Poster 3.304

SIMULATION OF THE PHARMACOKINETICS OF DIAZEPAM BUCCAL FILM IN ADULT PATIENTS WITH EPILEPSY WITH WEIGHT-ADJUSTED DOSING

Allen H. Heller[1], Gary Slatko[2], Michael A. Rogawski[3]

[1]Pharma Study Design, LLC, Woodbridge, CT; [2]Aquestive Therapeutics, Warren, NJ; [3]University of California, Davis, Sacramento, CA

INTRODUCTION

- Despite treatment with antiepileptic drugs, many patients with epilepsy experience seizure clusters, which can impair quality of life, emotional well-being, daily function, and productivity of patients and their caregivers[1,2]
- Diazepam rectal gel (DRG) is the legacy treatment indicated for management of patients with epilepsy who require intermittent use of diazepam to control bouts of increased seizure activity,[3] but has a less-than-ideal route of administration, which may affect adherence[1,4]
 - DRG is dosed according to weight category[3]
- Diazepam buccal film (DBF) is a novel dosage form of diazepam under development for the treatment of epilepsy patients with breakthrough or cluster seizures.[5,6] It is designed to be readily usable, with reliable and predictable pharmacokinetics (PK; dose-proportional for both maximal plasma drug concentration [C_{max}] and area under the plasma drug concentration-time curve [AUC])[6]
- In a previous study in patients with epilepsy, the PK performance of DBF (ie, diazepam exposure) following a single 12.5 mg fixed dose was shown to be similar under interictal and ictal/periictal conditions[7]
- Aquestive has developed a proposed weight-based dosing regimen for DBF based on population PK modeling intended to provide similar exposure to DRG administered according to the DRG label. This simulation was conducted in an effort to predict exposures following DBF using the proposed weight-based dosing regimen

OBJECTIVE

- To compare diazepam exposures obtained with DBF when administered according to a simulated weight-based dose regimen vs a 12.5 mg fixed dose under both interictal and ictal/periictal conditions

METHODS

STUDY DESIGN

- Data were obtained from a multicenter, open-label, crossover study that evaluated the PK of a single dose of DBF under interictal (no seizure activity observed in the preceding 3 hours) or ictal/periictal (during a clinically observed seizure or within 5 minutes following its cessation) conditions
- Eligibility was established at a screening visit ≤28 days prior to the first study drug administration
- For the interictal condition, patients received DBF 12.5 mg during a PK-only study visit (treatment period A); patients remained at the epilepsy monitoring unit (EMU) for at least 8 hours after treatment
- For the ictal/periictal condition, patients received DBF 12.5 mg while undergoing clinical observation at the EMU (treatment period B); patients remained in the EMU for at least 4 hours after treatment
 - Treatment periods A and B could occur in any order, with the visits separated by approximately 3 weeks between DBF doses
- An outpatient follow-up visit took place at 14 (± 2) days after the last treatment period

PATIENTS

- Adult men and women ages 17–65 years with epilepsy who had poorly controlled tonic-clonic seizures or focal seizures with impaired awareness were enrolled
- Patients reported on average ≥1 seizure every 3 days or ≥10 seizures per month
- Patients were receiving ≥1 antiepileptic drug as maintenance
- Investigators were permitted to adjust concomitant antiseizure drugs according to clinical judgment

PK SAMPLING AND ANALYSIS

- Blood samples for PK analyses were obtained before DBF administration and at the following time points (± 5 minutes): 0.25 , 0.5, 1.0, 1.5, 2.0, and 4.0 hours after dosing during both treatment periods
- Plasma was analyzed for diazepam by a high-performance liquid chromatography method using tandem mass spectroscopy and automated extraction for diazepam (1.00 to 1000 ng/mL) in human EDTA K2 plasma
- C_{max}, time to C_{max} (T_{max}), and partial AUC from time zero until 2 hours (AUC_{0-2h}) and 4 hours (AUC_{0-4h}) were calculated

SIMULATION

- PK profiles were simulated based on body weight and robust evidence of dose-proportionality of DBF from healthy volunteer studies[5,6] and population PK modeling
- Simulation was based on administration of a weight-based regimen as in **Table 1**

TABLE 1. WEIGHT-BASED DOSING REGIMEN SIMULATED

Weight (kg)	Dose
38–50	1 x 10 mg
51–62	1 x 12.5 mg
63–75	1 x 15 mg
76–87	1 x 15 mg
>88	1 x 17.5 mg

