SAFETY AND PHARMACOKINETICS OF DIAZEPAM BUCCAL SOLUBLE FILM

Allen H. Heller[1], Stephen Wargacki[2], Cassie Jung[2], David J. Wyatt[3], A. Mark Schobel[2]
[1]Pharma Study Design, LLC, Woodbridge, CT 06525, USA; [2]Aquestive Therapeutics, Warren, NJ 07059, USA; [3]Syneos Health Clinical Solutions Early Phase, Miami, FL 33131, USA

ABSTRACT

Objective: The objective of this study was to assess the dose-proportionality and safety of Diazepam Buccal Soluble Film (DBSF) at single doses of 5, 10, and 15 mg.

Background: Diazepam Buccal Soluble Film (DBSF) is a novel dosage form of diazepam under development for the management of selected, refractory, patients with epilepsy, on stable regimens of anti-epileptic drugs, who require intermittent use of diazepam to control bouts of increased seizure activity.

Design/Methods: Healthy adult men age 18-55 were administered single doses of DBSF containing diazepam 5 mg, 10 mg, or 15 mg under fasting conditions in an open-label, randomized sequence crossover with a 21 day washout between treatments. Subjects were confined from 14 hours prior to drug administration until 24 hours post-dose. Plasma samples for diazepam were drawn pre-dose and at intervals until 240 hours. Dose-proportionality was assessed by comparing area under the curve (AUC) and maximal plasma concentration (C_{max}) (both log-transformed) following the 10 mg dose with the corresponding parameters following 5 mg and 15 mg (both dose-normalized to a 10 mg dose). Treatment-emergent adverse events (TEAEs), vital signs, and standard laboratory evaluations were recorded.

Results: Thirty subjects received at least one dose of DBSF. Mean C_{max} values (SD) for diazepam at 5, 10, and 15 mg were 168.84 (46.5) n=26; 342.61 (59.03) n=25; and 492.21 (108.14) n=30, respectively, and median times to maximum concentration (T_{max}) were 0.81, 1.0, and 1.3 hours. C_{max}, AUC_{0-t}, and AUC_{0-inf} were dose-proportional. The most frequent TEAEs were dose-related somnolence and dizziness. None of these events were severe or serious.

Conclusions: The pharmacokinetics of diazepam following a single dose of DBSF are dose-proportional over the range 5 to 15 mg. DBSF administered as a single dose 5 to 15 mg under fasting conditions was safe and well tolerated.

BACKGROUND

Safety and Pharmacokinetics of Diazepam Buccal Soluble Film

- The route of administration of diazepam rectal gel is not ideal, and may affect adherence[1]
- Diazepam Buccal Soluble Film (DBSF) is a novel dosage form of diazepam under development as a direct alternative to the rectal gel formulation
- DBSF contains diazepam in a dissolvable polymer-based matrix designed to maximize transport across the buccal mucosa
- The current study was undertaken to assess the dose-proportionality and overall safety and tolerability of DBSF following single doses of 5 mg, 10 mg, and 15 mg in healthy male adult volunteers

OBJECTIVES

- The primary objective of this study was to assess the dose-proportionality of DBSF following single doses of 5, 10, and 15 mg in healthy male adult volunteers
- A secondary objective was to assess the overall safety and tolerability of single doses of DBSF of 5, 10, and 15 mg

METHODS

STUDY DESIGN

- Single-dose, randomized, 3-period, 3-treatment, 6-sequence, open-label crossover at a single site in Miami, FL (**Table 1**)

SUBJECTS

- Healthy, non-smoking (≥6 months prior to screening), male volunteers, 18-55 years of age with a body mass index (BMI) 18.5-29.9 kg/m²
- Individuals were excluded if they had clinically significant illnesses or conditions, history of drug abuse or dependence, use of benzodiazepines within 30 days, or tobacco use within 6 months of study drug administration
- All subjects provided informed consent. The study protocol and informed consent were reviewed and approved by an ethics committee (Chesapeake IRB) prior to study initiation

PROCEDURES

- Eligibility was established at a screening visit within 30 days of study drug administration
- Subjects were confined from 14 hours prior to drug administration until 24 hours post-dose, returning to the clinic for subsequent blood draws

TABLE 1: SUBJECTS RANDOMIZED TO ONE OF SIX SEQUENCES (DOSE OF DIAZEPAM BUCCAL SOLUBLE FILM)

