Poster 1.432

PHARMACOKINETICS OF DIAZEPAM BUCCAL FILM IN ADULT PATIENTS WITH EPILEPSY: COMPARISON WITH DIAZEPAM RECTAL GEL

Michael A. Rogawski[1], Allen H. Heller[2], Simon Farrow[3], Cassie Jung[4], Pavel Klein[5], Sylvie Boudreault[6], Gary Slatko[4]

[1]University of California, Davis, Sacramento, CA; [2]Pharma Study Design, LLC, Woodbridge, CT; [3]Clinical Research Center of Nevada, Las Vegas, NV; [4]Aquestive Therapeutics, Warren, NJ; [5]Mid-Atlantic Epilepsy and Sleep Center, Bethesda, MD; [6]Syneos Health, Quebec, Canada

INTRODUCTION

- Patients with refractory epilepsy may experience seizure exacerbations referred to as cluster seizures, which represent a series of seizures grouped consecutively, typically with short (or shorter than usual) interictal periods
- Cluster seizures raise concerns for seizure-associated risks, including postictal psychosis, physical injury, negative social and economic impact from frequent emergency department visits, hospitalizations, or missed school or work days, and for status epilepticus that may lead to persistent neurological impairment or death[1-7]
- Despite these increased risks, many patients with epilepsy do not have a rescue medication prescribed and/or a seizure action plan for when cluster seizures occur[1,8-10]
- Diazepam buccal film (DBF) is a novel dosage form of diazepam under development for the management of patients with refractory epilepsy requiring intermittent use of diazepam to control increased seizure activity[11,12]
 - DBF consists of a rectangular section of film smaller than the size of a postage stamp, which is affixed to the buccal mucosa inside the cheek
 - Diazepam carried within the dissolving polymer matrix of the film is absorbed transmucosally and is also swallowed
- Currently, diazepam rectal gel (DRG; Diastat® AcuDial™ rectal gel, Valeant Pharmaceuticals, Bridgewater, NJ, USA) and midazolam nasal spray (Nayzilam® nasal spray, UCB Biopharma, Smyrna, GA, USA) are the only FDA-approved treatments for seizure emergencies[2,13-16]; these current treatments have certain limitations:
 - Rectal administration can be difficult and time consuming, and is otherwise problematic for many patients and caregivers due to concerns regarding embarrassment and social acceptability[13,17,18]
 - Intranasal administration is often poorly accepted by patients, which can negatively impact compliance
- Pharmacokinetic (PK) data from a phase 1 study in healthy adults under fasted conditions showed DBF to exhibit greater overall consistency in diazepam exposure (area under the concentration-time curve [AUC], maximal plasma concentration [C_{max}]) than DRG[12]

OBJECTIVE

- This study was conducted to compare the PK performance in people with epilepsy of DBF in relation to DRG under fed conditions with DBF administered according to its proposed dosing regimen and DRG administered according to its labeled dosing regimen

METHODS

STUDY DESIGN AND PATIENTS

- Randomized, multicenter, single-dose, open-label, two-treatment, two-sequence crossover study (NCT03953820)
 - Adult patients with epilepsy receiving a stable regimen of antiseizure drugs were randomized to receive a single administration of DBF and a single administration of DRG in crossover fashion
 - There was a 28-day washout phase between doses of study drug
 - DRG was dosed according to the weight-based regimen in the FDA-approved label (dose range 10–20 mg) and DBF was dosed according to a weight-based regimen (dose range 10–17.5 mg) predicted to approximate the PK performance of DRG with respect to C_{max} (**Table 1**)
 - Both treatments were administered following a moderate-fat meal

TABLE 1. STUDY DRUG DOSING BY PATIENT WEIGHT CATEGORY

Weight Category (kg)	DBF Dose	DRG Dose[a]
38–50	10 mg	10 mg (2 mL)
51–62	12.5 mg	12.5 mg (2.5 mL)
63–75	15 mg	15 mg (3 mL)
76–87	15 mg	17.5 mg (3.5 mL)
≥88	17.5 mg	20 mg (4 mL)

[a]DRG was administered according to the weight-based regimen recommended in the FDA-approved label.[16]

- For each dose of study drug, patients were confined to the clinic from approximately 14 hours before dosing until after the 24-hour post-dose blood draw
 - For blood draws after 24 hours post-dose, patients could return to the clinical site or have a sample collected by a home-care nurse
 - While confined in the clinic, patients fasted overnight for at least 10 hours before being served a standardized moderate-fat meal