STATISTICAL ANALYSIS

- PK parameters were calculated and tabulated by treatment period with summary statistics; analyses used the actual time of blood sample collection
- Pairwise comparisons of diazepam exposure following treatment period A and treatment period B were evaluated by analysis of variance with treatment period as a fixed factor and patient as a random factor on the log-transformed C_{max}, AUC_{0-2h}, and AUC_{0-4h}
- T_{max} was compared using the nonparametric Wilcoxon signed-rank test

RESULTS

PATIENTS

- A total of 35 patients received at least 1 dose of study drug. Patients were excluded from PK analyses if both treatments were not completed (n=4), critical PK time points were missed as adjudicated by an expert blinded to the concentration data (n=3), predose diazepam concentrations were >5% of the subsequent C_{max} (n=2), or DBF was administered in a manner contrary to instructions (n=5)
- There were 21 patients who had PK profiles valid for analysis and available for both treatment conditions. Demographic characteristics are summarized in **Table 2**
 - All 21 patients had samples collected for ≤2 hours; 18 had samples collected for ≤4 hours
- The average dose predicted for this group of patients with simulated weight-based dosing was 14.9 mg/kg

TABLE 2. SUMMARY OF DEMOGRAPHIC CHARACTERISTICS

Demographic Characteristic	PK Analysis Set (n=21)
Age, mean (SD), years	34 (13)
Sex, n (%)	
Female	12 (57)
Male	9 (43)
Ethnicity, n (%)	
Not Hispanic or Latino	18 (86)
Hispanic or Latino	3 (14)
Race, n (%)	
White	12 (57)
Hawaiian	5 (24)
Black	3 (14)
American Indian	1 (5)
Height, mean (SD), cm	166.6 (11.6)
Weight, mean (SD), kg	76.8 (17.6)
BMI, mean (SD), kg/m²	28.0 (7.0)

BMI, body mass index; PK, pharmacokinetic; SD, standard deviation.

PK PARAMETERS

- PK parameters obtained following DBF administered at a fixed dose or simulated weight-based dose under interictal and ictal/periictal conditions are summarized in **Table 3**
 - There were no statistical differences between the geometric mean C_{max}, AUC_{0-2h}, AUC_{0-4h}, or median T_{max} values in the 2 treatment conditions with either the 12.5 mg fixed-dose regimen or the simulated weight-based dose regimen

TABLE 3. PK PARAMETERS FOLLOWING DBF IN THE INTERICTAL AND ICTAL/PERIICTAL STATE USING A FIXED-DOSE (12.5 MG) OR SIMULATED WEIGHT-BASED REGIMEN

2-Hour Profiles (n=21)

	Fixed-Dose (12.5 mg) Regimen				Simulated Weight-Based Regimen			
	Period A: Interictal	Period B: Ictal/ Periictal	Ratio of Geometric Means, B/A (%)*	90% CI (%) for Ratio†	Period A: Interictal	Period B: Ictal/ Periictal	Ratio of Geometric Means, B/A (%)*	90% CI (%) for Ratio†
	Geometric Mean	Geometric Mean			Geometric Mean	Geometric Mean		
C_{max}, ng/mL	198.76	184.40	92.77	74.50–112.53	234.73	217.77	92.77	74.50–112.53
AUC_{0-2h}, ng•h/mL	264.57	248.06	93.76	73.86–119.02	321.50	301.52	93.76	73.86–119.02
	Median	Median			Median	Median		
T_{max}, h	0.680‡	0.983‡			0.680‡	0.983‡		

4-Hour Profiles (n=18)

	Fixed-Dose (12.5 mg) Regimen				Simulated Weight-Based Regimen			
	Period A: Interictal	Period B: Ictal/ Periictal	Ratio of Geometric Means, B/A*	90% CI (%) for Ratio†	Period A: Interictal	Period B: Ictal/ Periictal	Ratio of Geometric Means, B/A (%)*	90% CI (%) for Ratio†
	Geometric Mean	Geometric Mean			Geometric Mean	Geometric Mean		
C_{max}, ng/mL	190.34	179.96	95.54	73.34–121.89	224.19	211.96	95.54	73.34–121.89
AUC_{0-4h}, ng•h/mL	483.79	433.32	89.57	69.21–115.91	569.82	510.37	89.57	69.21–115.91
	Median	Median			Median	Median		
T_{max}, h	0.767§	0.533§			0.767§	0.533§		

**Calculated using least-squares means according to the formula: exp (difference) x 100.*
†90% geometric confidence interval using log-transformed data.
‡Difference in T_{max} between treatment periods was not significant, P=0.8620.
§Difference in T_{max} between treatment periods was not significant, P=0.5708 (Wilcoxon signed-rank test).
AUC, area under the plasma concentration-time curve; CI, confidence interval; C_{max}, maximum plasma drug concentration; T_{max}, time to reach maximum plasma concentration.