Sequence	Period 1		Period 2		Period 3
1	5 mg	21-day washout	10 mg	21-day washout	15 mg
2	10 mg		15 mg		5 mg
3	15 mg		5 mg		10 mg
4	5 mg		10 mg		15 mg
5	10 mg		15 mg		5 mg
6	15 mg		5 mg		10 mg

PHARMACOKINETIC SAMPLING AND BIOANALYSIS

- Plasma samples for diazepam were drawn pre-dose and post-dose at 0.25, 0.5, 0.75, 1, 1.5, 2, 3, 4, 6, 8, 12, 24, 48, 72, 96, 120, 144, 192, and 240 hours in each study period
- There was a 21 day washout period between treatments
- Plasma was analyzed for diazepam by a high-performance liquid chromatography method using tandem mass spectroscopy and automated extraction for diazepam (1.00 to 1000 ng/mL) in human EDTA K_2 plasma.

SAFETY AND TOLERABILITY

- Treatment-emergent adverse events (TEAE), vital signs, and standard laboratory evaluations were recorded.

STATISTICAL ANALYSIS

- Plasma pharmacokinetic (PK) parameters were summarized by treatment group using descriptive statistics, and geometric means were calculated for all PK parameters
- Dose-proportionality was assessed by comparing area under the curve (AUC) and maximal plasma concentration (C_{max}), both log-transformed, following the 10 mg dose with the corresponding parameters following 5 mg and 15 mg doses (both dose-normalized to a 10 mg dose)

Dose-proportionality

- Comparisons between treatment groups (5 mg vs 10 mg and 15 mg vs 10 mg) were evaluated using an analysis of variance (ANOVA), with sequence, period, and treatment as fixed factors, and subject nested within sequence as a random factor on the ln-transformed values of AUC_{0-t}, AUC_{0-inf}, and C_{max}, dose normalized to 10 mg
- Sequence was tested using subject nested within sequence as the error term
- Each ANOVA included calculations of least-squares (LS) means for each treatment, the LS-mean for treatment differences, and the standard error associated with this difference
- T_{max} was analyzed using the Wilcoxon Signed Rank test
- The ratio of geometric means (5 mg/10 mg and 15 mg/10 mg) and corresponding 90% confidence intervals (CI), based on LS means from the ANOVA of the ln-transformed data dose-normalized to a 10 mg dose, were calculated for AUC_{0-t}, AUC_{0-inf}, and C_{max}
- Criteria for dose-proportionality: For the ranges 5 mg to 10 mg and 10 mg to 15 mg, the 90% geometric CI of the ratio (5 mg/10 mg and 15 mg/10 mg, respectively) of LS means from the ANOVA of the ln-transformed AUC_{0-t}, AUC_{0-inf}, and C_{max}, dose-normalized to 10 mg were required to be within 80% to 125%

RESULTS

SUBJECTS

- All subjects (n=30) were male and the majority were white (66.7%), Hispanic or Latino (90.0%), with mean age of 39.9 years and mean BMI of 26.69 kg/m²
- All 30 subjects received at least one dose of DBSF
- Baseline demographic characteristics are summarized in **Table 2**

TABLE 2: BASELINE DEMOGRAPHIC CHARACTERISTICS (SAFETY POPULATION)

Characteristic	Result (N=30)
Age (years), mean ± SD	39.9 ± 10.0
Male, n (%)	30 (100)
Ethnicity	
Not Hispanic or Latino, n (%)	3 (10.0)
Hispanic or Latino, n (%)	27 (90.0)
Race	
White, n (%)	20 (66.7)
Black, n (%)	10 (33.3)
Height (cm), mean ± SD	176.62 ± 7.96
Weight (kg), mean ± SD	83.54 ± 11.39
BMI (kg/m²), mean ± SD	26.69 ± 2.20

BMI, body mass index; SD, standard deviation.

