Poster 2.258

UNRELIABLE ABSORPTION WITH RECTAL ADMINISTRATION OF DIAZEPAM

Gary Slatko[1], Stephen Wargacki[1], David J. Wyatt[2], Allen H. Heller[3]

[1]Aquestive Therapeutics, Warren, NJ; [2]Syneos Health Clinical Solutions Early Phase, Miami, FL; [3]Pharma Study Design, LLC, Woodbridge, CT

INTRODUCTION

- For many patients with epilepsy, breakthrough seizures or cluster seizures continue to occur despite treatment with anti-epilepsy drugs, and rapid treatment of these seizures is required to help prevent neuronal damage and other morbidities[1-3]
- Until recently, diazepam rectal gel (DRG) was the only FDA-approved treatment for the management of selected patients with epilepsy who experience bouts of increased seizure activity[4]
- The route of administration for DRG presents challenges for patients and caregivers, as it may be more difficult and time-consuming to administer than nonrectal medications.[2,3,5] Rectal administration can also cause embarrassment and may be less socially acceptable than other forms of treatment[1-3]
- Diazepam buccal film (DBF) is a novel formulation of diazepam under development as a therapeutic alternative to DRG for the treatment of breakthrough or cluster seizures[6,7]
- In a previously reported study comparing the bioavailability of DBF and DRG in healthy adults, high variability in pharmacokinetic (PK) parameters was observed following administration of DRG but not DBF, with some subjects showing extremely low plasma concentrations following single doses of DRG on multiple occasions[7]

OBJECTIVE

- To assess the available published literature to identify whether other instances of low systemic exposure following rectal administration of diazepam in individual subjects have been reported

METHODS

LITERATURE SEARCH AND ANALYSIS

- We conducted a search of the PubMed database (from database inception to September 18, 2019) to identify primary studies of rectal absorption of diazepam
 - The following search protocol was used: ("diazepam"[title/abstract] AND "absorption" [title/abstract] AND "rectal"[title/abstract])
- Publications that reported diazepam plasma concentration data after rectal administration for individual study subjects were included
- Subjects in each study were categorized as having low rectal absorption if they had diazepam plasma levels that were either: ≥1.5 standard deviations below the group mean[8-12]; <20% of the group median[13]; ≤50 ng/mL and group outliers[8,14]; or explicitly reported as having low drug plasma levels by study investigators[15-17]
- Findings from the literature analysis were compared with observed data from the previously reported randomized-sequence, open-label, crossover study that evaluated bioavailability of diazepam following DBF vs DRG administration in healthy adults (N=36)[7]
 - Each subject received single doses of DRG 5 mg, DRG 12.5 mg, DRG 20 mg, and DBF 15 mg under fasting conditions in 1 of 4 randomized treatment sequences, with a 28-day washout period between doses
 - Subjects were administered a Fleet® enema the evening before receiving each dose of DRG

RESULTS

- The PubMed search yielded 30 publications; of these, 10 studies reporting plasma levels of diazepam following rectal administration were determined to be relevant and were included in the analysis (**Table 1**)[8-17]
 - A total of 155 subjects across the 10 studies were included
- No instances of low absorption were reported in 5 of the studies, including the three smallest studies (N=6 subjects in each study; **Table 1**)[9-12,16]
- In the remaining 5 studies encompassing 106 patients, the proportions of subjects with low absorption of rectal diazepam ranged from 4.2% to 13.6% (**Table 1**)[8,13-15,17]
 - Two of these studies[16,17] reported the use of a cleansing enema prior to diazepam rectal administration; 3 studies[14,16,17] required subjects to fast prior to dosing

TABLE 1. SUMMARY OF LITERATURE SEARCH RESULTS

Study*	Population	Formulation	Predose cleansing enema?	Dosing under fasting conditions?	Subjects with low plasma levels of diazepam, n (%)
Mattila MAK, et al. *Br J Anaesth.* 1981;53:1269-1272[8]	Children undergoing surgery (N=22)	Solution	No	Not reported	3 (13.6)[†,‡]
Dhillon S, et al. *Arch Dis Child.* 1982;57:264-267[14]	Children with epilepsy (N=9)	Solution and suppository	No	Yes	1 (11.1)[‡]
Ivaturi V, et al. *Epilepsy Res.* 2013;103:254-261[13]	Healthy adults (N=12)	Gel	Not reported	Not reported	1 (8.3)[§]
Remy C, et al. *Epilepsia.* 1992;33:353-358[15]	Adults with epilepsy (N=39)	Solution	Not reported	Not reported	2 (5.1)[‖]
Garnett WR, et al. *Epilepsy Res.* 2011;93:11-16[17]	Healthy adults (N=24)	Gel	Yes	Fasting and nonfasting conditions	1 (4.2)[‖]
Agurell S, et al. *Epilepsia.* 1975;16:277-283[12]	Children with epilepsy (N=11)	Solution	Not reported	Not reported	0[†]
Cloyd JC, et al. *Epilepsia.* 1998;39:520-526[16]	Healthy male adults (N=20)	Gel	Yes	Yes	0[‖]
Magnussen I, et al. *Acta Pharmacol Toxicol.* 1979;45:87-90[9]	Adults with history of severe headache (N=6)	Solution	Not reported	Not reported	0[†]
Milligan N, et al. *Epilepsia.* 1982;23:323-331[10]	Adults with epilepsy (N=6)	Solution and suppository	No	Not reported	0[†]
Viukari M, et al. *Acta Pharmacol Toxicol.* 1981;49:59-64[11]	Hospitalized geriatric patients (N=6)	Solution and suppository	No	Not reported	0[†]

**All were single-dose studies; †Low absorption defined as diazepam plasma levels ≥1.5 standard deviations below the mean[8-12]; ‡Low absorption defined as diazepam plasma levels ≤50 ng/mL AND subject was an outlier[8,14]; §Low absorption defined as diazepam plasma levels <20% of the median[13]; ‖Low absorption explicitly reported by investigator.[15-17]*

