Poster 3.444

SAFETY AND TOLERABILITY ASSOCIATED WITH CHRONIC INTERMITTENT USE OF DIAZEPAM BUCCAL FILM IN PEDIATRIC, ADOLESCENT, AND ADULT PATIENTS WITH EPILEPSY

Syndi Seinfeld[1], Michael A. Gelfand[2], Allen H. Heller[3], Carla Buan[4], Gary Slatko[4]

[1]Virginia Commonwealth University, Richmond, VA; [2]University of Pennsylvania, Philadelphia, PA; [3]Pharma Study Design, LLC, Woodbridge, CT; [4]Aquestive Therapeutics, Warren, NJ

INTRODUCTION

- Seizure clusters and other bouts of more frequent or more severe seizures occur in many patients with epilepsy despite treatment with antiepileptic medications, and can lead to injury, hospitalizations, status epilepticus, and risk of death[1-3]
- Available treatment options for these episodes are suboptimal, particularly in terms of speed of onset of action and ease of administration[2,4,5]
- Currently, diazepam rectal gel and midazolam nasal spray are the only treatments approved by the United States Food and Drug Administration for refractory patients with epilepsy experiencing bouts of increased seizure activity[6,7]
 - Rectal administration may be difficult, time consuming, and embarrassing for patients and/or caregivers, which may affect adherence[1,5,8,9]
 - Intranasal administration may increase mucosal secretions, which can be associated with unpredictable absorption[5]
- Diazepam buccal film (DBF) is a novel formulation of diazepam in development that is designed to be easily administered,[10] with a more consistent, predictable pharmacokinetic profile compared with rectally administered diazepam[11]
- In studies conducted to date, single doses of DBF 5 mg to 20 mg have been shown to be safe and well tolerated in healthy adults and adults with epilepsy[11-13]

OBJECTIVES

- The primary objective of this ongoing study (NCT03428360) is to assess the safety and tolerability of self- or caregiver-administered DBF in people with epilepsy in the outpatient setting
 - The usability of DBF is being assessed as a secondary objective

METHODS

STUDY DESIGN

- This is an ongoing, phase 3, multicenter, open-label, long-term safety and tolerability study; interim data from patients receiving ≥1 dose of DBF as of May 2019 are reported here
- An overview of the study schema is shown in **Figure 1**