STUDY ASSESSMENTS

- In each period, blood samples for PK analyses were obtained pre-dose and 0.5, 0.75, 1, 1.5, 2, 3, 4, 6, 9, 12, 24, 48, 72, 96, 120, 144, 192, and 240 hours post-dose to enable PK assessments
 - A window of ±1 minute was allowed for blood samples obtained within the first 8 hours post-dose, ±3 minutes for samples obtained within 8 to 24 hours post-dose, and ±60 minutes for all subsequent blood samples
- Key PK parameters of interest included C_{max}, time to C_{max} (T_{max}), AUC from time zero to the last non-zero concentration ($AUC_{0\text{-}T}$), and AUC from time zero extrapolated to infinity ($AUC_{0\text{-}INF}$)
- Adverse events (AEs) were monitored throughout the study

DATA ANALYSIS

- The PK population included all patients who completed period 1 and period 2, had no significant violations of the study protocol, and for whom the PK profile could be adequately characterized
- PK data were summarized overall and for each weight category using descriptive statistics
- ANOVA was performed on log-transformed $AUC_{0\text{-}T}$, $AUC_{0\text{-}INF}$, and C_{max} at alpha 0.05. Factors in the model were sequence, subject within sequence, period, and treatment. T_{max} was analyzed using the non-parametric Wilcoxon signed-rank test
- The ratios of geometric means with 90% confidence intervals based on least-squares means from ANOVA of log-transformed data were calculated for C_{max}, $AUC_{0\text{-}T}$, and $AUC_{0\text{-}INF}$ for DBF versus DRG values for all subjects irrespective of weight category, and for subjects within each weight category

RESULTS

PATIENTS

- Among 31 patients enrolled, PK profiles valid for analysis for both DBF and DRG were available for 28 subjects (13 males, 15 females; mean [SD] weight: 84.6 [20.6 kg])
 - Patients were excluded from PK analyses if both DBF and DRG treatments were not completed (n=2), or if pre-dose diazepam concentrations were >5% of C_{max} (n=1)
- Diazepam mean (SD) doses were 15.4 (1.9) mg and 17.1 (3.0) mg for DBF and DRG, respectively

PHARMACOKINETICS RESULTS

- Geometric means for PK parameters with ratios of the geometric means (DBF/DRG) for the overall study population (N=28 for C_{max} and T_{max}; N=27 for $AUC_{0\text{-}T}$ and $AUC_{0\text{-}INF}$) are shown in **Table 2**
 - The ratio of geometric mean C_{max} values for the study population (DBF/DRG) was 96.7%, demonstrating that diazepam peak plasma concentrations for DBF and DRG were similar
 - $AUC_{0\text{-}T}$ and $AUC_{0\text{-}INF}$ values were higher for DBF than for DRG
 - In this study, T_{max} values for both DBF and DRG were smaller than 1.5 hours, the value reported in DRG (Diastat) labeling
 - Three of 28 subjects following DRG dosing failed to achieve a plasma concentration ≥70 ng/mL (70 ng/mL is the estimated threshold plasma concentration of diazepam associated with seizure threshold elevation as determined by a pharmacodynamic-PK study in rats[19])

TABLE 2. PHARMACOKINETIC PARAMETERS FOLLOWING DBF AND DRG ADMINISTERED TO ADULTS WITH EPILEPSY FOLLOWING A MODERATE-FAT MEAL IN THE OVERALL STUDY POPULATION (N=28)

Parameter	DBF	DRG	Ratio of Geometric Means, DBF/DRG (%)[a]	90% CI (%) for Ratio[b]
C_{max} (ng/mL), geometric mean	204.26	211.22	96.70	70.53, 132.58
$AUC_{(0\text{-}T)}$ (ng•h/mL), geometric mean[c]	7290.40	5682.09	128.31	95.93, 171.61
$AUC_{(0\text{-}INF)}$ (ng•h/mL), geometric mean[c]	8672.09	6880.96	126.03	103.67, 153.21
T_{max} (h), median	1.0	0.517[d]	NA	NA

[a]Calculated using least-square means according to the formula $e^{(Difference)}$ x 100. [b]90% geometric confidence interval using log-transformed data. [c]N=27. [d]P<0.05 vs DBF.