- Of the 36 subjects in the previously reported study of bioavailability of DBF versus DRG, 2 (5.6%) subjects consistently exhibited extremely low diazepam exposure (C_{max} and $AUC_{0\text{-}t}$ >2 standard deviations below the group mean) following ≥2 DRG administrations separated by time (**Figures 1A and 1B**)[18]
 - Both subjects showed systemic exposure at or near the group mean following administration of DBF
 - Mean diazepam plasma concentrations over time for the overall study population are shown in **Figure 1C**

Figure 1. Diazepam Plasma Concentration-Time Curves Following Single Doses of DBF and DRG Through 6 Hours Post-Dose: A) Subject 1; B) Subject 2; and C) Overall Study Population

A. Subject 1

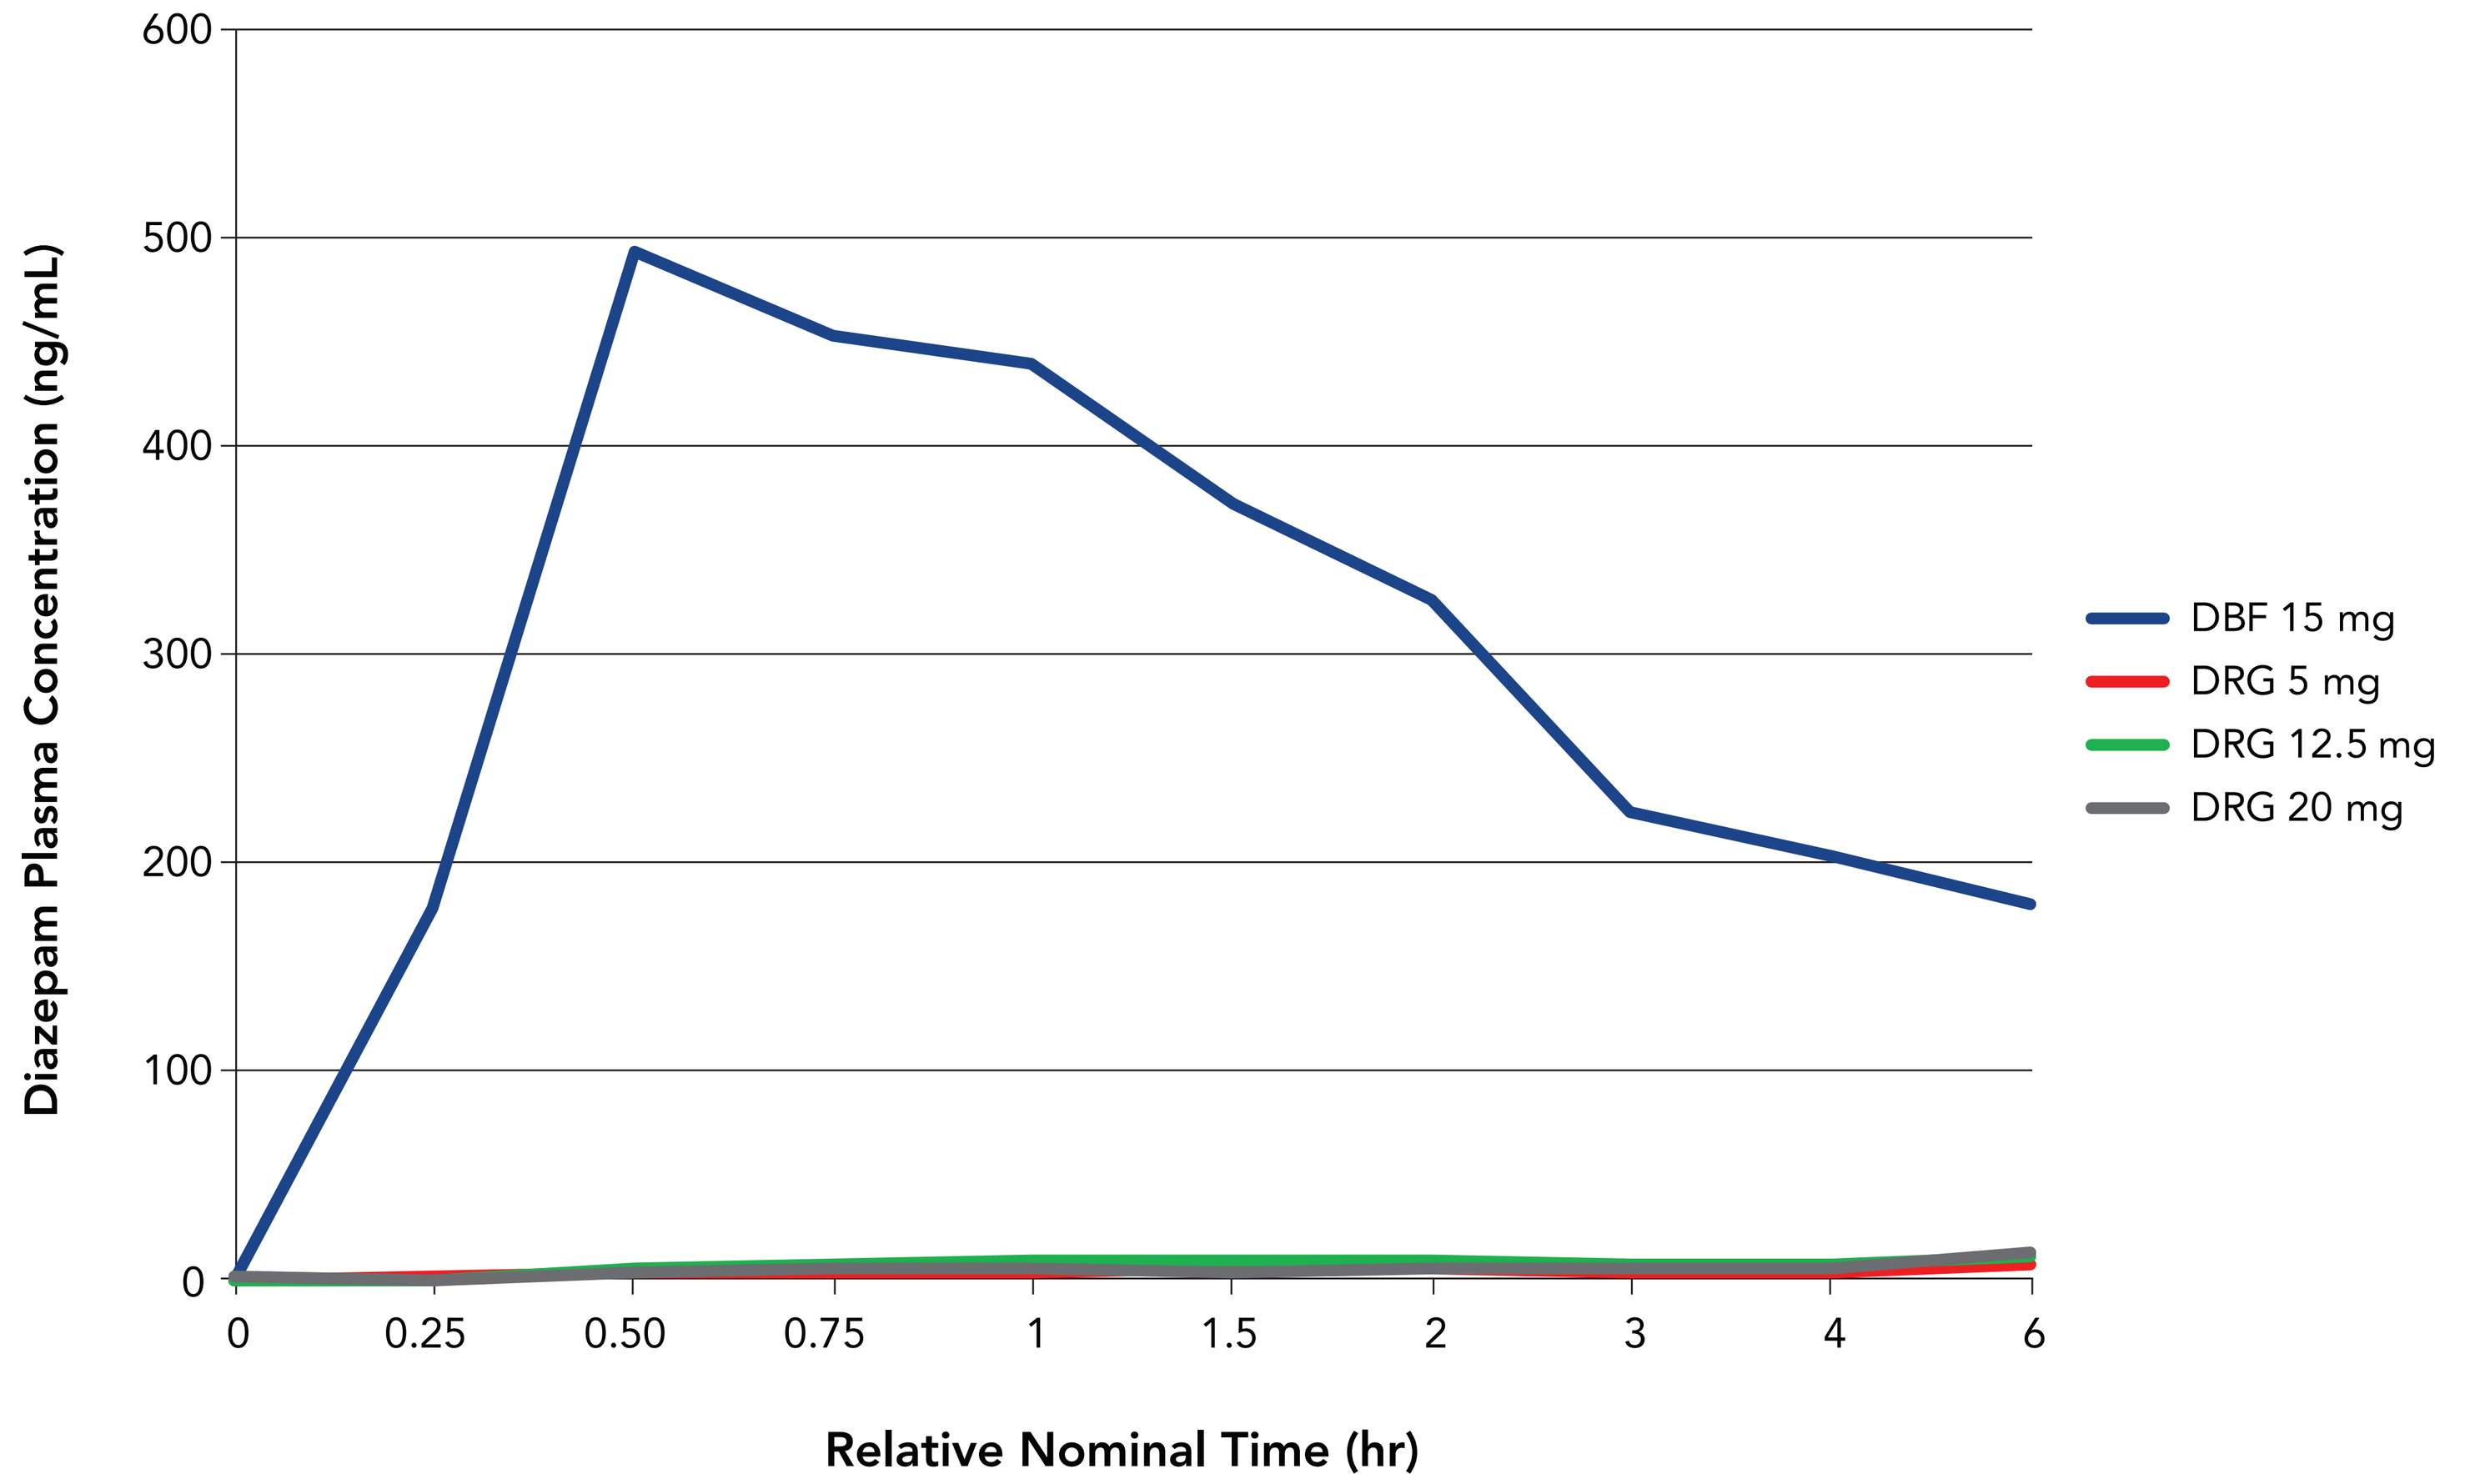
Diazepam Plasma Concentration (ng/mL)
0
100
200
300
400
500
600
0
0.25
0.50
0.75
1
1.5
2
3
4
6
Relative Nominal Time (hr)
DBF 15 mg
DRG 5 mg
DRG 12.5 mg
DRG 20 mg