Figure 1. Study Schema

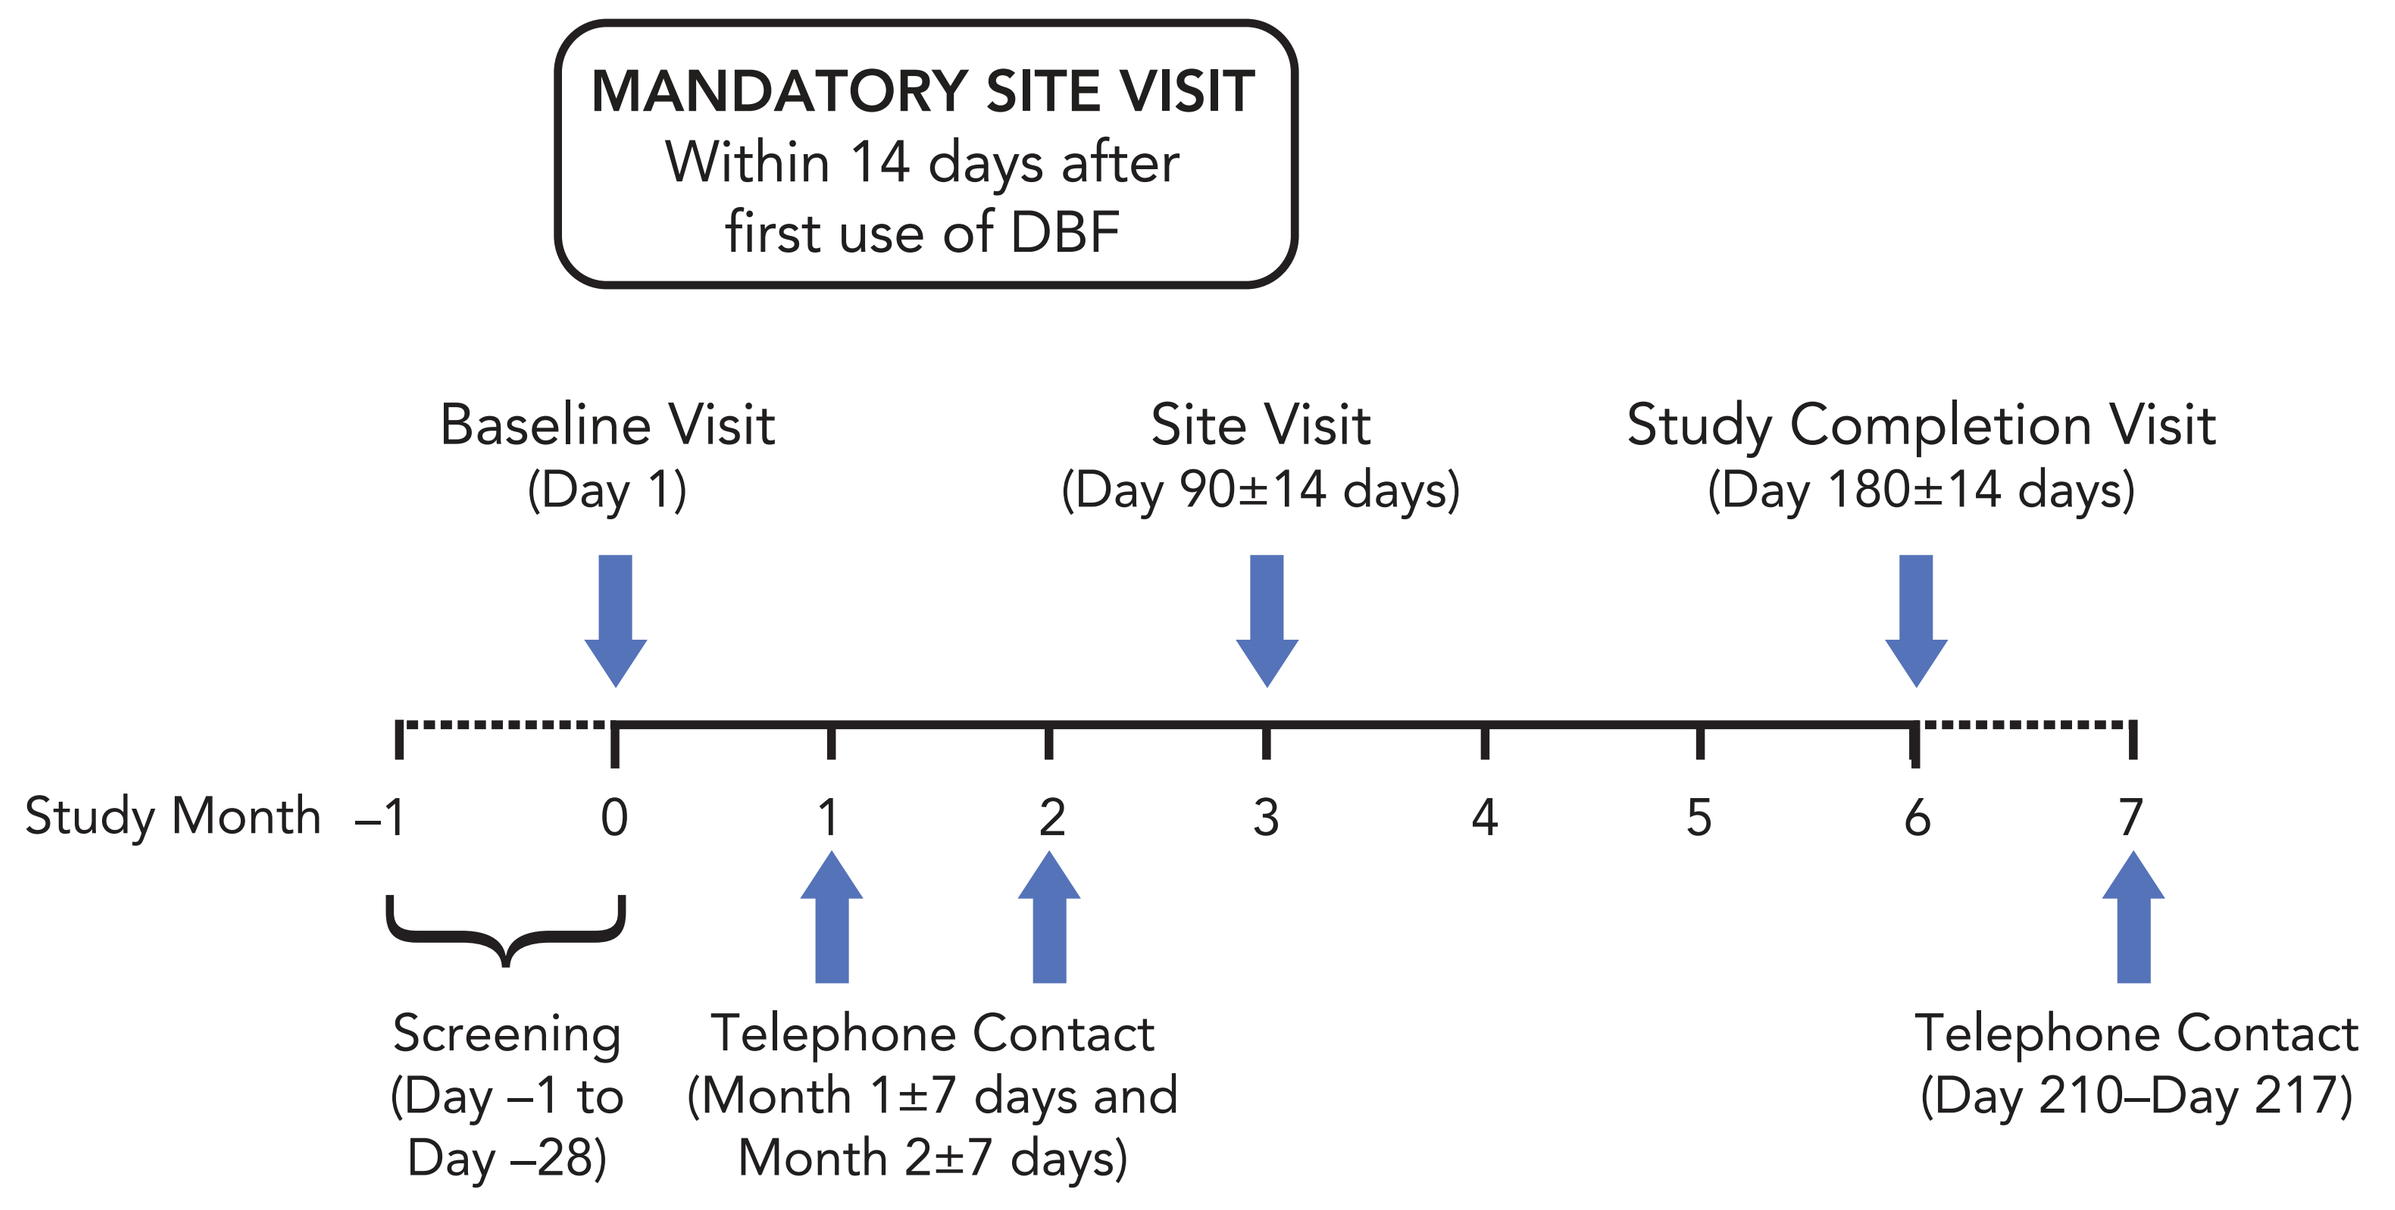
MANDATORY SITE VISIT
Within 14 days after
first use of DBF
Baseline Visit
(Day 1)
Site Visit
(Day 90±14 days)
Study Completion Visit
(Day 180±14 days)
Study Month
–1
0
1
2
3
4
5
6
7
Screening
(Day –1 to
Day –28)
Telephone Contact
(Month 1±7 days and
Month 2±7 days)
Telephone Contact
(Day 210–Day 217)