- Geometric mean diazepam plasma concentrations over time in the overall study population following DBF and DRG are illustrated in **Figure 1**
 - For the study population overall, geometric mean C_{max} values following DBF and DRG administration were 204.26 ng/mL (geometric SD [GSD]: 136.12–306.49) and 211.22 ng/mL (GSD 87.71–508.63), respectively, indicating that C_{max} values following DBF were comparable but significantly less variable than C_{max} values following DRG (*P*<0.0001) (**Table 2** and **Figure 1 inset**)

Figure 1. Geometric Mean (Geometric SE) Diazepam Plasma Concentrations Over Time Following Administration of DBF and DRG in the Overall Study Population (N=28)

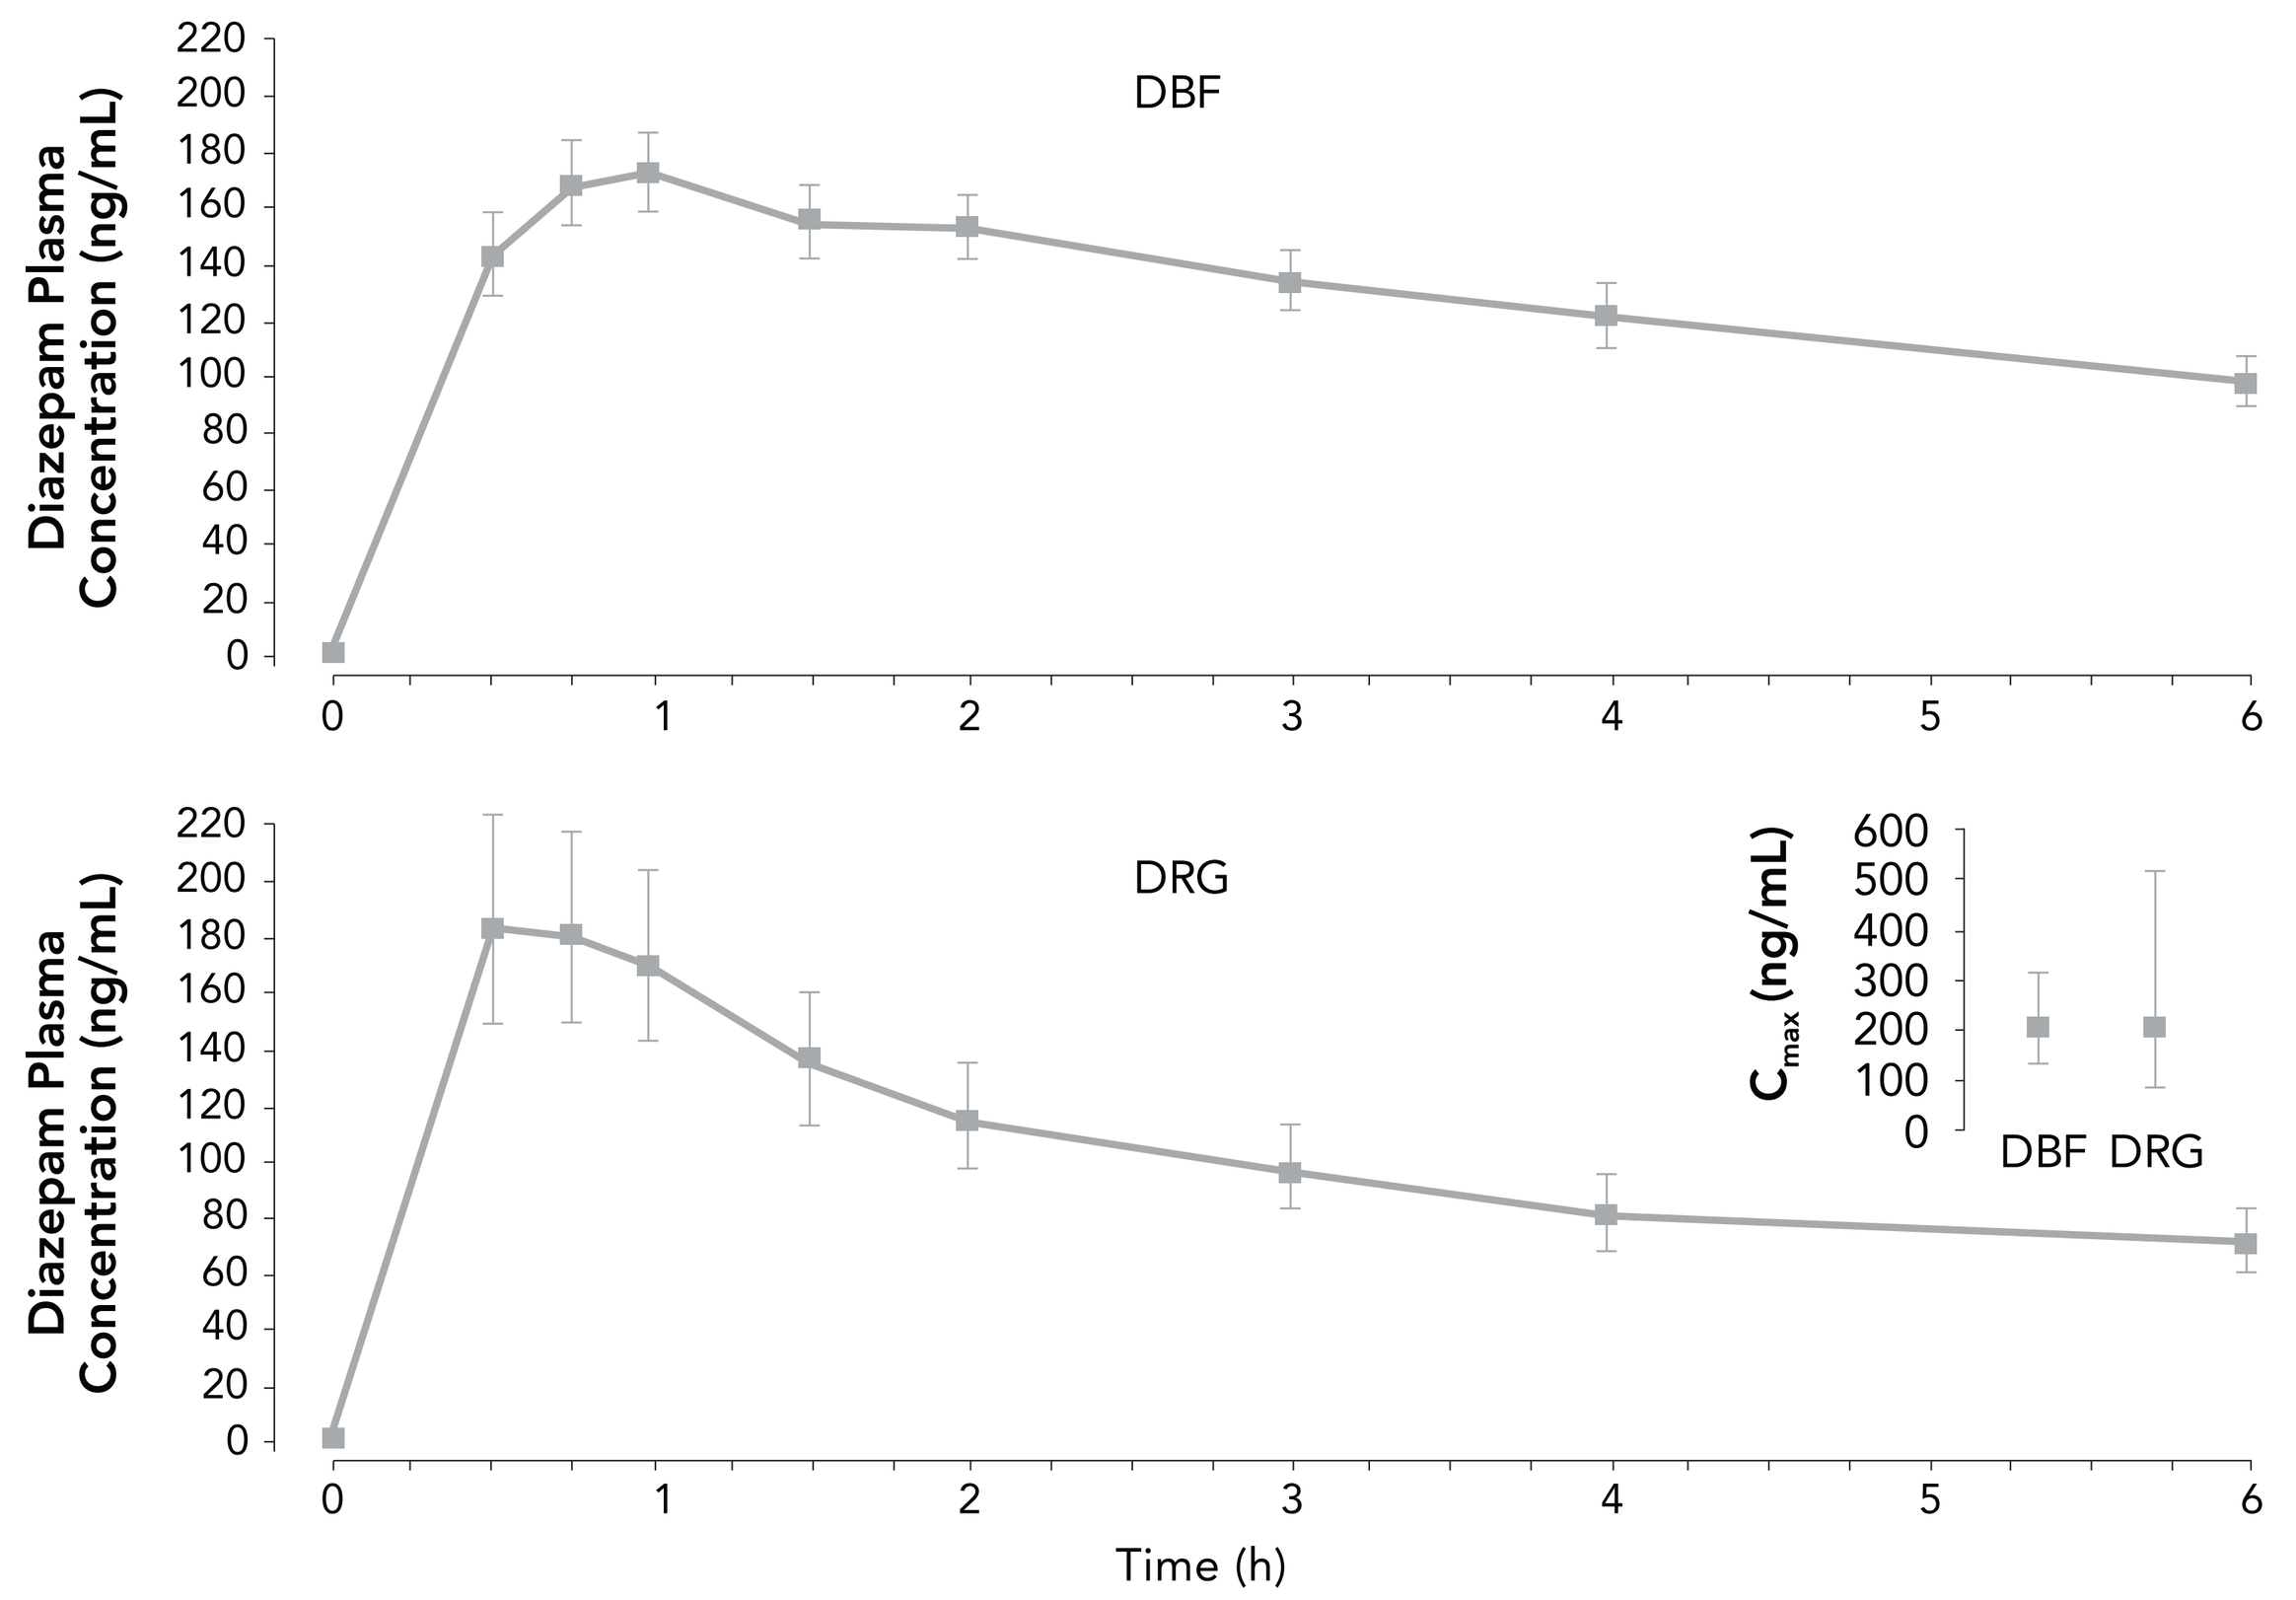

DBF
DRG
Diazepam Plasma Concentration (ng/mL)
Time (h)
C_{max} (ng/mL)
DBF DRG

Values graphed are geometric mean (geometric SE) plasma concentrations. Inset shows geometric mean (geometric SD) C_{max} values.
C_{max}, maximum observed plasma drug concentration; DBF, diazepam buccal film; DRG, diazepam rectal gel; SD, standard deviation; SE, standard error.