B. Subject 2

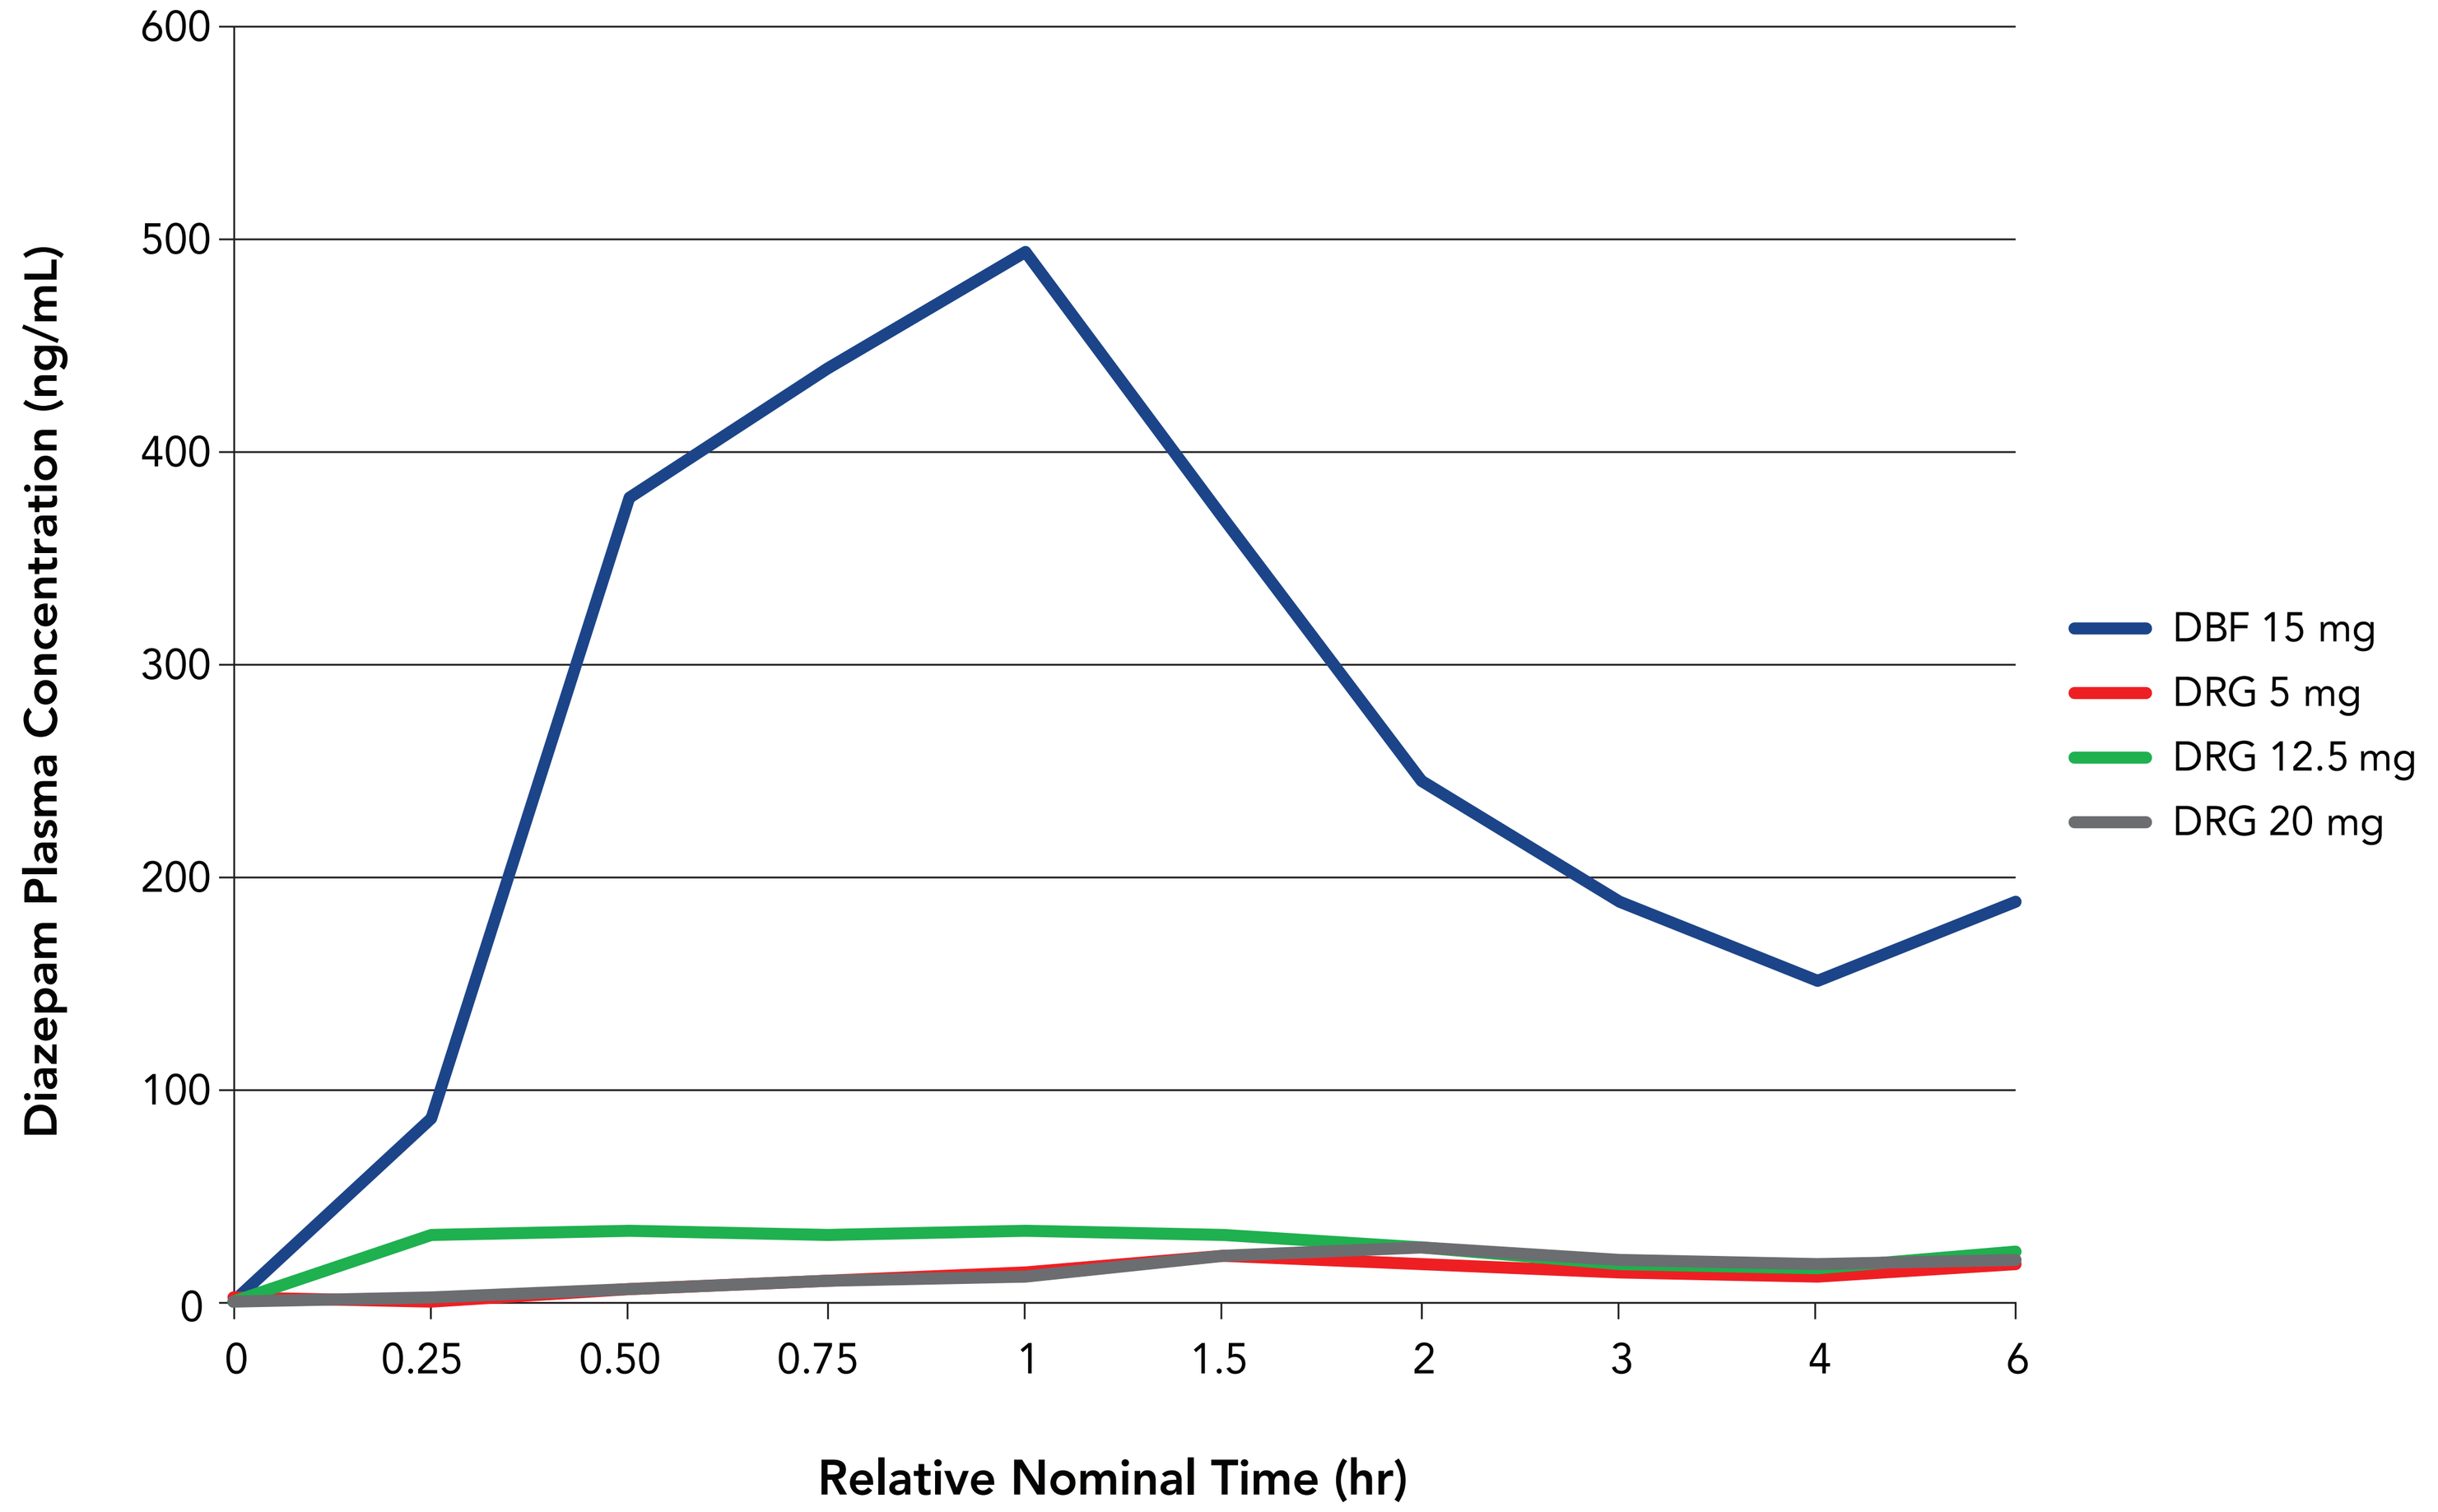
Diazepam Plasma Concentration (ng/mL)
0
100
200
300
400
500
600
0
0.25
0.50
0.75
1
1.5
2
3
4
6
Relative Nominal Time (hr)
DBF 15 mg
DRG 5 mg
DRG 12.5 mg
DRG 20 mg