DBF, diazepam buccal film.

PATIENTS

- Eligible patients include males and females between the ages of 2 and 65 years, inclusive, with an established diagnosis of epilepsy who were deemed to need benzodiazepine treatment for bouts of increased seizures at least once monthly on average
- Patients are ineligible if not treated with at least 1 antiepileptic medication or if they have a history of clinically significant disease or any other clinically significant abnormalities that could, in the opinion of the investigator, interfere with study procedures or the safety of the patient

PROCEDURES AND ASSESSMENTS

- Informed consent was obtained prior to study entry
- Patients and caregivers were trained on how to administer DBF and use of an electronic diary to document seizures, use of DBF, adverse events (AEs), and changes in concomitant medications during the baseline site visit on study day 1; these procedures were reviewed at all subsequent study visits and contacts
 - Study participants were instructed to administer DBF for treatment of seizure episodes occurring in their home environments in which a benzodiazepine would ordinarily be administered for seizure rescue
- DBF was dispensed at weight- and age-based doses ranging from 5 to 17.5 mg, which could be adjusted with aging or change in body weight during the study. Modest adjustments to dose could also be made clinically depending on initial response (**Table 1**)

TABLE 1. DOSING ALGORITHM

Age/Weight Category	DBF Dose (mg)[a]
Age 2–5 years	
6–10 kg	5
11–20 kg	7.5
21–25 kg	10
26–44 kg	12.5
Age 6–11 years	
10–16 kg	5
17–33 kg	7.5
34–41 kg	10
42–74 kg	12.5
Age 12–16 years	
14–25 kg	5
26–50 kg	7.5
51–62 kg	10
63–111[b] kg	12.5
Age ≥17 years	
14–25 kg	5
26–37 kg	7.5
38–50 kg	10
51–62 kg	12.5
63–87 kg	15
88–111[b] kg	17.5

[a]Where there is overlap in body weight with age, the age-adjustment takes precedence for dose administered. Investigators may make dose adjustments as medically indicated.
[b]Patients with body weight in the range 112–134 kg may participate in the study at the discretion of the investigator.

- DBF could be administered for up to 5 seizure episodes per month; administration of a second dose was permitted within 4 to 12 hours after the first dose
- Adjustments to patients' baseline antiepileptic medication regimens were allowed as per standard clinical practice

OUTCOMES OF INTEREST FOR THE CURRENT ANALYSIS

- Treatment emergent AEs (including severity and relationship to study drug) occurring during the course of the study or up to 30 days after last study drug administration or until all drug-related toxicities have resolved, whichever is later
- DBF administration and usability, as assessed by patients and/or caregivers, recorded after each use of study drug, including successful placement/buccal insertion of DBF, oral cavity retention of DBF, and ability to open packaging and remove DBF

DATA ANALYSIS

- All data analyses for this interim report were conducted using the safety population, which includes all patients who received ≥1 dose of DBF
- Data are summarized using descriptive statistics with no formal statistical testing performed

RESULTS

STUDY PATIENTS

- As of May 31, 2019, a total of 72 patients have enrolled and used DBF at least once (adults, n=59; adolescents, n=7; pediatric, n=6)
 - Of the 72 total patients, 53 (73.6%) continue to be actively enrolled in the study, 14 (19.4%) have completed the study, and 5 (6.9%) have withdrawn
- The mean (SD) duration of time on study at the cutoff date for this interim analysis was 192.0 (97.0) days
- The mean (SD) number of DBF doses administered was 8.7 (10.1), with a median (range) of 5 (1–49) doses
- Patient demographics and baseline characteristics are shown in **Table 2**