- On average, geometric mean predicted PK parameters were 18%–22% higher with the simulated weight-based dose regimen compared with the 12.5-mg fixed-dose regimen
- Box plots for median C_{max} and AUC values observed at 2 and 4 hours after dosing during interictal and ictal/periictal conditions for both the 12.5 mg fixed-dose regimen and the simulated weight-based dose regimen are shown in **Figure 1**
 - Similarly to the geometric means, median predicted PK parameter values were numerically higher with the simulated weight-based dose regimen

Figure 1. Box Plots of PK Parameter Values Observed With the Fixed-Dose (12.5 mg) vs Simulated Weight-Based Dose Regimen in the Interictal and Ictal/Periictal States: A) C_{max} (2-hour profiles), B) AUC_{0-2h}, C) C_{max} (4-hour profiles), and D) AUC_{0-4h}

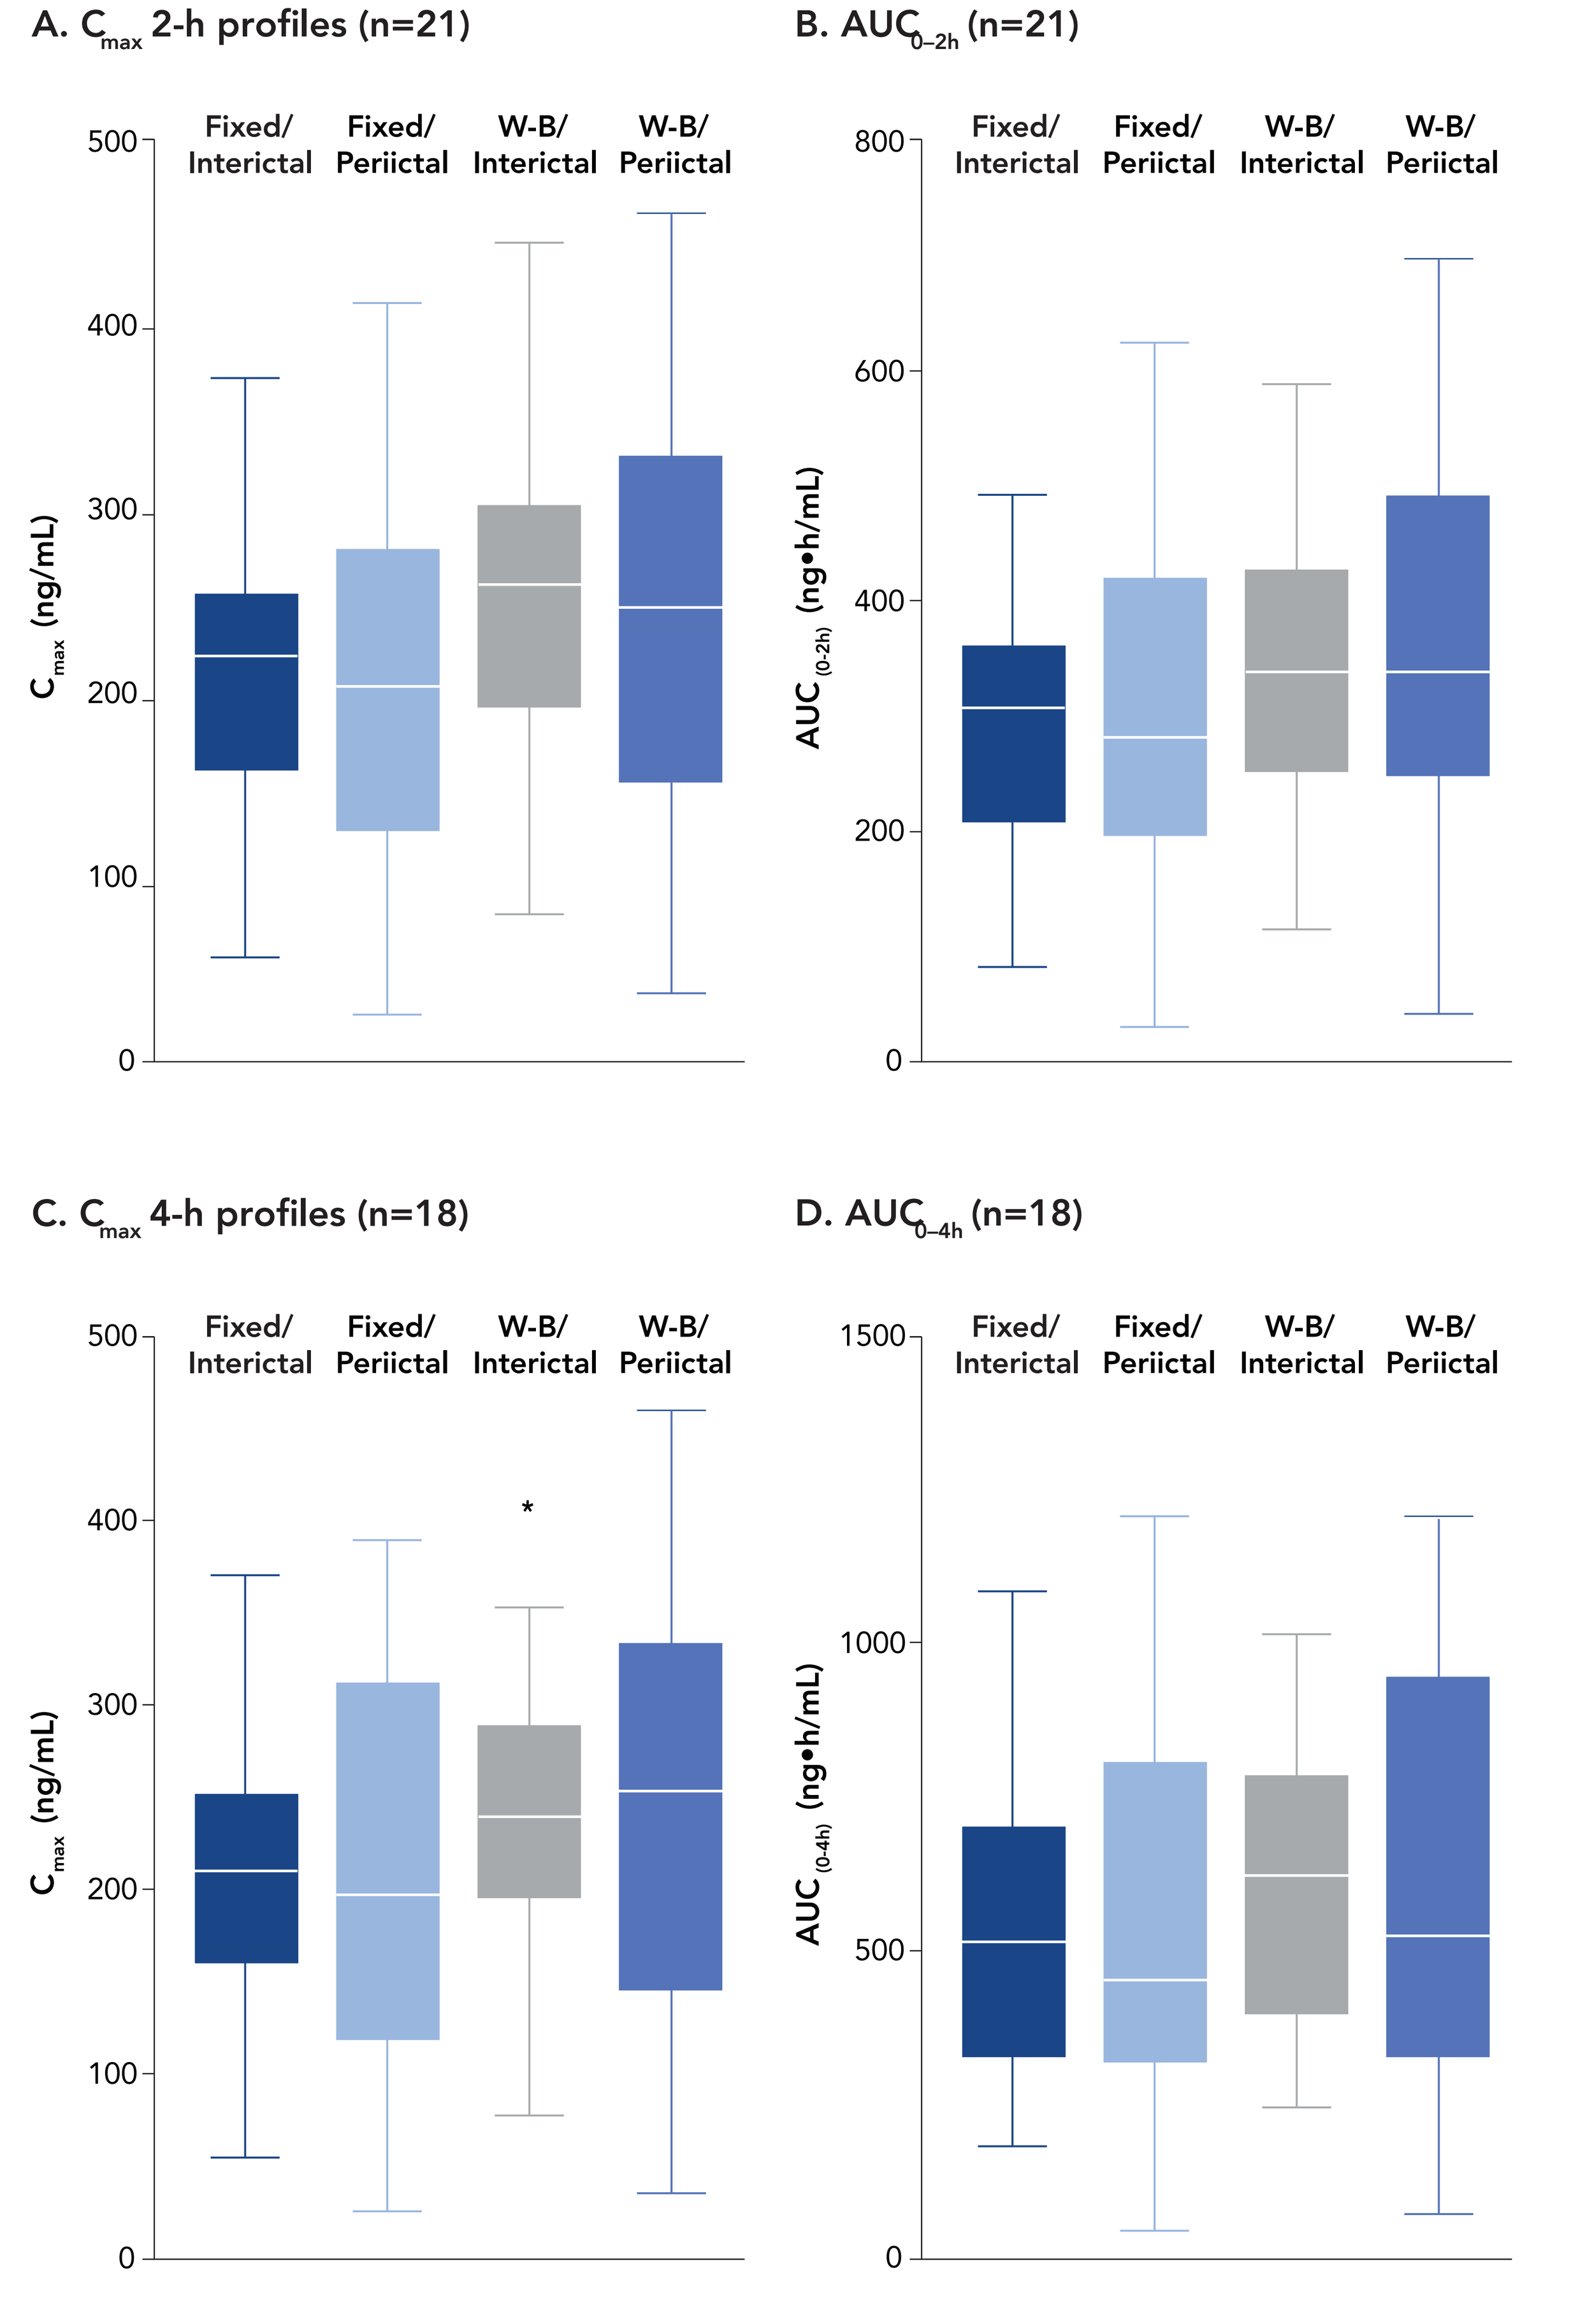

A. C_{max} 2-h profiles (n=21)
B. AUC_{0-2h} (n=21)
C. C_{max} 4-h profiles (n=18)
D. AUC_{0-4h} (n=18)
Fixed/ Interictal
Fixed/ Periictal
W-B/ Interictal
W-B/ Periictal
C_{max} (ng/mL)
$AUC_{(0-2h)}$ (ng•h/mL)
$AUC_{(0-4h)}$ (ng•h/mL)

The horizontal line within each box indicates the median value; the top and bottom edges of each box represent 75th and 25th percentiles, respectively; the upper and lower limit of the bars represent the maximum and minimum observed values within 1.5 x the interquartile range (IQR) measured from the second and third quartiles. Values outside this range are plotted as points, indicated by an asterisk marker.
AUC, area under the plasma concentration-time curve; C_{max}, maximum plasma drug concentration; W-B, weight-based.