C. Mean Plasma Concentration for the Overall Study Population (N=36)*

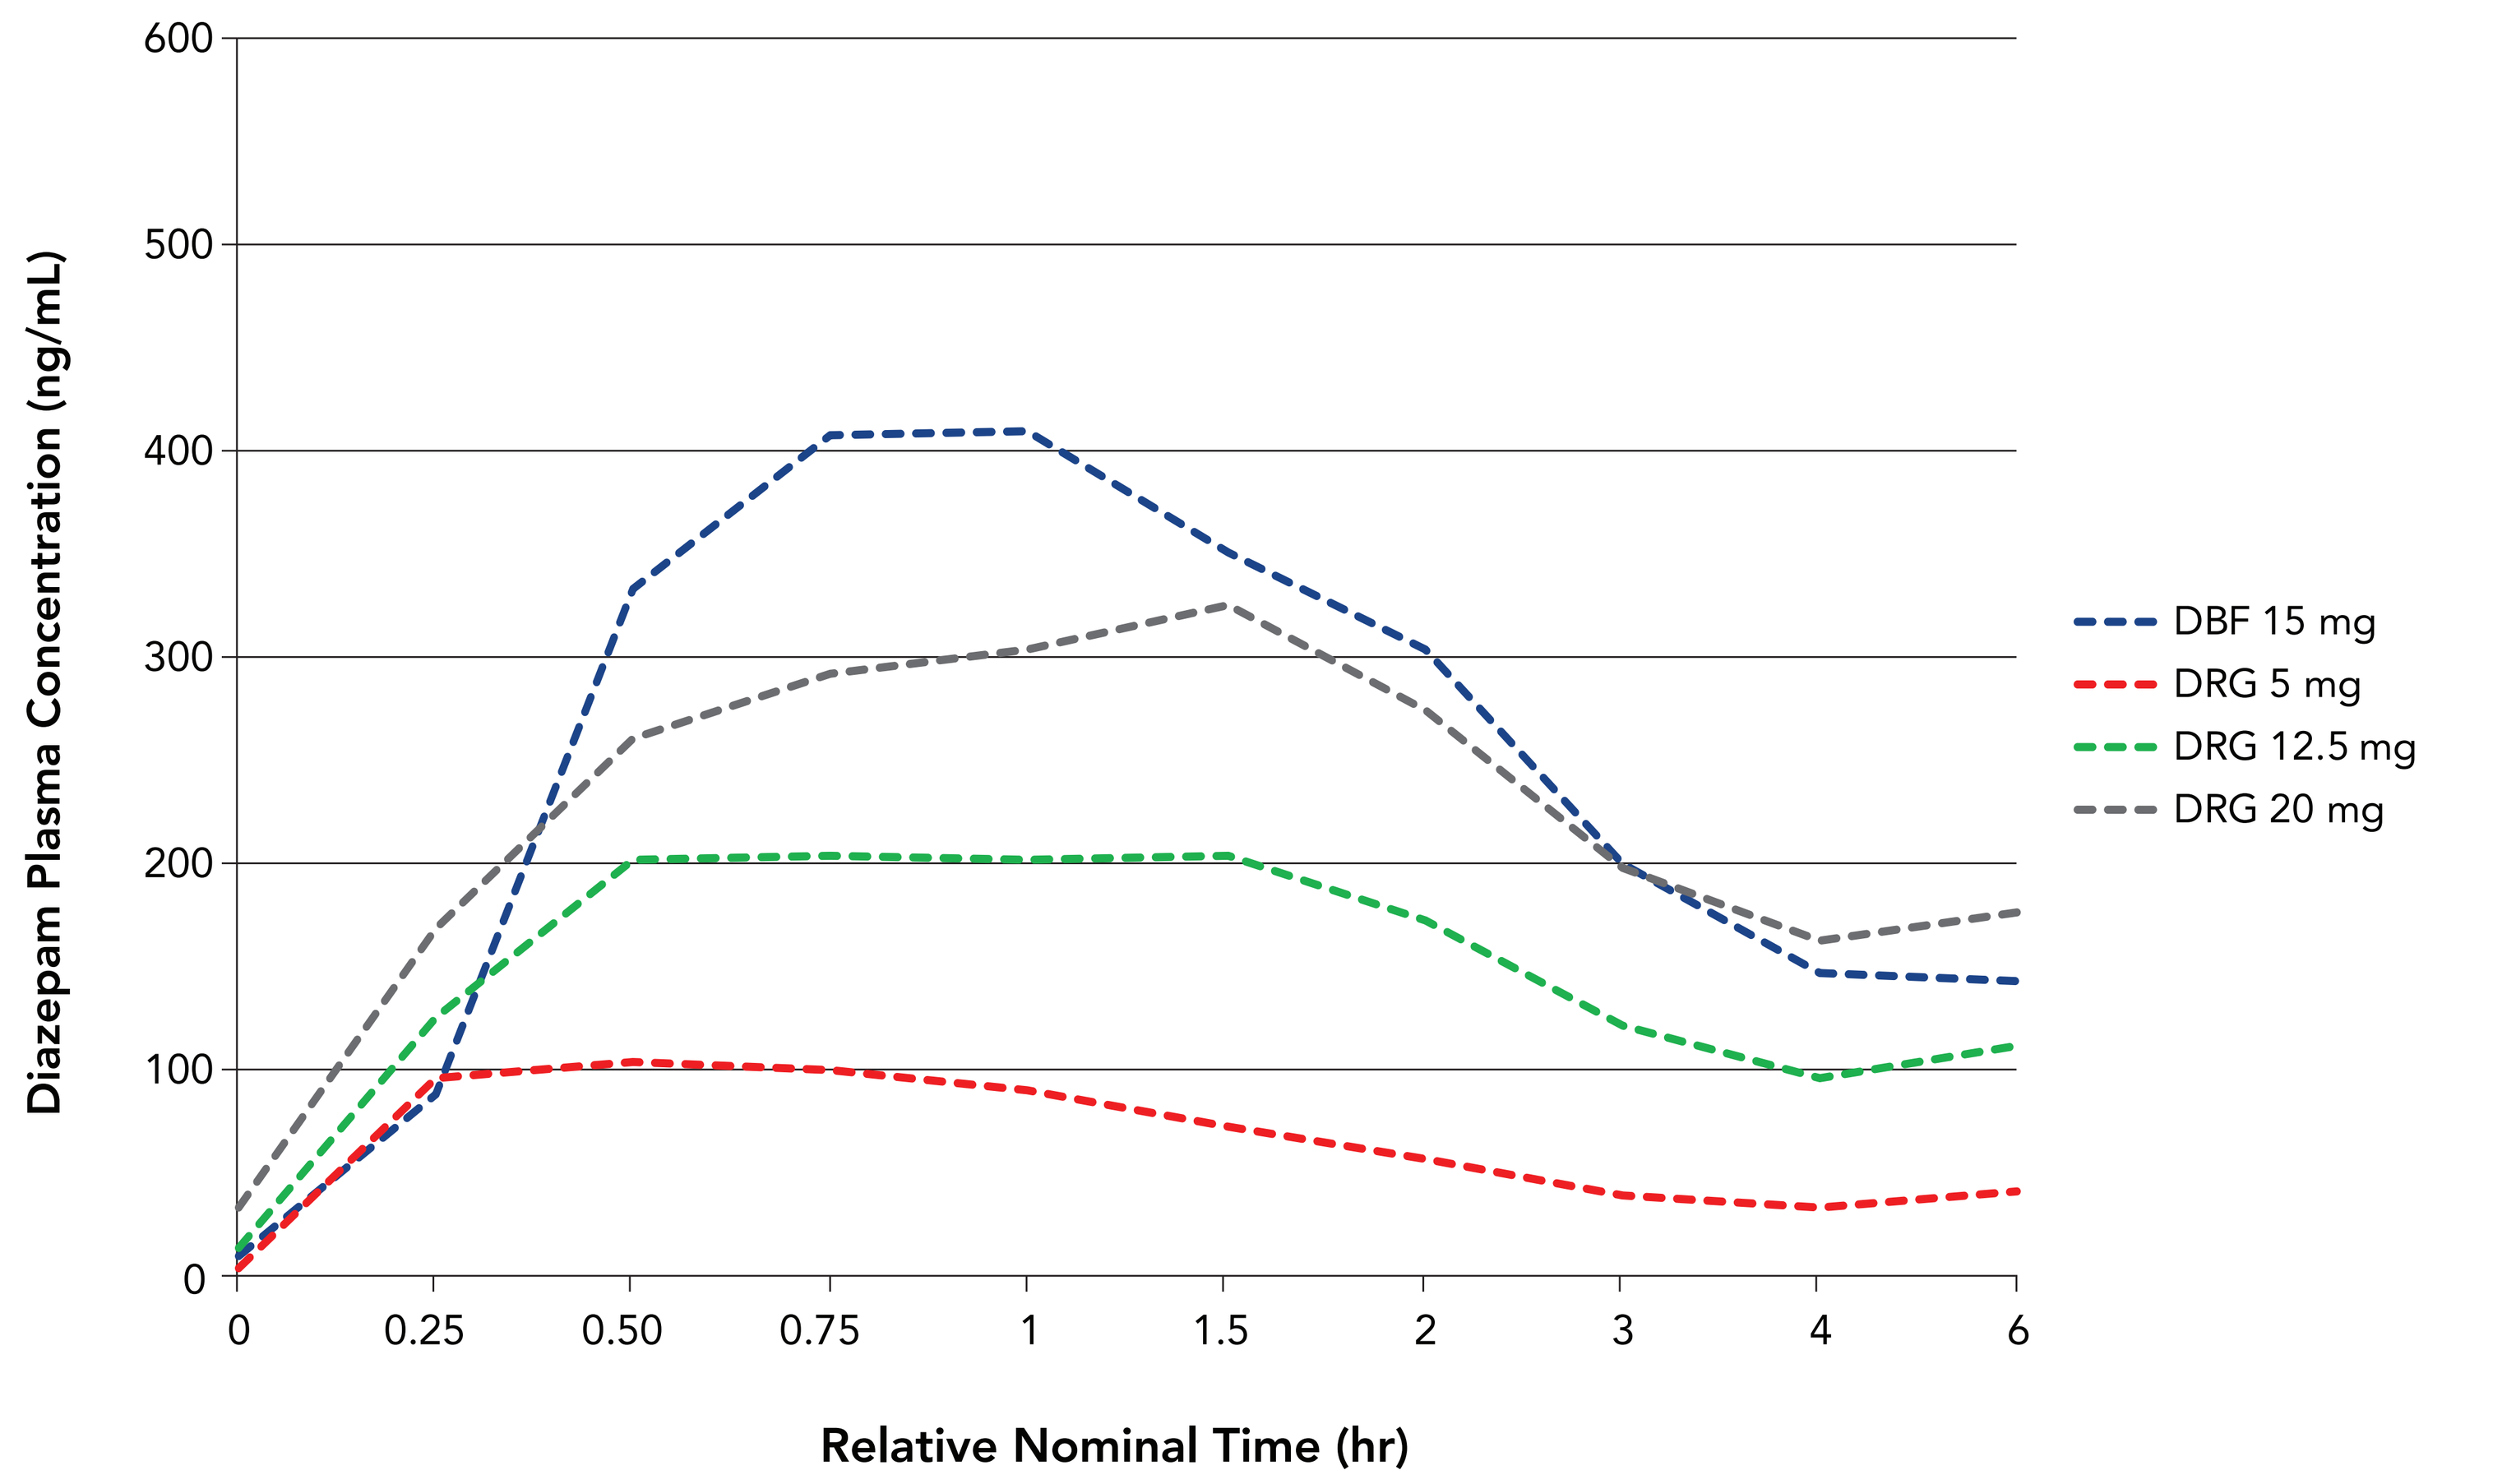
Diazepam Plasma Concentration (ng/mL)
0
100
200
300
400
500
600
0
0.25
0.50
0.75
1
1.5
2
3
4
6
Relative Nominal Time (hr)
DBF 15 mg
DRG 5 mg
DRG 12.5 mg
DRG 20 mg