TABLE 2. PATIENT DEMOGRAPHICS AND BASELINE CHARACTERISTICS

Characteristic	Adults (n=59)	Adolescents (n=7)	Pediatric (n=6)
Age, mean (SD), years	31.9 (10.5)	14.1 (1.5)	7.5 (2.7)
Female, n (%)	27 (45.8)	6 (85.7)	2 (33.3)
Race, n (%)			
White	45 (76.3)	4 (57.1)	4 (66.7)
Black or African American	4 (6.8)	2 (28.6)	1 (16.7)
Asian	4 (6.8)	1 (14.3)	0
Native Hawaiian or Other Pacific Islander	5 (8.5)	0	0
Other[a]	1 (1.7)	0	1 (16.7)
Ethnicity, n (%)			
Hispanic or Latino	12 (20.3)	2 (28.6)	3 (50.0)
Not Hispanic or Latino	45 (76.3)	5 (71.4)	3 (50.0)
Unknown	2 (3.4)	0	0
Height (cm), mean (SD)	162.8 (13.1)[b]	144.9 (15.7)	118.5 (16.1)
Weight (kg), mean (SD)	69.4 (19.9)	46.1 (11.5)	23.0 (7.4)
BMI (kg/m^2), mean (SD)	26.0 (6.4)	22.1 (4.5)	16.1 (3.0)
Duration of epilepsy (years), mean (SD)	20.2 (11.7)[c]	12.3 (4.2)[d]	6.6 (3.0)[e]

[a]Not white, black or African American, native Hawaiian or other Pacific Islander, or American Indian or Alaska native.
[b]n=58; [c]n=44; [d]n=4; [e]n=5.
BMI, body mass index; SD, standard deviation.

DBF SAFETY

- An overview of AEs is shown in **Table 3**
 - No pediatric patient has reported an AE
 - Overall, 13 serious AEs were reported by 9 (12.5%) patients; none were considered to be related to treatment
 - No patient discontinued study participation due to an AE

TABLE 3. SUMMARY OF ADVERSE EVENTS

Parameter	Pediatric (n=6)	Adolescent (n=7)	Adult (n=59)	Total (N=72)
	Number (%) of Patients, Number of Events			
Any AE	0	5 (71.4), 10	29 (49.2), 80	34 (47.2), 90
Any serious AE	0	2 (28.6), 2	7 (11.9), 11	9 (12.5), 13
Any severe AE	0	0	5 (8.5), 11	5 (6.9), 11
Any treatment-related AE[a]	0	1 (14.3), 1	4 (6.8), 6	5 (6.9), 7
Discontinued due to AE	0	0	0	0
Death	0	0	0	0

[a]Defined as an AE categorized as having a "possible" or "probable" relationship to the study drug. AE, adverse event.

- AEs reported in >2% of patients are shown in **Table 4**
 - The most frequently reported AE was seizure; a total of 6 seizure events were reported in 4 (5.6%) patients overall

TABLE 4. ADVERSE EVENTS REPORTED IN >2% OF THE OVERALL POPULATION

AE[a]	Overall Population (N=72)
Seizure	4 (5.6), 6
Pyrexia	4 (5.6), 4
Dizziness	3 (4.2), 3
Nausea	3 (4.2), 3
Somnolence	3 (4.2), 3
Weight decreased	3 (4.2), 3
Cough	2 (2.8), 2
Dehydration	2 (2.8), 2
Gastroenteritis viral	2 (2.8), 2
Lethargy	2 (2.8), 2
Vomiting	2 (2.8), 2
Weight increased	2 (2.8), 2

[a]All data are reported as number (%) of patients, number of events. AE, adverse event.

- Seven treatment-related AEs occurred in 5 (6.9%) patients (**Table 5**)
 - All treatment-related AEs were mild in severity and resolved without sequelae
 - Local buccal discomfort was reported in 1 adult patient and resolved within 1 day
 - There were no reports of injury during buccal placement of DBF

TABLE 5. TREATMENT-RELATED[a] ADVERSE EVENTS

Treatment-related AEs[b]	Pediatric (n=6)	Adolescent (n=7)	Adult (n=59)	Total (N=72)
	Number (%) of Patients, Number of Events			
Somnolence	0	0	1 (1.7), 1	1 (1.4), 1
Lethargy	0	0	1 (1.7), 1	1 (1.4), 1
Altered state of consciousness	0	0	1 (1.7), 1	1 (1.4), 1
Mouth swelling	0	1 (14.3), 1	0	1 (1.4), 1
Oral discomfort	0	0	1 (1.7), 1	1 (1.4), 1
Gait disturbance	0	0	1 (1.7), 1	1 (1.4), 1
Skin sensitization	0	0	1 (1.7), 1	1 (1.4), 1

[a]Defined as an AE categorized as having a "possible" or "probable" relationship to the study drug.
[b]All treatment-related AEs were mild in severity.