- Mean diazepam plasma concentrations over time after DBF administration for the 12.5 mg fixed-dose regimen and the simulated weight-based dose regimen are presented in **Figure 2**

Figure 2. Mean Plasma Diazepam Concentrations Following Administration of DBF in the Interictal or Ictal/Periictal State (n=16–21) Using A) the 12.5 mg Fixed-Dose Regimen* or B) the Simulated Weight-Based Dose Regimen*

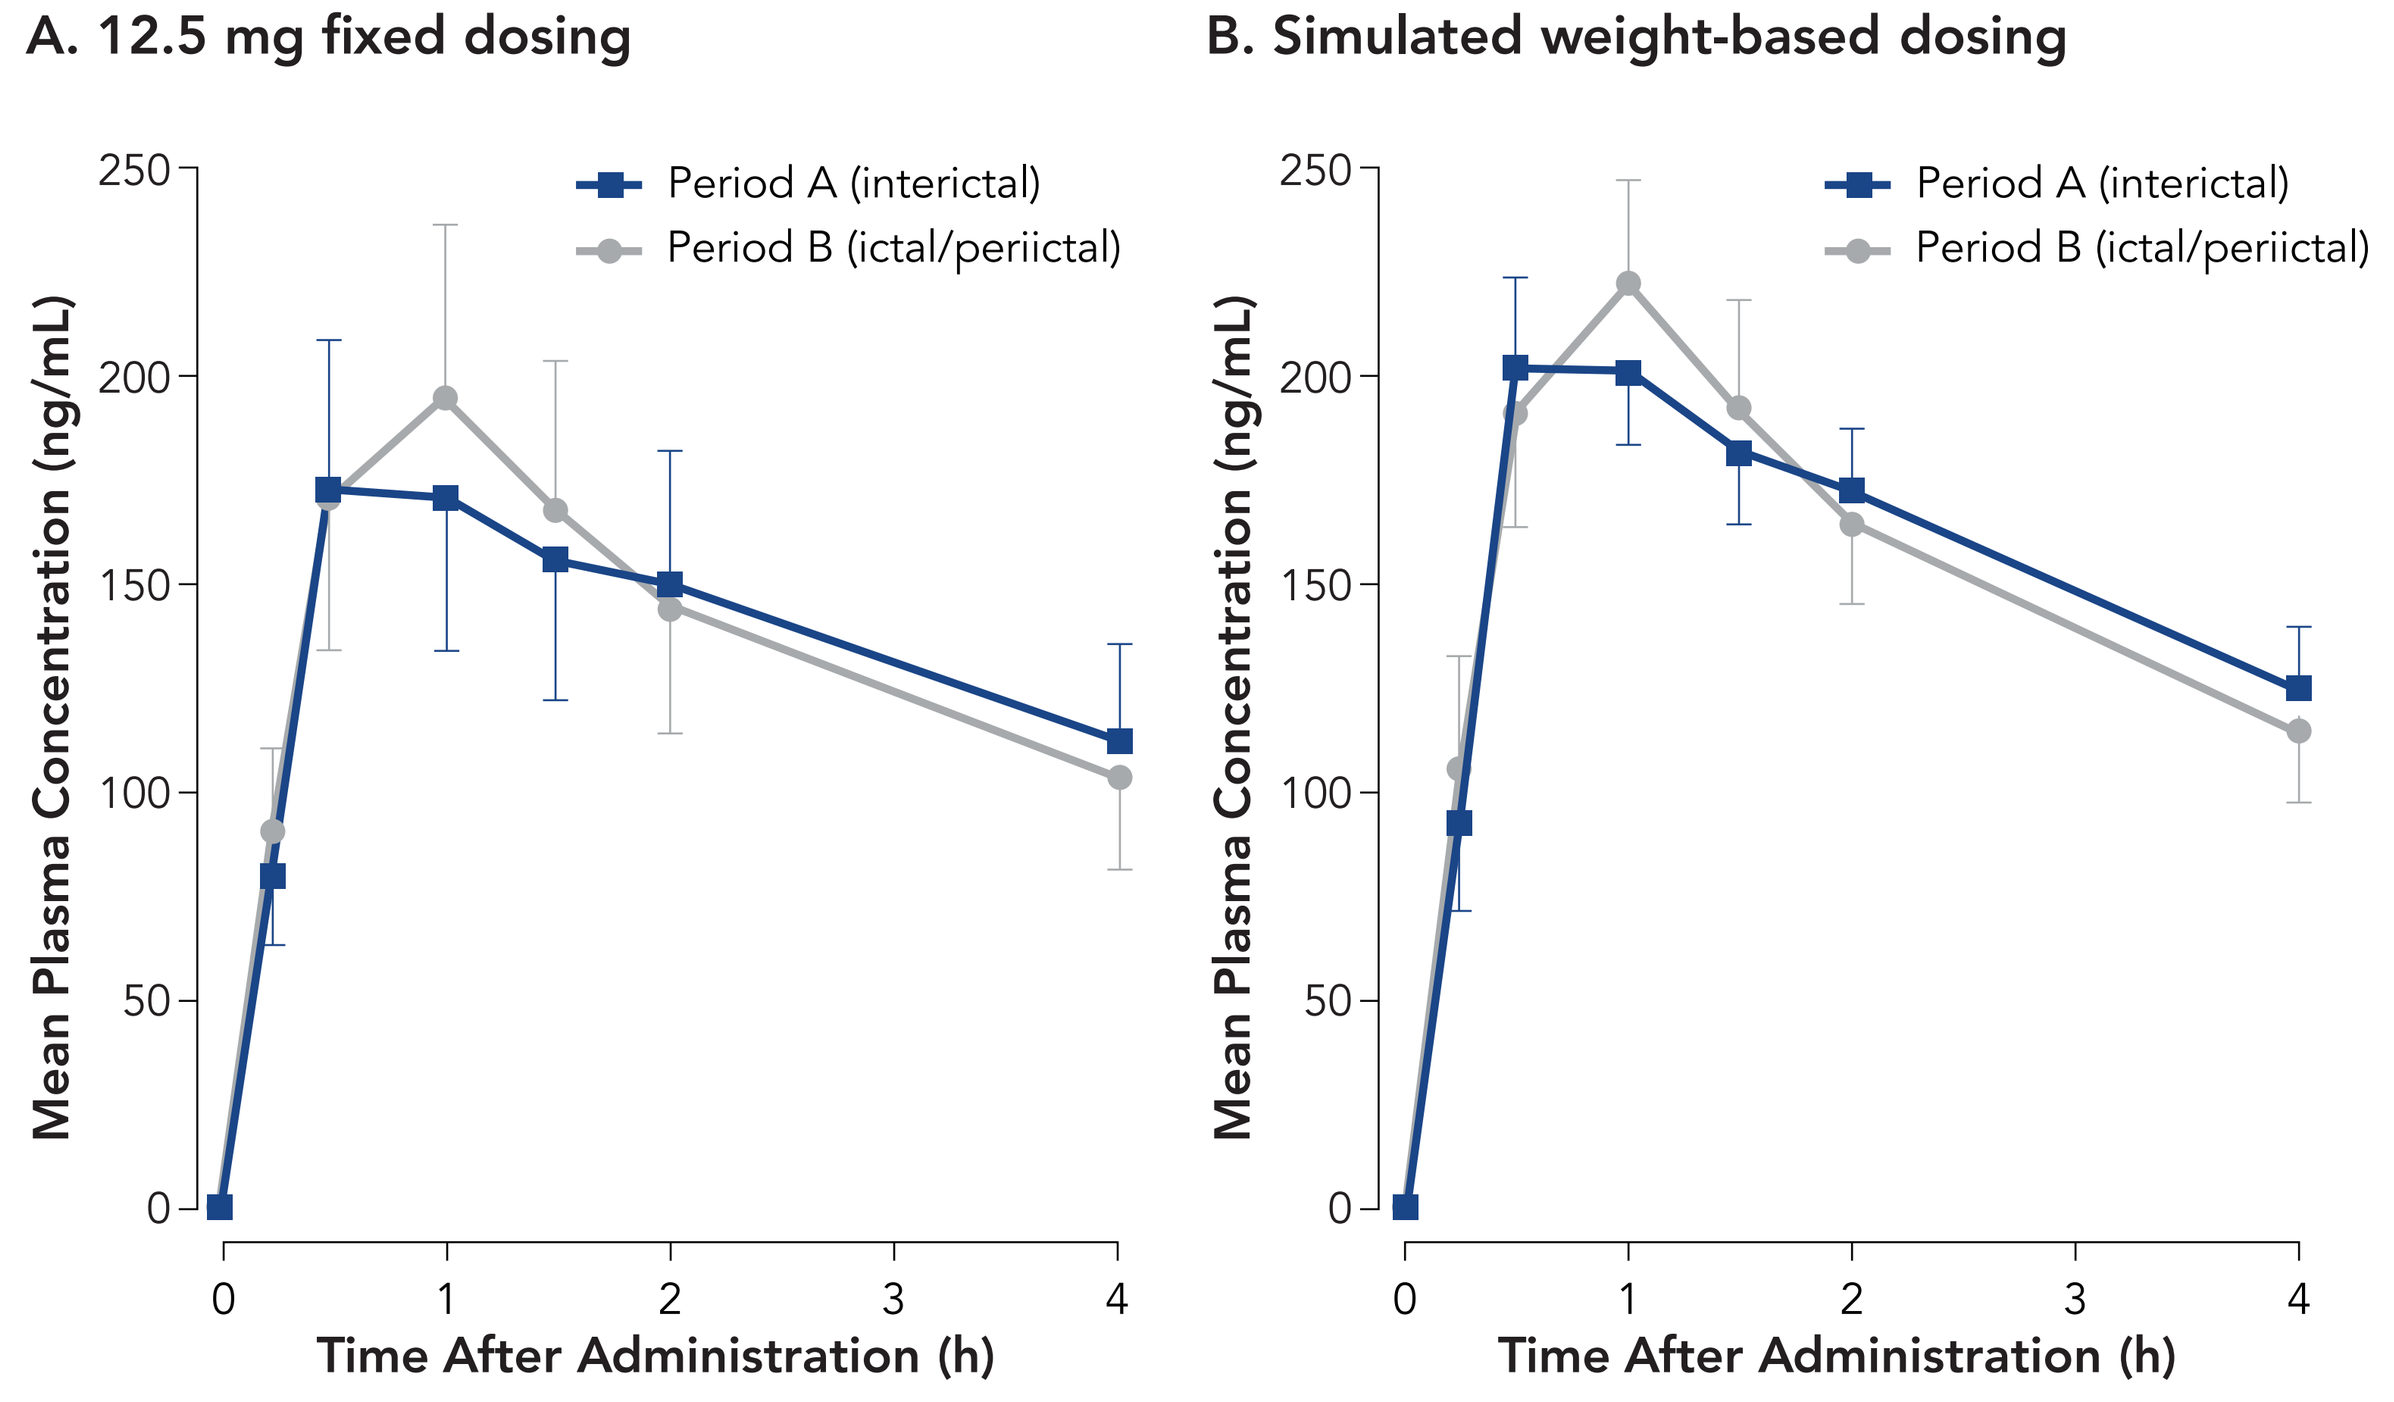

A. 12.5 mg fixed dosing
B. Simulated weight-based dosing
Period A (interictal)
Period B (ictal/periictal)
Mean Plasma Concentration (ng/mL)
Time After Administration (h)

**Predicted plasma concentrations based on data from 21 patients with valid profiles for both interictal and ictal/periictal periods. Each time point is the mean of concentration data from 16–21 patients.*
DBF, diazepam buccal film.
Error bars are standard error of the mean.

CONCLUSIONS

- Simulation of the weight-based dose regimen for administration of DBF to adults with epilepsy predicted a higher exposure than that observed with a fixed dose of 12.5 mg