**Data values plotted for the overall study population represent arithmetic means. DRG 5 mg, n=33; DRG 12.5 mg, n=32, DRG 20 mg, n=31; DBF 15 mg, n=33. DBF, diazepam buccal film; DRG, diazepam rectal gel.*

CONCLUSIONS

- Half the studies identified by the literature review reported a group of subjects with low rectal absorption of diazepam; up to 13.6% of subjects in reports included in the literature analysis showed low rectal absorption of DRG[8,12-15,17]
- These findings were consistent with observations from a recent comparative bioavailability study of DBF and DRG, in which some subjects had extremely low diazepam exposure after DRG administration[7]
- In the comparative bioavailability study, low diazepam exposure was not observed after DBF administration, suggesting that some individuals may be predisposed to be nonabsorbers of rectally administered diazepam[7]

REFERENCES

1. Tatum IW. *Epilepsy Behav.* 2002;3:535-8. **2.** Jafarpour S, et al. *Seizure.* 2019;68:9-15. **3.** Haut SR, et al. *Epilepsy Behav.* 2016;63:109-17. **4.** Diastat C-IV (diazepam rectal gel) [package insert]. San Antonio, TX: DPT Laboratories; 2016. **5.** Hartman AL, et al. *Pediatrics.* 2016;137. **6.** Heller AH, et al. *Neurology.* 2018;90(15 suppl):P4.272. **7.** Heller AH, et al. *Neurology.* 2018;90(15 suppl):P4.273. **8.** Mattila MA, et al. *Br J Anaesth.* 1981;53:1269-72. **9.** Magnussen I, et al. *Acta Pharmacol Toxicol (Copenh).* 1979;45:87-90. **10.** Milligan N, et al. *Epilepsia.* 1982;23:323-31. **11.** Viukari M, et al. *Acta Pharmacol Toxicol (Copenh).* 1981;49:59-64. **12.** Agurell S, et al. *Epilepsia.* 1975;16:277-83. **13.** Ivaturi V, et al. *Epilepsy Res.* 2013;103:254-61. **14.** Dhillon S, et al. *Arch Dis Child.* 1982;57:264-7. **15.** Remy C, et al. *Epilepsia.* 1992;33:353-8. **16.** Cloyd JC, et al. *Epilepsia.* 1998;39:520-6. **17.** Garnett WR, et al. *Epilepsy Res.* 2011;93:11-6. **18.** Heller AH, et al. Presented at: Annual Meeting of the American College of Clinical Pharmacology; September 15-17, 2019; Chicago, IL.

ACKNOWLEDGMENTS

This analysis was sponsored by Aquestive Therapeutics, Inc. Editorial support was provided by Michael Morren, RPh, MBA, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, and funded by Aquestive Therapeutics, Inc.

DISCLOSURES

Gary Slatko is an employee of Aquestive Therapeutics, Inc. Stephen Wargacki is an employee of Aquestive Therapeutics, Inc. David J. Wyatt is an employee of Syneos Health, which received clinical research funding from Aquestive Therapeutics, Inc. Allen H. Heller has served as a paid consultant to Aquestive Therapeutics, Inc.

Presented at: American Epilepsy Society Annual Meeting; December 6–10, 2019; Baltimore, Maryland