DBF USABILITY

- DBF usability data were reported by 64 of the 72 (88.9%) patients, representing a total of 471 DBF use occasions with a mean (SD) of 7.4 (7.7) administrations per patient
- DBF was successfully administered during a first attempt on 443 of 471 (94.1%) use occasions (**Figure 2**) and was ultimately successfully placed in all but 2 (0.4%) use occasions
 - All 64 patients had first-attempt success at DBF administration on at least one use occasion

Figure 2. Success Rates for DBF Insertion Stratified by Number of Attempts

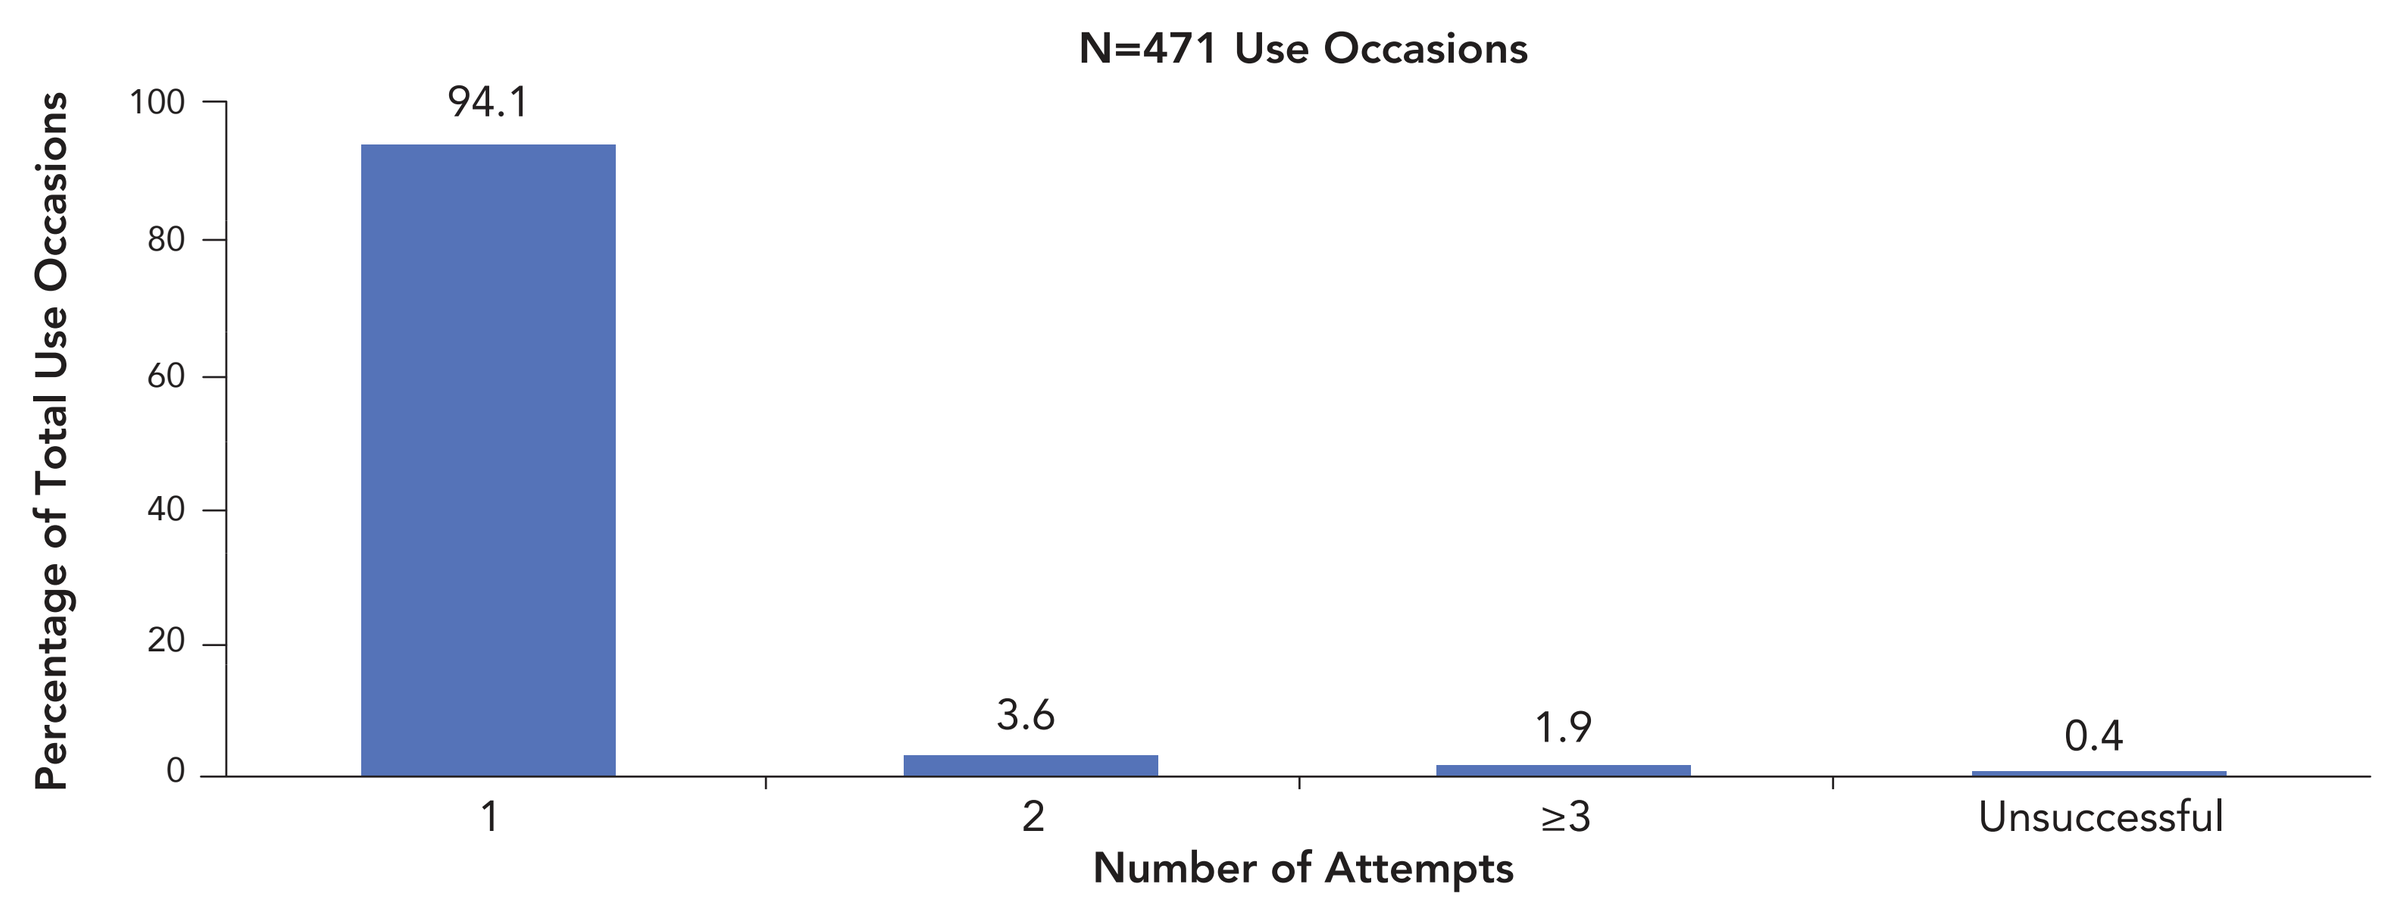
N=471 Use Occasions
Percentage of Total Use Occasions
100
80
60
40
20
0
94.1
3.6
1.9
0.4
1
2
≥3
Unsuccessful
Number of Attempts

- Reasons for unsuccessful placement among patients with ≥1 unsuccessful attempt are summarized in **Table 6**
 - There were no incidents of unsuccessful placement due to DBF being swallowed before it adhered to the buccal mucosa

