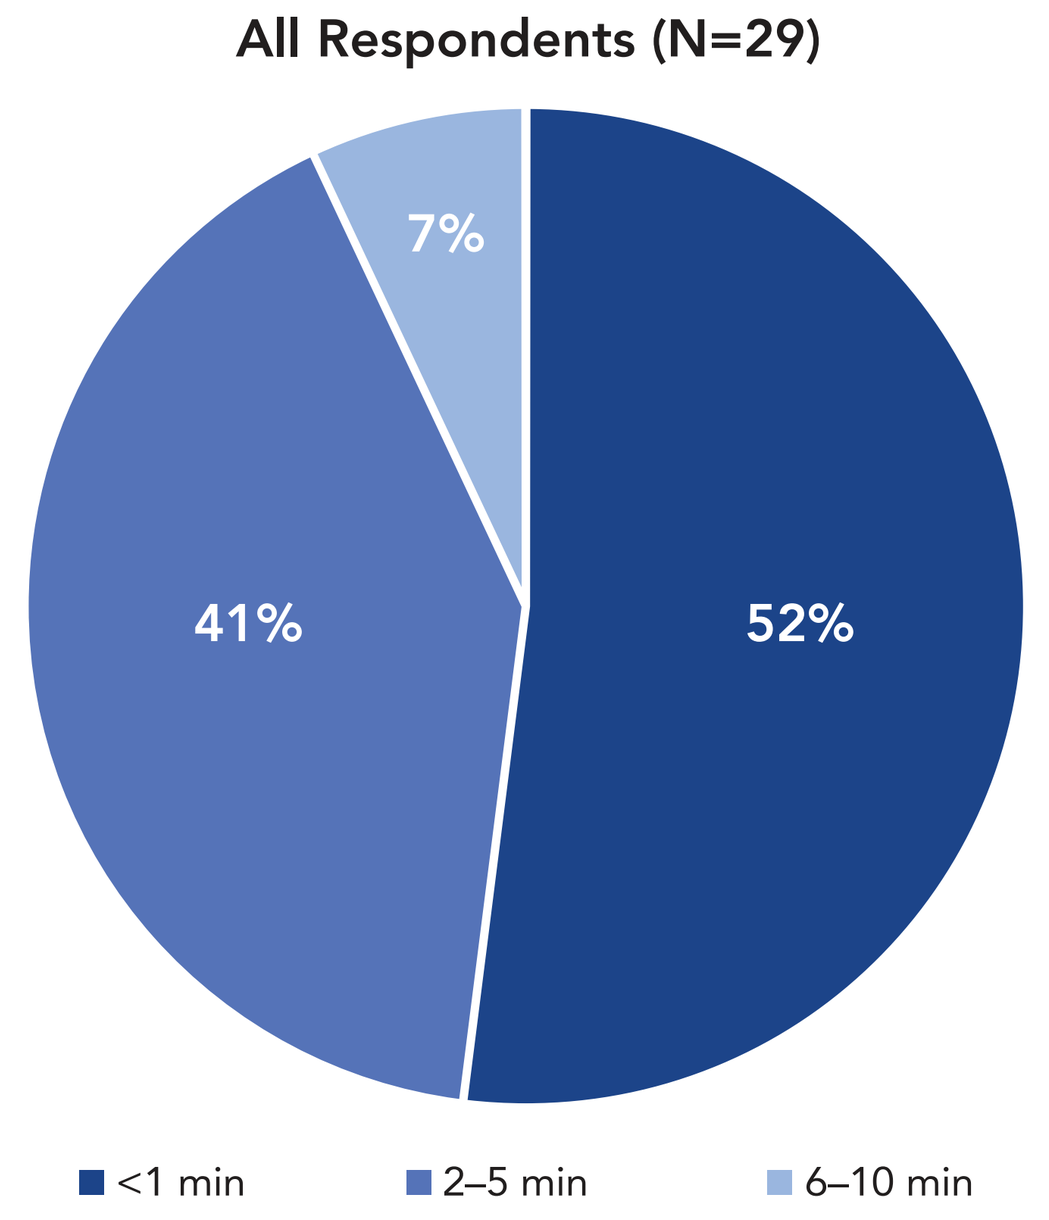
All Respondents (N=29)
52%
41%
7%
<1 min
2–5 min
6–10 min

CONCLUSIONS

- Rescue medications appear to be underutilized; only 21% (6/29) of survey respondents reported current use of any rescue medications to control increased seizure activity
- The majority of patients and their caregivers chose an oral dissolving film over nasal spray as their preferred route of administration for a benzodiazepine to control increased seizure activity
- Effectiveness, immediacy, ease of use, and tolerability were chosen as the most important considerations in an ideal rescue medication

REFERENCES

1. Brodie MJ. *Epilepsia*. 2013;54 Suppl 2:5-8.
2. Haut SR. *Epilepsy Behav*. 2006;8:50-5.
3. Mitchell WG. *J Child Neurol*. 2002;17 Suppl 1:S36-43.
4. Bergen DC. *Epilepsy Curr*. 2006;6:117-8.
5. Sillanpaa M, Schmidt D. *Brain*. 2008;131:938-44.
6. Diastat C-IV (diazepam rectal gel) [package insert]. San Antonio, TX: DPT Laboratories; 2016.
7. Jafarpour S, et al. *Seizure*. 2019;68:9-13.
8. Hartman AL, et al. *Pediatrics*. 2016;137.
9. Haut SR, et al. *Epilepsy Behav*. 2016;63:109-17.
10. Tatum IW. *Epilepsy Behav*. 2002;3:535-8.
11. Cereghino JJ. *Curr Treat Options Neurol*. 2007;9:249-55.
12. Nayzilam [package insert]. Smyrna, GA: UCB, Inc.; 2019.
13. Nunley S, et al. *Epilepsy Behav*. 2019;92:53-6.

ACKNOWLEDGMENTS

The survey and this analysis were sponsored by Aquestive Therapeutics, Inc. Medical writing support was provided by Michael Morren, RPh, MBA, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, and funded by Aquestive Therapeutics, Inc.

DISCLOSURES

Ayanna Santos has served as a paid consultant to Aquestive Therapeutics, Inc. Gary Slatko is an employee of Aquestive Therapeutics, Inc.

Presented at: American Epilepsy Society Annual Meeting; December 6–10, 2019; Baltimore, Maryland