- Geometric means for C_{max} and corresponding ratios within each weight category are shown in **Table 3**
 - C_{max} values were less variable with DBF versus DRG (**Figure 2**)

TABLE 3. DBF AND DRG MAXIMAL PLASMA CONCENTRATIONS (C_{MAX}) ACCORDING TO WEIGHT CATEGORY (N=28)

C_{max} Geometric Mean by Weight Group	DBF	DRG	Ratio of C_{max} Geometric Means DBF/DRG (%)[a]	90% CI (%) for Ratio[b]
51-62 kg (n=6)				
C_{max} (ng/mL)	258.38	358.06	72.16	51.17, 101.76
Dose (mg)	12.5	12.5	—	—
63-75 kg (n=4)				
C_{max} (ng/mL)	234.45	258.88	90.56	27.89, 295.09
Dose (mg)	15.0	15.0	—	—
76-87 kg (n=7)				
C_{max} (ng/mL)	201.39	293.00	68.74	46.77, 101.01
Dose (mg)	15.0	17.5	—	—
≥88[c] (n=11)				
C_{max} (ng/mL)	175.56	115.82	151.58	71.59, 320.94
Dose (mg)	17.5	20.0	—	—

[a]Calculated using least-square means according to the formula $e^{(Difference)}$ x 100. [b]90% geometric confidence interval using log-transformed data. [c]The highest weight category included 4 individuals with body weight 112–124.5 kg.
CI, confidence interval; C_{max}, maximum observed plasma drug concentration; DBF, diazepam buccal film; DRG, diazepam rectal gel.

Figure 2. C_{max} (Geometric Mean) by Weight Group (N=28)