TABLE 6. OVERVIEW OF REPORTED REASONS FOR UNSUCCESSFUL PLACEMENT OF DBF

Reasons for Unsuccessful Insertion Attempts	Unsuccessful Attempts Based on 471 Use Occasions, n (%)[a]
Clenching jaw/won't open mouth	10 (2.1)
Excessive drooling	9 (1.9)
Spit out before DBF adhered to buccal mucosa	7 (1.5)
Clenching jaw/won't open mouth/excessive drooling	1 (0.2)
Other	8 (1.7)

[a]Respondents could choose more than 1 reason for an unsuccessful insertion attempt; 35 reasons were given for 28 unsuccessful attempts.

- Patients and caregivers reported no difficulty opening the outer and inner packaging in 78.3% and 67.7% of total use occasions, respectively
- Patients and caregivers reported no difficulty removing DBF from the inner packaging in nearly all (96.6%) use occasions
- Patient self-administration was recorded on 107 of 471 use occasions (22.7%), and 308 use occasions (65.4%) were under ictal/periictal conditions

CONCLUSIONS

- In this observational study of chronic, intermittent administration and use, DBF was found to be safe and well tolerated by pediatric, adolescent, and adult patients with epilepsy experiencing seizure emergencies
- Treatment-related AEs, including somnolence, were relatively uncommon following DBF use, and all treatment-related AEs were mild in severity
- DBF was ultimately successfully placed on nearly all (99.6%) use occasions and readily used without difficulty when administered by patients and caregivers
- Patient self-administration of DBF is feasible, and ictal/periictal use is commonly observed
- These results support further development and use of DBF as an easily administered alternative to diazepam rectal gel for patients with epilepsy who experience cluster seizures and other bouts of more frequent or more severe seizures despite treatment with antiepileptic medications

REFERENCES

1. Jafarpour S, et al. *Seizure.* 2019;68:9-15. **2.** Penovich PE, et al. *Neurologist.* 2017;22:207-14. **3.** Sillanpaa M, Schmidt D. *Brain.* 2008;131:938-44. **4.** Tatum IW. *Epilepsy Behav.* 2002;3:535-8. **5.** Cereghino JJ. *Curr Treat Options Neurol.* 2007;9:249-55. **6.** Diastat C-IV (diazepam rectal gel) [package insert]. San Antonio, TX: DPT Laboratories; 2016. **7.** Nayzilam [package insert]. Smyrna, GA: UCB, Inc.; 2019. **8.** Fisgin T, et al. *J Child Neurol.* 2002;17:123-6. **9.** Haut SR, et al. *Epilepsy Behav.* 2016;63:109-17. **10.** Jung C, et al. Presented at: Annual Meeting of the American Epilepsy Society; November 30-December 4, 2018; New Orleans, LA. **11.** Heller AH, et al. *Neurology.* 2018;90(15 suppl):P4.273. **12.** Heller AH, et al. *Neurology.* 2018;90(15 suppl):P4.272. **13.** Rogawski MA, et al. Presented at: Annual Meeting of the American Epilepsy Society; November 30-December 4, 2018; New Orleans, LA.

ACKNOWLEDGMENTS

Sponsored by Aquestive Therapeutics, Inc. Editorial support was provided by Michael Morren, RPh, MBA, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, and funded by Aquestive Therapeutics, Inc.

DISCLOSURES

Syndi Seinfeld is a member of the speaker bureau for LivaNova PLC and has served as a consultant for Aquestive Therapeutics, Inc. Michael A. Gelfand has received research support from Aquestive Therapeutics, Inc., Engage Therapeutics, Inc., UCB, Inc., Biogen, Eisai Co., Ltd., LivaNova PLC, Otsuka Pharmaceutical Co., Ltd., and SK Pharma. Allen H. Heller has served as a paid consultant to Aquestive Therapeutics, Inc. Carla Buan and Gary Slatko are employees of Aquestive Therapeutics, Inc.

Presented at: American Epilepsy Society Annual Meeting; December 6–10, 2019; Baltimore, Maryland