PHARMACOKINETICS

- Plasma concentration-time profiles (geometric mean) are shown in **Figure 1**
- Pharmacokinetic parameters (arithmetic mean and SD) are shown in **Table 3**

Figure 1. Diazepam plasma concentrations over time (geometric means; linear scale)[a]

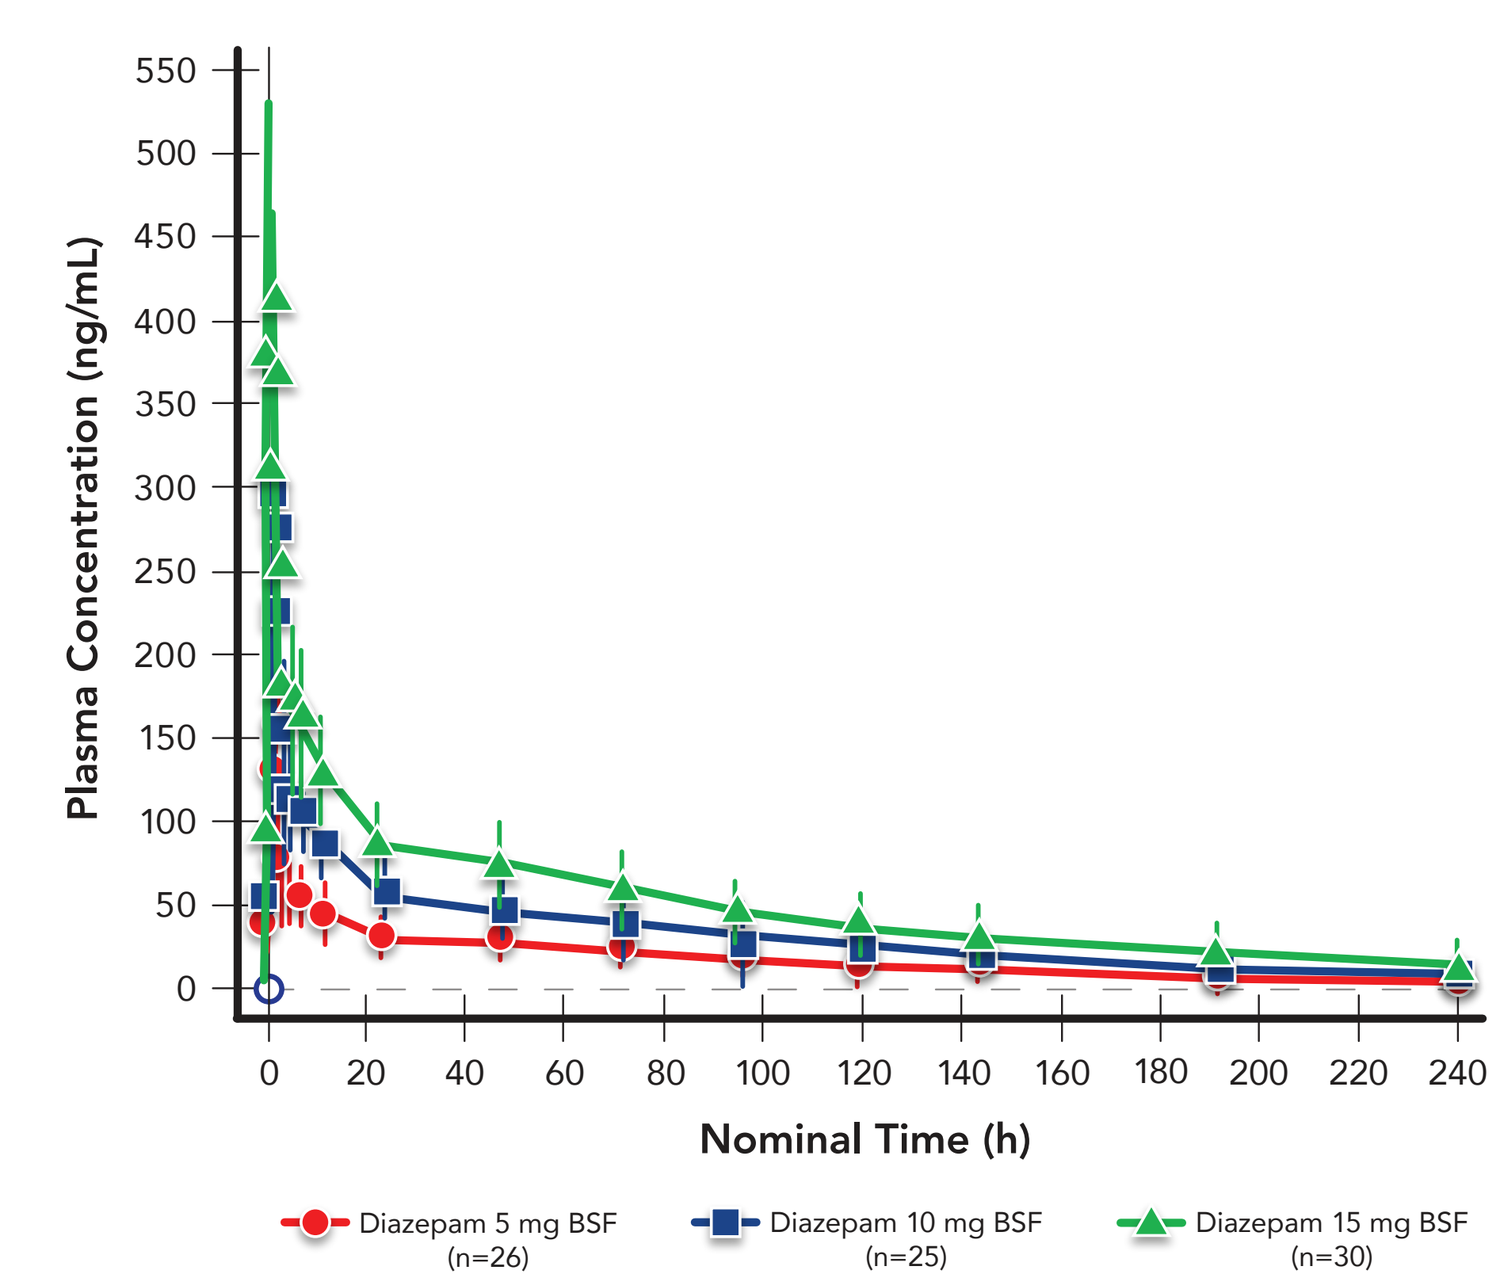
Plasma Concentration (ng/mL)
550
500
450
400
350
300
250
200
150
100
50
0
0
20
40
60
80
100
120
140
160
180
200
220
240
Nominal Time (h)
Diazepam 5 mg BSF (n=26)
Diazepam 10 mg BSF (n=25)
Diazepam 15 mg BSF (n=30)