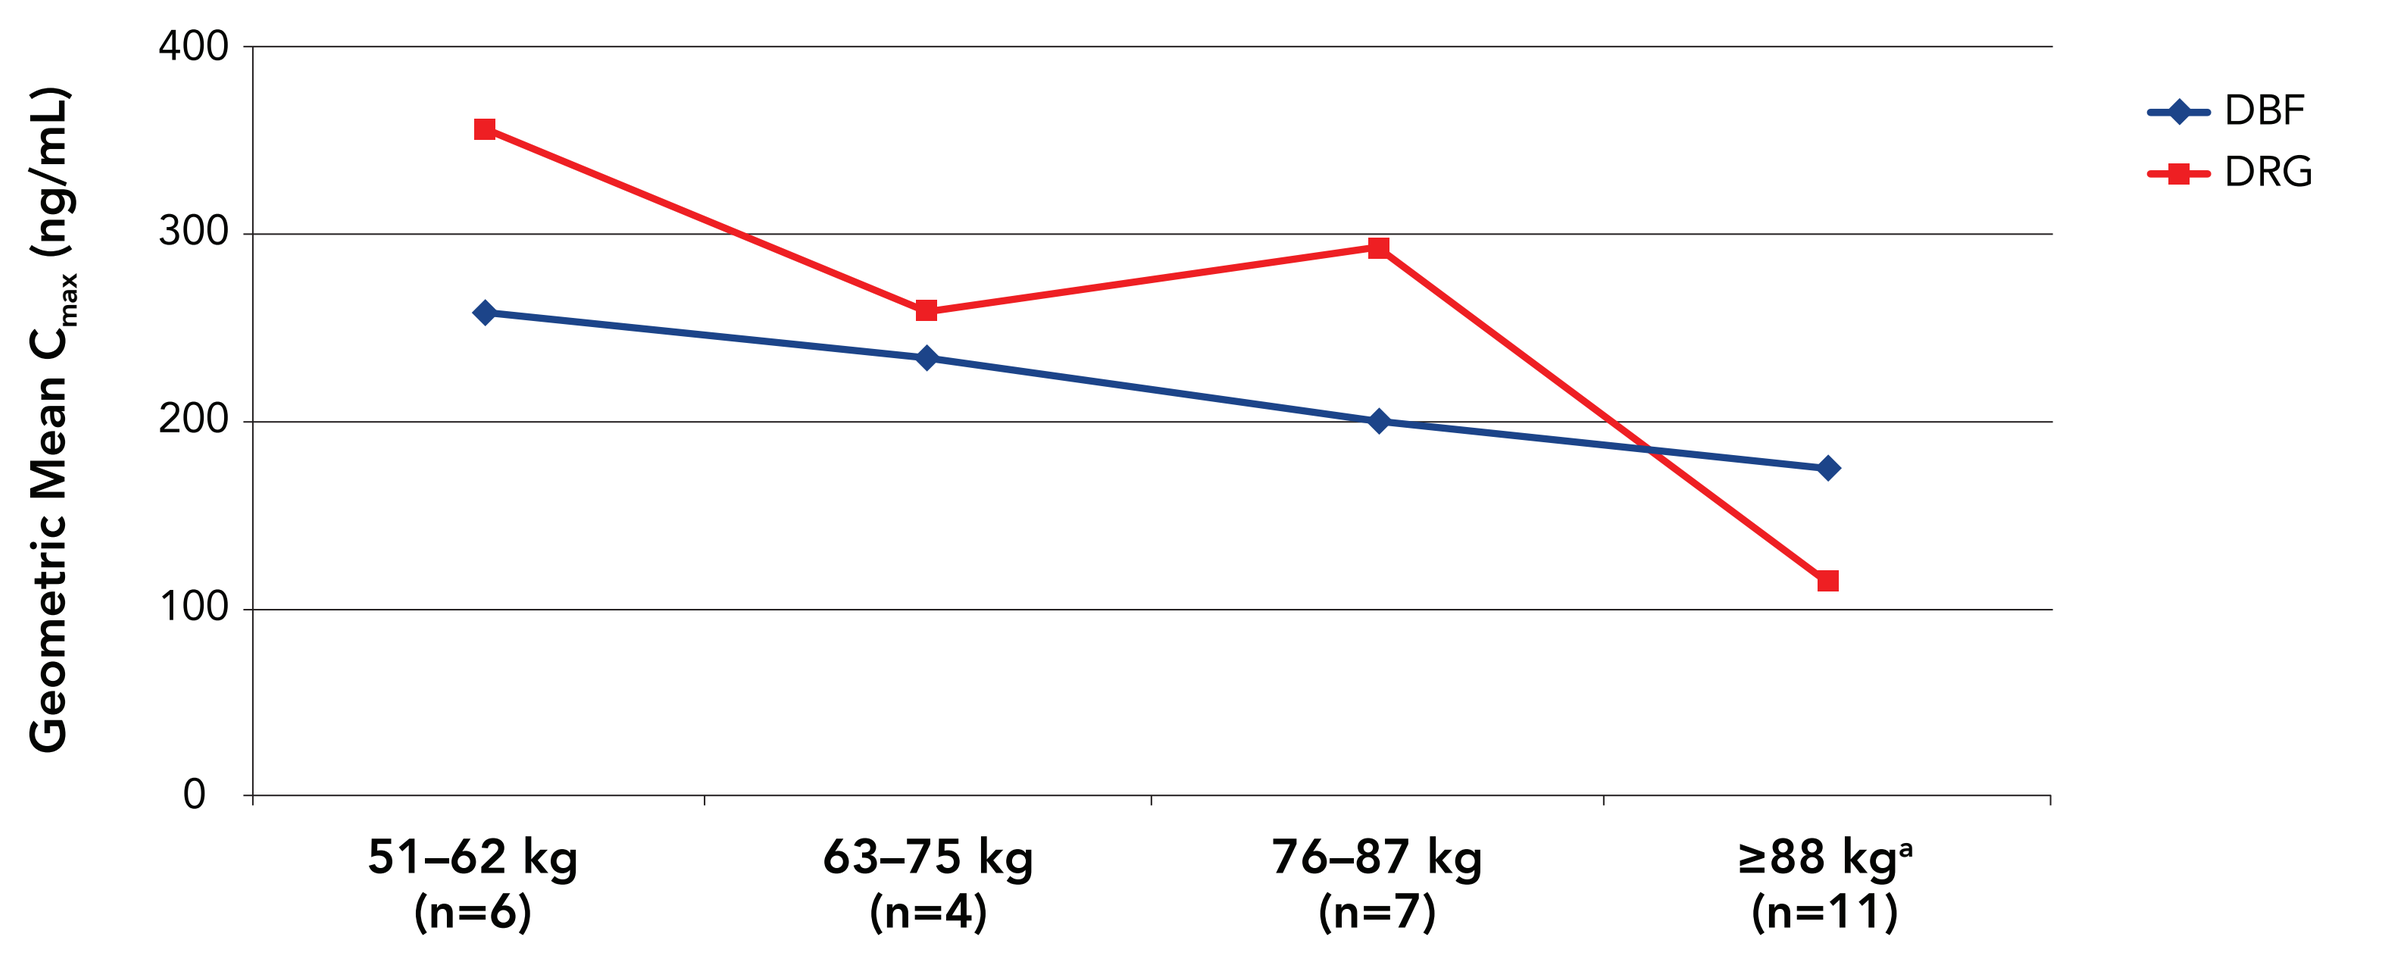

Geometric Mean C_{max} (ng/mL)
DBF
DRG
51–62 kg (n=6)
63–75 kg (n=4)
76–87 kg (n=7)
≥88 kg[a] (n=11)

[a]Includes 4 patients with weight 112–124.5 kg.
C_{max}, maximum observed plasma drug concentration; DBF, diazepam buccal film; DRG, diazepam rectal gel.

- There were no serious AEs related to study drug reported

CONCLUSIONS

- These results demonstrate that a single dose of DBF provides similar exposure to diazepam as DRG with significantly less variability when administered to adults with epilepsy according to weight-based regimens following a moderate-fat meal and confirm that the proposed weight-based dosing regimen for DBF provides diazepam exposure comparable to DRG
- The geometric mean values for C_{max} following DBF were consistently ≥150 ng/mL for each of the weight categories
- These results support the use of DBF as an easily administered alternative to DRG for patients with epilepsy who experience seizure emergencies despite treatment with antiseizure medications

REFERENCES

1. Penovich PE, et al. *Neurologist.* 2017;22:207-14.
2. Haut SR. *Curr Opin Neurol.* 2015;28:143-50.
3. Kanner AM, et al. *Arch Neurol.* 1996;53:258-63.
4. Pellock JM. *J Child Neurol.* 2007;22:9S-13S.
5. Sillanpaa M, Schmidt D. *Brain.* 2008;131:938-44.
6. Bergen DC. *Epilepsy Curr.* 2006;6:117-8.
7. Haut SR, et al. *Epilepsia.* 1999;40:1832-4.
8. Detyniecki K, et al. *Epilepsy Behav.* 2018;88:349-56.
9. Gainza-Lein M, et al. *Seizure.* 2017;52:188-94.
10. Vigevano F, et al. *Eur J Paediatr Neurol.* 2018;22:56-63.