- The weight-based dose regimen simulated here (average dose 14.9 mg/kg) predicts geometric mean C_{max} values greater than 200 ng/mL under both interictal and ictal/periictal conditions, well within the range generally considered as therapeutic for diazepam antiepileptic activity
- The predicted geometric mean C_{max} values observed here are consistent with actual values from a subsequent crossover study in which patients actually received single administrations of DBF dosed according to the weight-based regimen in the current analysis[8]

REFERENCES

1. Jafarpour S, et al. *Seizure.* 2019;68:9-15. **2.** Penovich PE, et al. *Neurologist.* 2017;22:207-14. **3.** Diastat C-IV (diazepam rectal gel) [package insert]. San Antonio, TX: DPT Laboratories; 2016. **4.** Cereghino JJ. *Curr Treat Options Neurol.* 2007;9:249-55. **5.** Heller AH, et al. *Neurology.* 2018;90(15 suppl):P4.272. http://n.neurology.org/content/90/15_Supplement/P4.272. **6.** Heller AH, et al. *Neurology.* 2018;90(15 suppl):P4.273. http://n.neurology.org/content/90/15_Supplement/P4.273. **7.** Rogawski MA, et al. Presented at: Annual Meeting of the American Epilepsy Society; November 30-December 4, 2018; New Orleans, LA. https://www.aesnet.org/meetings_events/annual_meeting_abstracts/view/551841. **8.** Rogawski MA, et al. Presented at: Annual Meeting of the American Epilepsy Society; December 6-10, 2019; Baltimore, MD.

ACKNOWLEDGMENTS

This analysis was sponsored by Aquestive Therapeutics, Inc. Editorial support was provided by Michael Morren, RPh, MBA, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, and funded by Aquestive Therapeutics, Inc.

DISCLOSURES

Allen H. Heller has served as a paid consultant to Aquestive Therapeutics, Inc. Gary Slatko is an employee of Aquestive Therapeutics, Inc. Michael A. Rogawski has served as a paid consultant to Aquestive Therapeutics, Inc.

Presented at: American Epilepsy Society Annual Meeting; December 6–10, 2019; Baltimore, Maryland