TABLE 3: SUMMARY OF PHARMACOKINETIC PARAMETERS FOR DIAZEPAM

Parameter (units)	Diazepam Buccal Soluble Film								
	5 mg (n=26)			10 mg (n=25)			15 mg (n=30)		
	Mean		SD	Mean		SD	Mean		SD
AUC_{0-t} (h*ng/mL)	4268.10		1383.58	8413.04		2347.79	12794.42		3350.80
AUC_{0-inf} (h*ng/mL)	4915.89		1913.40	9731.75		3438.78	15241.45		5375.37
Residual area (%)	11.24		7.53	11.17		9.00	13.21		10.33
C_{max} (ng/mL)	186.84		46.49	342.61		59.03	492.21		108.14
$T_{1/2el}$ (h)	76.35		27.74	76.67		32.93	86.11		40.35
K_{el} (/h)	0.0106		0.0049	0.0108		0.0047	0.0099		0.0046
Correlation	-0.9641		0.0410	-0.9739		0.0261	-0.9764		0.023
Cl/F (L/h)	1.15		0.39	1.16		0.40	1.11		0.38
Cl/F/kg (L/(h*kg))	0.0139		0.0041	0.0138		0.0043	0.0134		0.0047
V_d/F (L)	117.04		37.93	115.90		36.42	121.45		34.81
V_d/F/kg (L/kg)	1.42		0.39	1.39		0.40	1.46		0.40
	Median	Min	Max	Median	Min	Max	Median	Min	Max
T_{max} (h)	0.808	0.490	2.016	0.999	0.493	1.998	1.268	0.487	1.997

DOSE-PROPORTIONALITY

- Comparisons for C_{max} and AUC between 5 mg and 10 mg and between 15 mg and 10 mg DBSF (**Table 4**) indicate that criteria for dose-proportionality were achieved
- P-values for treatment, period, and sequence for comparisons between 5 mg and 10 mg and between 15 mg and 10 mg DBSF are shown in **Table 5**
- Plots of mean values C_{max}, AUC_{0-t}, and AUC_{0-inf} as a function of dose are shown in **Figure 2**