11. Heller AH, et al. *Neurology.* 2018;90(15 suppl):P4.272.
12. Heller AH, et al. *Neurology.* 2018;90(15 suppl):P4.273.
13. Cereghino JJ. *Curr Treat Options Neurol.* 2007;9:249-55.
14. Jafarpour S, et al. *Seizure.* 2019;68:9-15.
15. Nayzilam [package insert]. Smyrna, GA: UCB, Inc.; 2019.ß
16. Diastat C-IV (diazepam rectal gel) [package insert]. San Antonio, TX: DPT Laboratories; 2016.
17. Fisgin T, et al. *J Child Neurol.* 2002;17:123-6.
18. Tatum IW. *Epilepsy Behav.* 2002;3:535-8.
19. Dhir A, Rogawski MA. *Epilepsia.* 2018;59:935-44.

ACKNOWLEDGMENTS

This analysis was sponsored by Aquestive Therapeutics, Inc. Editorial support was provided by Michael Morren, RPh, MBA, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, and funded by Aquestive Therapeutics, Inc.

DISCLOSURES

Michael A. Rogawski has served as a paid consultant to Aquestive Therapeutics, Inc. Allen H. Heller has served as a paid consultant to Aquestive Therapeutics, Inc. Simon Farrow has received research support from Syneos Health. Cassie Jung is an employee of Aquestive Therapeutics, Inc. Pavel Klein has served as a consultant or advisory board member for Abbott, Alliance, Aquestive, Eisai, SK Life Science, and UCB Pharma; has served as a speaker for Aquestive, Eisai, Sunovion, and UCB Pharma; and has received research grant support from Eisai. Sylvie Boudreault has nothing to disclose. Gary Slatko is an employee of Aquestive Therapeutics, Inc.

Presented at: American Epilepsy Society Annual Meeting; December 6–10, 2019; Baltimore, Maryland