TABLE 4: TESTS FOR DOSE-PROPORTIONALITY OF DBSF 5, 10, 15 MG

	5 mg (n=26)	10 mg (n=25)	Ratio of Geometric Means (90% CI)
C_{max} (ng/mL)	187	343	104.6 (97.9-111.7)
AUC_{0-t} (ng*h/mL)	4268	8413	103.5 (99.5-107.7)
AUC_{0-inf} (ng*h/mL)	4916	9732	104.1 (99.4-109.0)
	15 mg (n=26)	**10 mg (n=25)**	
C_{max} (ng/mL)	492	343	98.8 (92.4-105.8)
AUC_{0-t} (ng*h/mL)	12,794	8413	101.9 (97.6-106.4)
AUC_{0-inf} (ng*h/mL)	15,241	9732	103.6 (98.5-109.0)

AUC_{0-t}, area under the concentration-time curve from time zero until the last measurable concentration; AUC_{0-inf}, area under the concentration-time curve from time zero to infinity (extrapolated); C_{max}, maximum plasma concentration

TABLE 5: P-VALUES FOR ANOVA DIAZEPAM DOSE-PROPORTIONALITY ANALYSES[a]

	5 mg vs 10 mg		
	Treatment	Period	Sequence
Parameter	P-values		
AUC_{0-t}	0.1466	0.0885	0.3743
AUC_{0-inf}	0.1474	0.2410	0.4012
C_{max}	0.2548	0.2375	0.2247
	15 mg vs 10 mg		
	Treatment	Period	Sequence
Parameter	P-values		
AUC_{0-t}	0.4544	0.4984	0.3723
AUC_{0-inf}	0.2381	0.4709	0.5903
C_{max}	0.7708	0.7397	0.0221

AUC_{0-t}, area under the concentration-time curve from time zero until the last measurable concentration; AUC_{0-inf}, area under the concentration-time curve from time zero to infinity (extrapolated); C_{max}, maximum plasma concentration

[a]All PK parameters values were dose-normalized to the 10 mg dose

Figure 2. Mean Values of (A) C_{max}, (B) AUC_{0-t}, and (C) AUC_{0-inf} by dose of diazepam (linear scale)

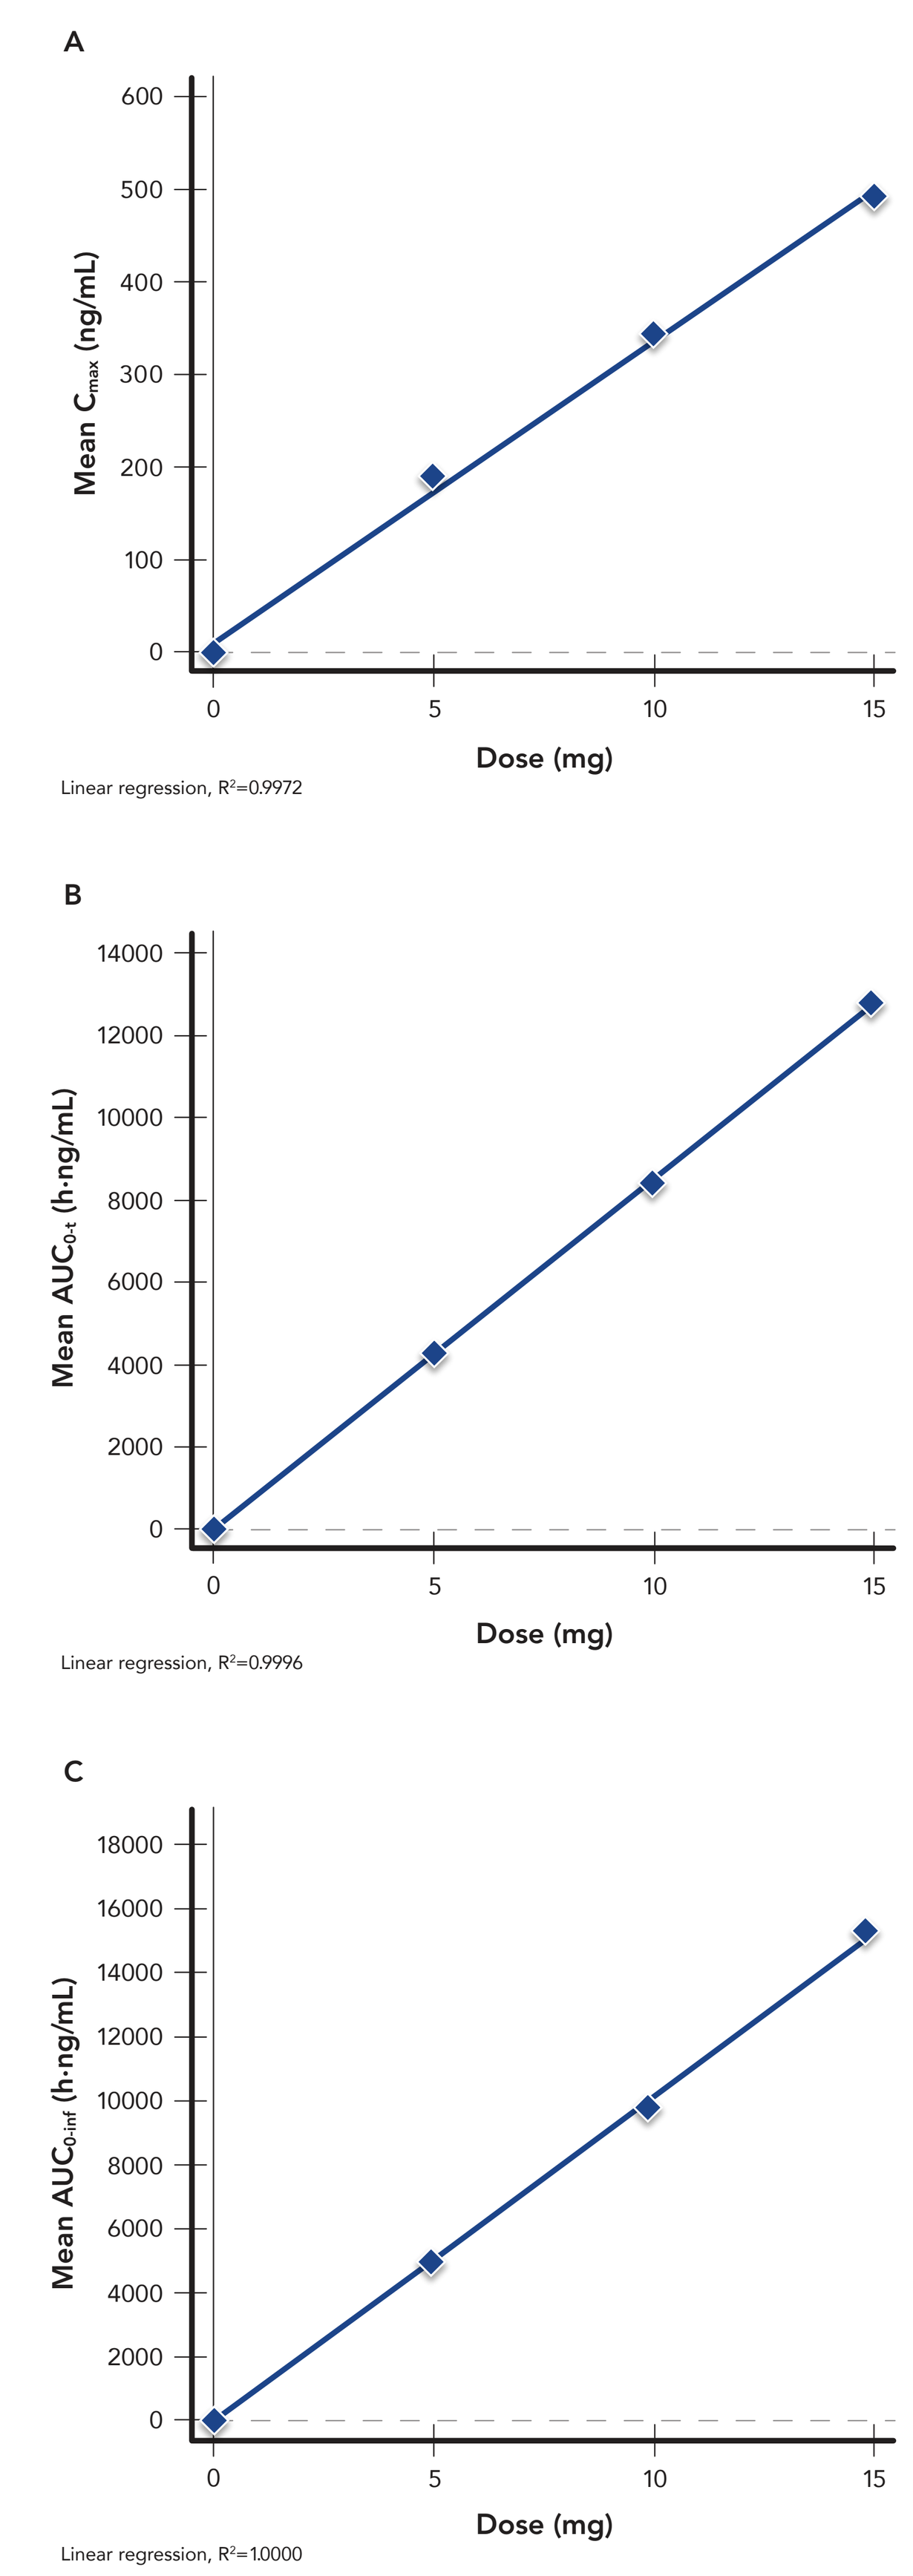
A
Mean C_{max} (ng/mL)
600
500
400
300
200
100
0
0
5
10
15
Dose (mg)
Linear regression, R²=0.9972
B
Mean AUC_{0-t} (h·ng/mL)
14000
12000
10000
8000
6000
4000
2000
0
0
5
10
15
Dose (mg)
Linear regression, R²=0.9996
C
Mean AUC_{0-inf} (h·ng/mL)
18000
16000
14000
12000
10000
8000
6000
4000
2000
0
0
5
10
15
Dose (mg)
Linear regression, R²=1.0000

SAFETY AND TOLERABILITY

- A total of 59 TEAEs were reported by 24 (80.0%) of the 30 subjects who received ≥1 dose of DBSF (**Table 6**)
- As expected, in the number of adverse events (AEs) and the number of subjects experiencing AEs was dose-related.
 - 9 TEAEs were reported by 8/27 (29.6%) subjects who received DBSF 5 mg
 - 18 TEAEs were reported by 15/26 (57.7%) subjects who received DBSF 10 mg
 - 32 TEAEs were reported by 21/30 (70.0%) subjects who received DBSF 15 mg
- The most frequent TEAEs were dose-related somnolence and dizziness
- All other TEAEs were each reported by no more than one subject by treatment group, and included fatigue, chills, and procedural dizziness
- The majority of TEAEs were mild in severity and resolved spontaneously by the end of the study
- No TEAEs were severe or serious (**Table 7**)
- 1 subject was discontinued due to an AE (increased blood pressure prior to dosing in Period 3, unrelated to study drug)
- No safety observations for vital signs or clinical laboratory results

TABLE 6: SUMMARY OF ADVERSE EVENTS

	5 mg	10 mg	15 mg	Overall
Subjects dosed, n	27	26	30	30
Subjects with ≥1 TEAE, n (%)	8 (29.6)	15 (57.7)	21 (70.0)	24 (80.0)
TEAEs, n	9	18	32	59
Severe TEAEs, n	0	0	0	0
Serious TEAEs, n	0	0	0	0
Discontinuations due to TEAEs, n	0	0	1	1
Deaths, n	0	0	0	0
Most frequent adverse events, n (%)				
Somnolence	6 (22.2)	10 (38.5)	15 (50.0)	19 (63.3)
Dizziness	1 (3.7)	1 (3.8)	3 (10.0)	4 (13.3)

TEAE: treatment-emergent adverse event

TABLE 7: FREQUENCY OF TREATMENT-EMERGENT ADVERSE EVENTS BY SEVERITY

Treatment	Severity		
	Mild	Moderate	Severe
5 mg	8	1	0
10 mg	17	1	0
15 mg	25	7	0
Overall	50	9	0

CONCLUSIONS

- These data demonstrate that the pharmacokinetics of diazepam (both C_{max} and AUC) are dose-proportional over the dosing range of 5 to 15 mg following single doses of DBSF under fasting conditions
- Overall, the administration of DBSF was safe and well tolerated in healthy subjects following a single oral dose of 5 mg, 10 mg, or 15 mg

REFERENCES

1. Tatum WO. *Epilepsy Behav.* 2002;3:535-538.

ACKNOWLEDGEMENTS

Medical writing support was provided by Nate Connors, PhD, of ACCESS Medical, LLC; medical writing support was funded by Aquestive Therapeutics.

FUNDING AND DISCLOSURES

Financial support for this study was provided by Aquestive Therapeutics. Aquestive Therapeutics participated in the interpretation of data, review, and approval of the poster. All authors contributed to the development of the poster and maintained control over the final content. Allen H. Heller and David J. Wyatt are consultants to Aquestive Therapeutics. Stephen Wargacki, Cassie Jung, and A. Mark Schobel are employees of Aquestive Therapeutics.